AI-Ready by Design: How Adimab's 15+ Years of Data Integration Makes Every Antibody Program Smarter Than the Last

Bingjie Ouyang, Kirt Johnson, Chris Sumner, Cody Williams, Jason Azze, Drew Prescott, Max Vasquez · Adimab, LLC
PEGS Boston 2026 · Session C6A: ML and Digital Integration in BiotherapeuticAnalytics

ADIMAB

INTRODUCTION

Antibody discovery generates interconnected data across library construction, selection, clonal characterization, and therapeutic lead delivery. In many organizations, this information is distributed across separate LIMS, ELN, and spreadsheet systems, limiting cross-program analysis.

Retrospective queries such as "Get all thermal stability data for Fab samples derived from Adimab 5.0 library selections" or "How do selection protocols compare across target classes?" require substantial manual data reconciliation when systems are not integrated.

Effective application of machine learning to antibody development depends on structured, linked experimental data rather than algorithmic sophistication alone. Here we describe Atlas, Adimab's integrated data platform, and how 15+ years of consistent data capture provide a foundation for machine learning models that depend on large-scale, linked experimental data, a complement to structure-based computational methods in antibody discovery.

OBJECTIVES

• Illustrate how structured, linked operational data supports downstream machine learning applications
• Demonstrate end-to-end clone traceability across 1,431 programs and 2.85M clones
• Present natural language database querying via the Model Context Protocol (MCP)
• Quantify the relationship between accumulated experimental measurements and ML prediction throughput

DATA SCALE: 15+ YEARS OF GROWTH

Atlas has captured antibody discovery data continuously since 2009. The cumulative growth of projects and clones reflects the expanding scope of Adimab's discovery operations, with each new program contributing to a shared, queryable dataset.

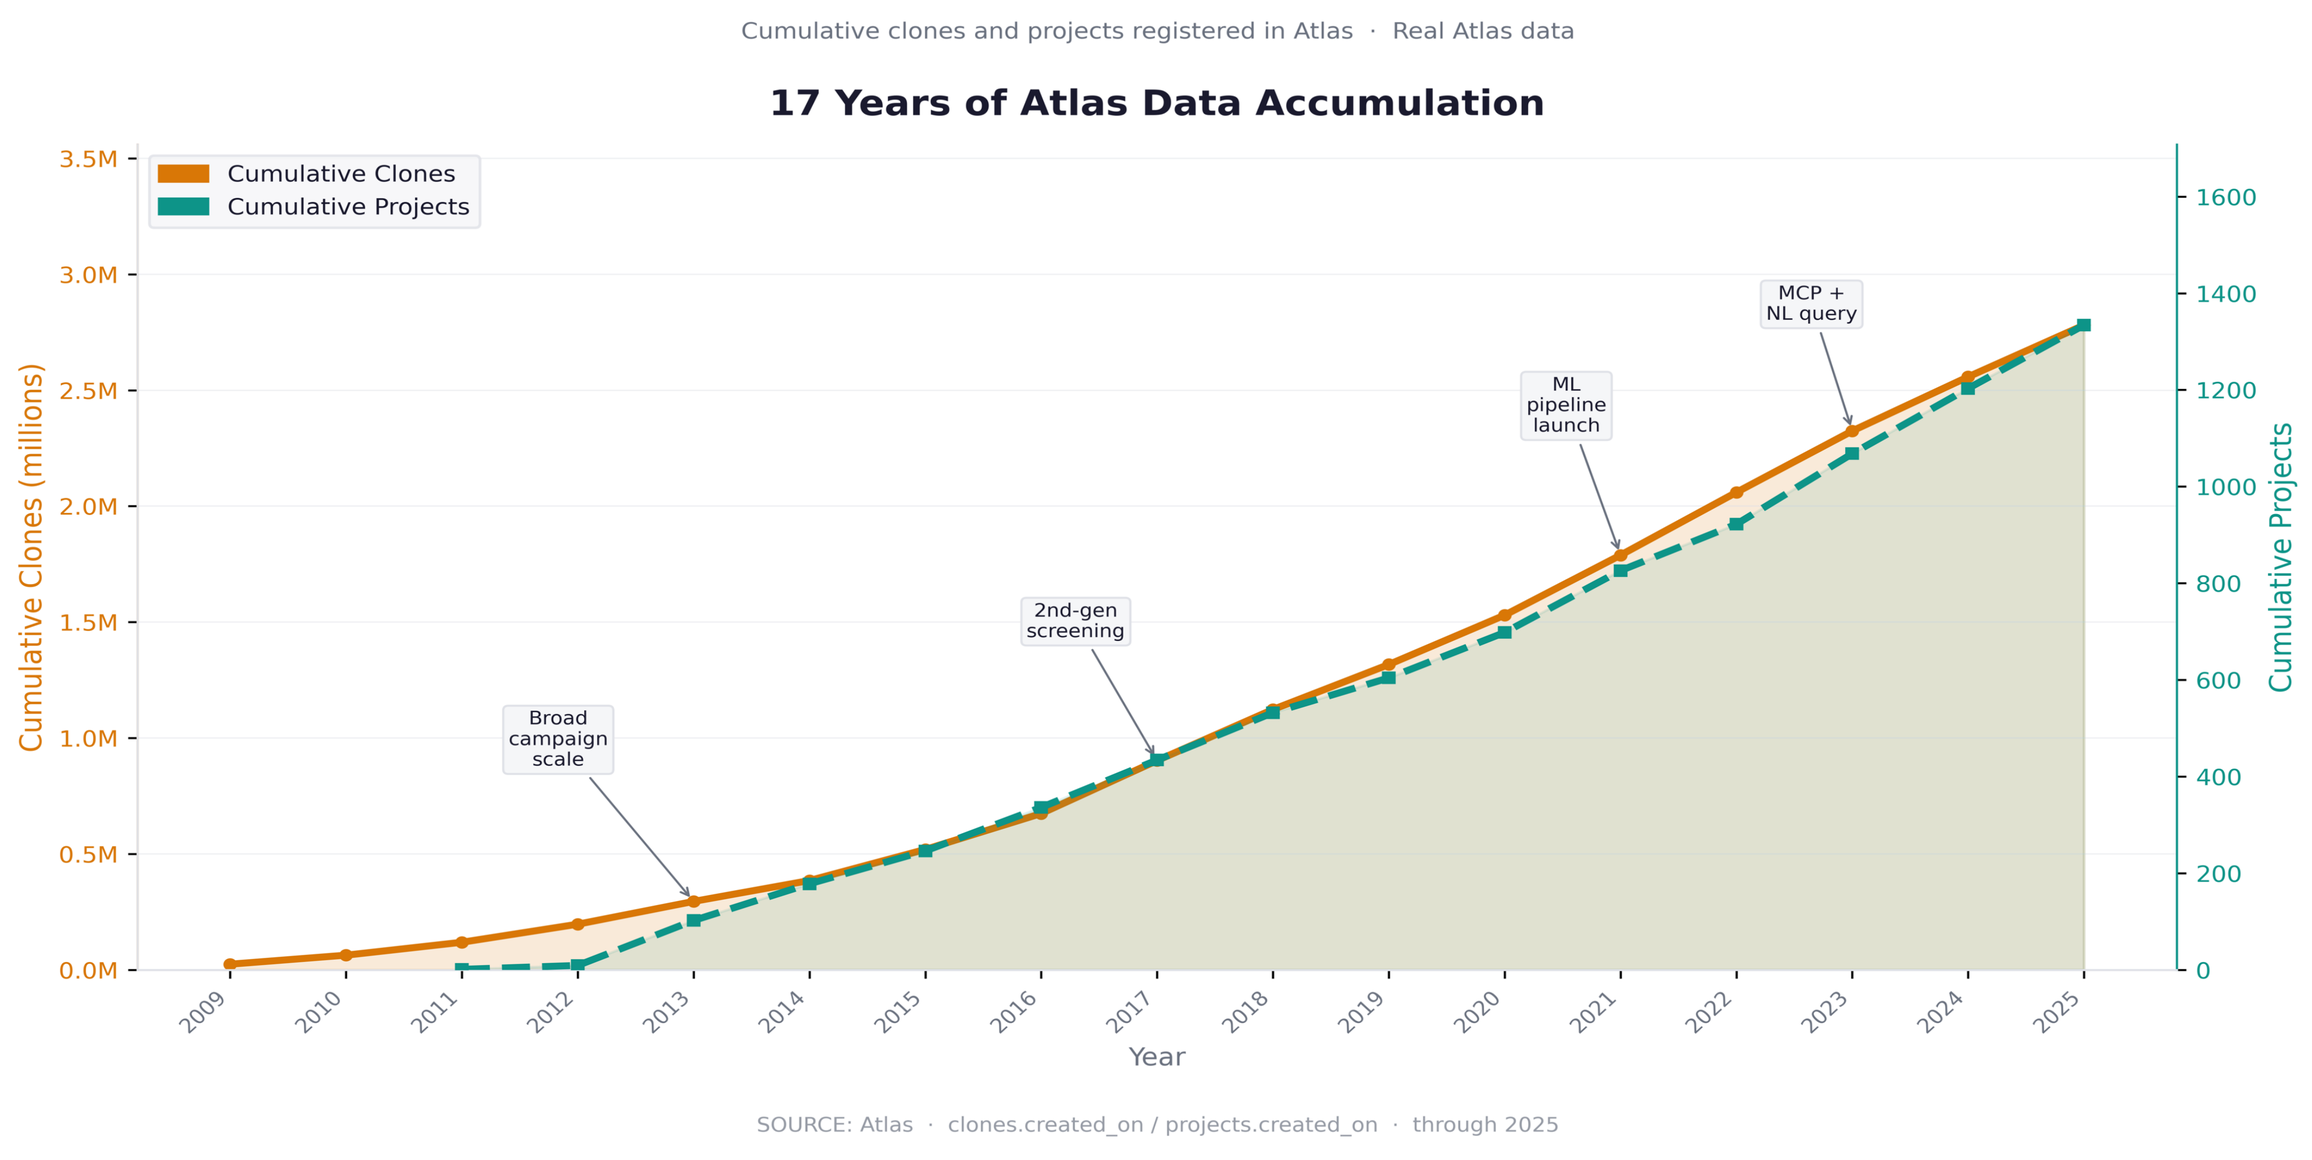
Cumulative clones and projects registered in Atlas · Real Atlas data
17 Years of Atlas Data Accumulation
Cumulative Clones
Cumulative Projects
Cumulative Clones (millions)
Cumulative Projects
Broad campaign scale
2nd-gen screening
ML pipeline launch
MCP + NL query
Year
SOURCE: Atlas · clones.created_on / projects.created_on · through 2025

FROM DISCOVERY TO DELIVERY

Atlas tracks clone progression through the full development pipeline: from project inception through clone identification and delivery to partners. Conversion rates at each stage are quantifiable from the linked dataset. Clone origins include selection-acquired (SAD, 73%), library-acquired (LAD, 25%), and other sources including engineered variants and external clones (2%).

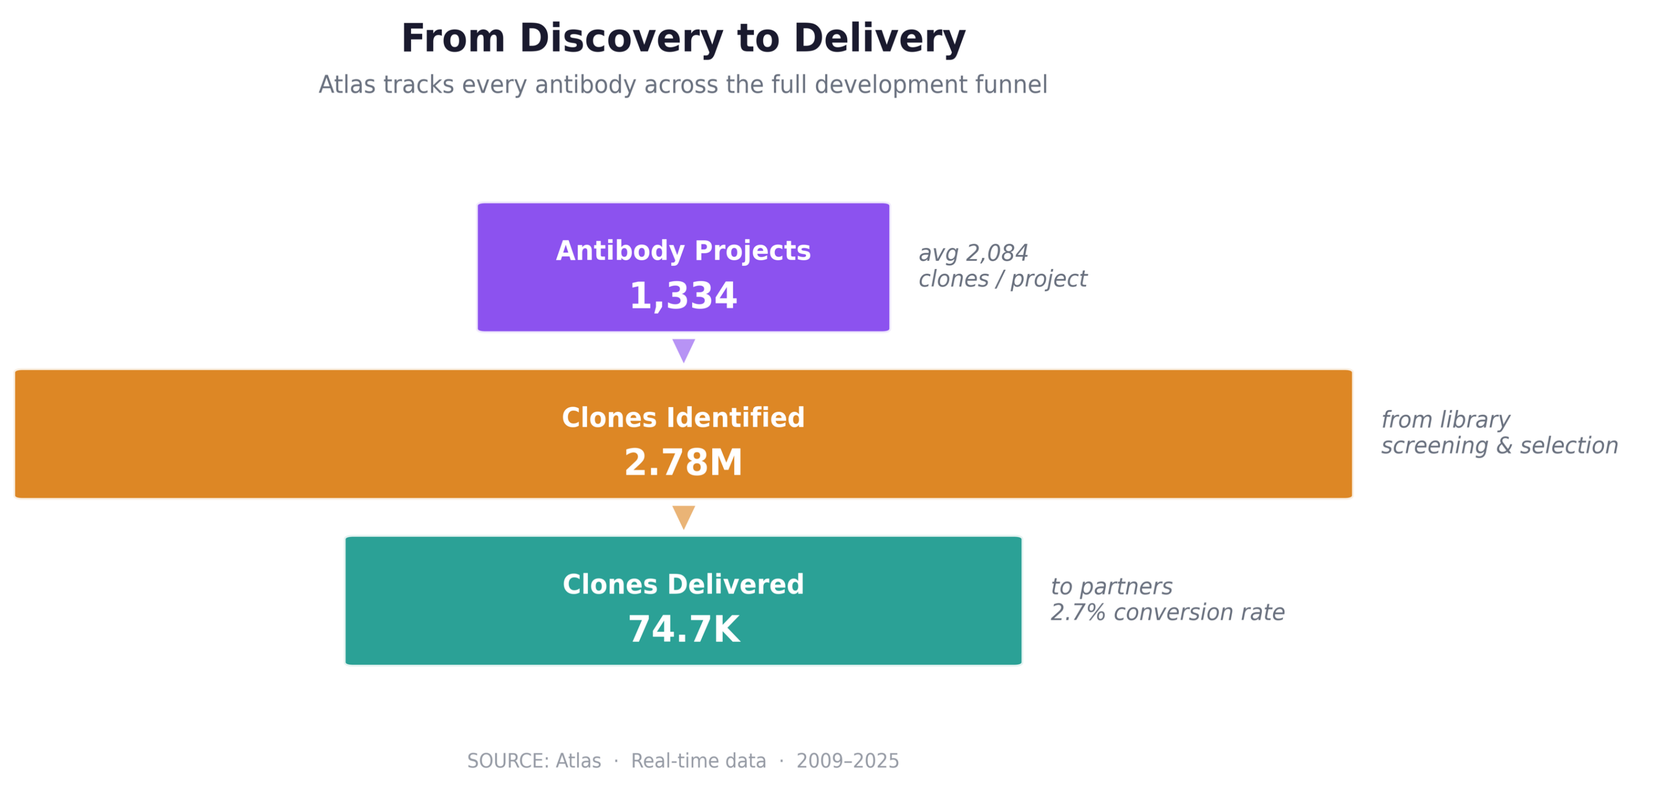
From Discovery to Delivery
Atlas tracks every antibody across the full development funnel
Antibody Projects
1,334
avg 2,084 clones / project
Clones Identified
2.78M
from library screening & selection
Clones Delivered
74.7K
to partners
2.7% conversion rate
SOURCE: Atlas · Real-time data · 2009–2025

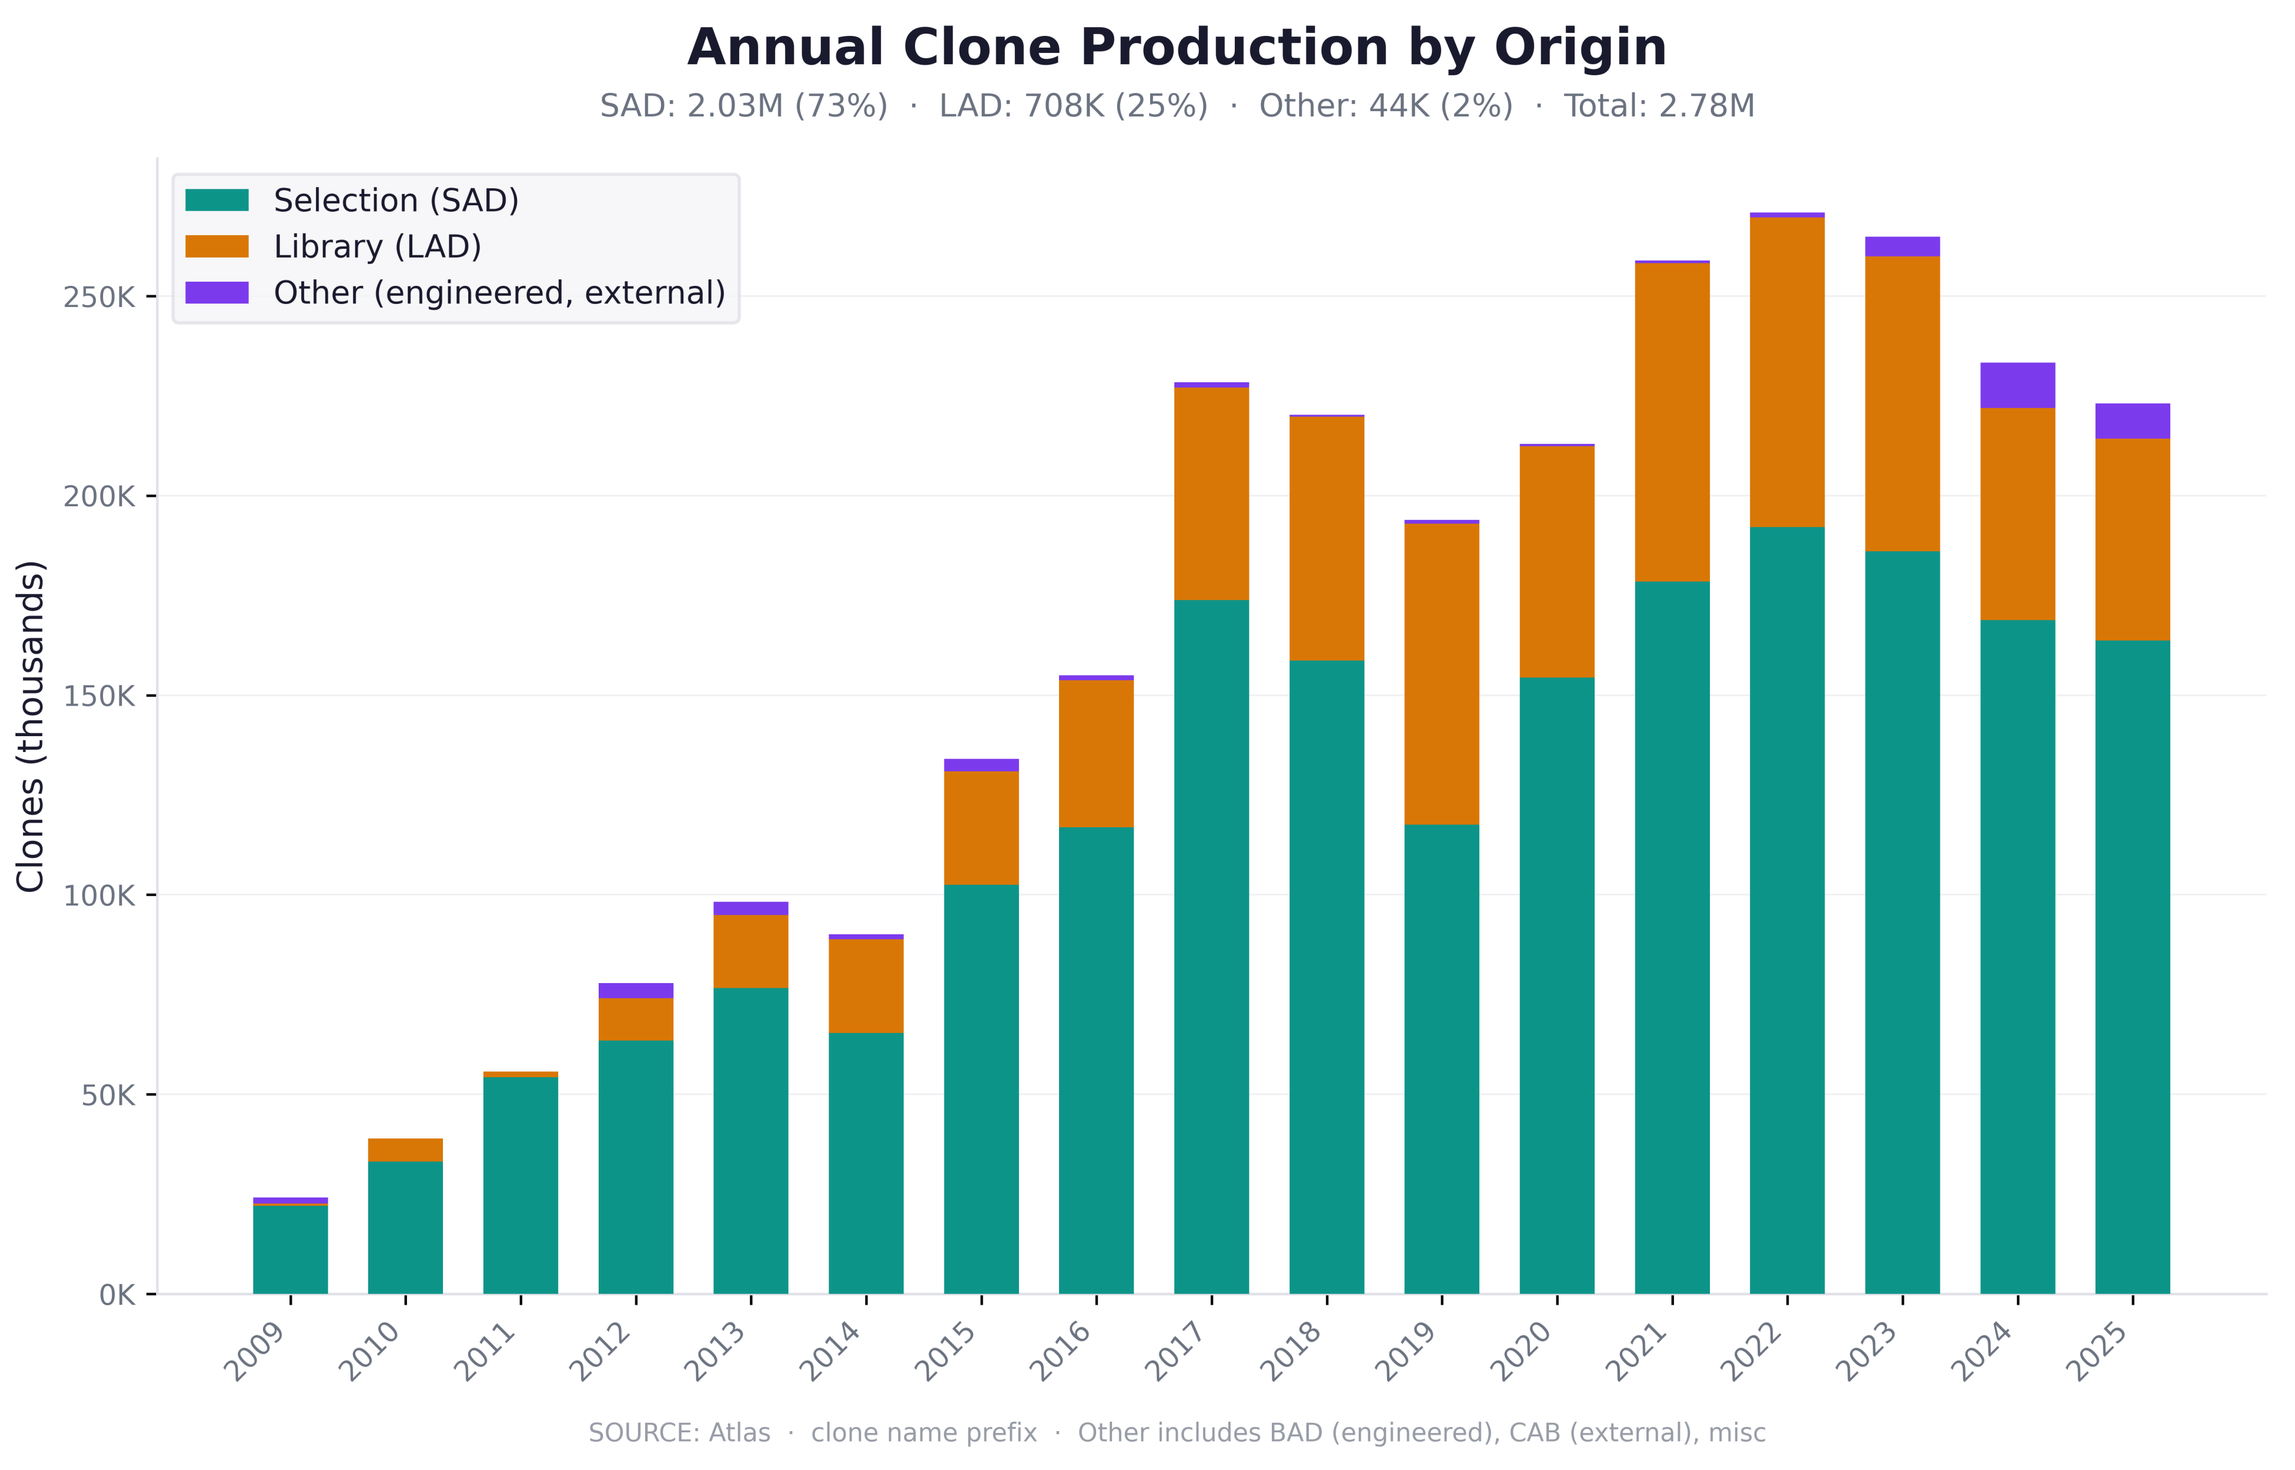
Annual Clone Production by Origin
SAD: 2.03M (73%) · LAD: 708K (25%) · Other: 44K (2%) · Total: 2.78M
Selection (SAD)
Library (LAD)
Other (engineered, external)
Clones (thousands)
SOURCE: Atlas · clone name prefix · Other includes BAD (engineered), CAB (external), misc

PROJECT DIVERSITY

Atlas captures data across the full range of Adimab's discovery programs: 1,431 projects spanning Type I (52%), Type II (37%), Type III (10%), and other formats.

This breadth of the data spans multiple project types, therapeutic targets, and antibody formats. This contributes to the diversity of the training dataset. Models trained on data from a single project type or target class may not generalize well; Atlas's cross-program dataset mitigates this limitation.

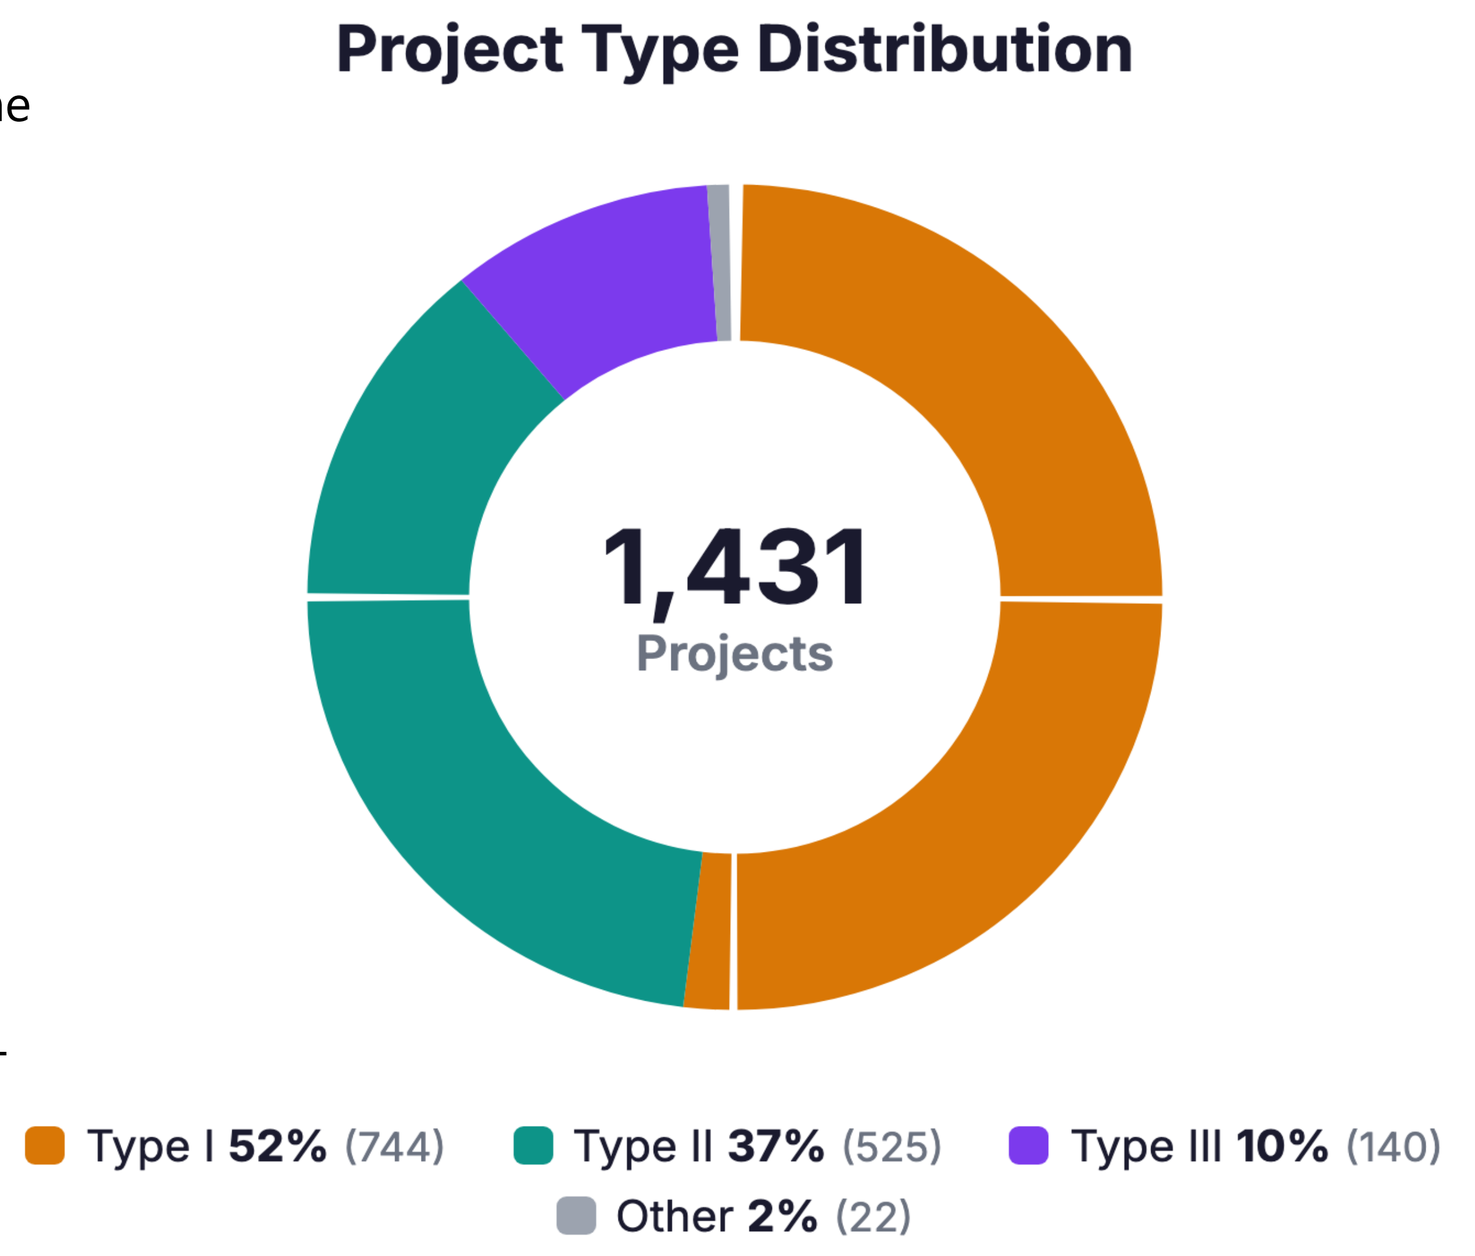
Project Type Distribution
1,431
Projects
Type I 52% (744)
Type II 37% (525)
Type III 10% (140)
Other 2% (22)

ATLAS: FOUR INTEGRATED MODULES

Atlas is not a conventional LIMS. It is a purpose-built relational data system designed to support scientific decision-making by linking experimental data across the full antibody discovery workflow.

• Data Integration Layer — PostgreSQL relational schema enforcing naming conventions and entity relationships at creation time

• Assay Data Warehouse — 34+ assay types with standardized units, QC flags, and cross-program benchmarking

• Natural Language Interface — MCP-based querying enabling plain-English access to the full database

• ML Prediction Engine — AWS SageMaker integration for sequence-to-property prediction

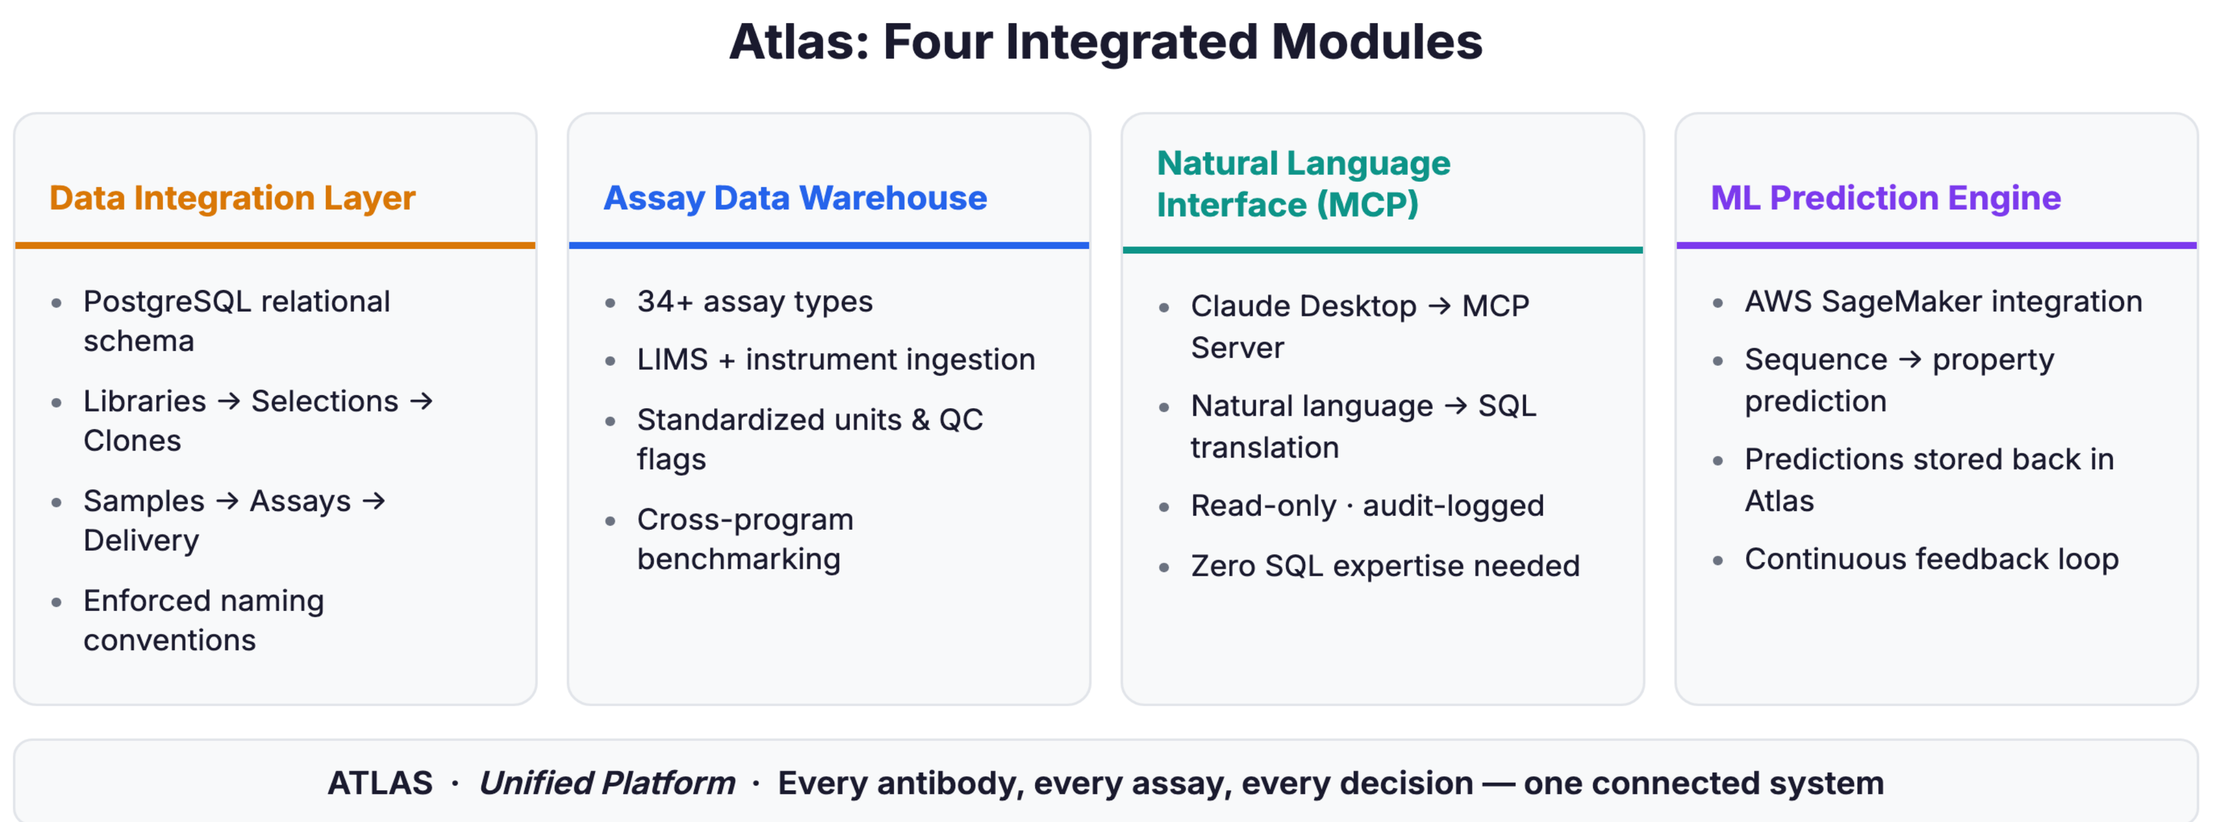
Atlas: Four Integrated Modules
Data Integration Layer
PostgreSQL relational schema
Libraries → Selections → Clones
Samples → Assays → Delivery
Enforced naming conventions
Assay Data Warehouse
34+ assay types
LIMS + instrument ingestion
Standardized units & QC flags
Cross-program benchmarking
Natural Language Interface (MCP)
Claude Desktop → MCP Server
Natural language → SQL translation
Read-only · audit-logged
Zero SQL expertise needed
ML Prediction Engine
AWS SageMaker integration
Sequence → property prediction
Predictions stored back in Atlas
Continuous feedback loop
ATLAS · Unified Platform · Every antibody, every assay, every decision — one connected system

ATLAS USER INTERFACE

The Atlas web interface provides each researcher with a personalized view of active projects, resource allocation, recent productions, and selection campaigns. Because scientists use Atlas as their primary operational tool, data is captured at the point of origin. They are structured, linked, and complete without retroactive curation.

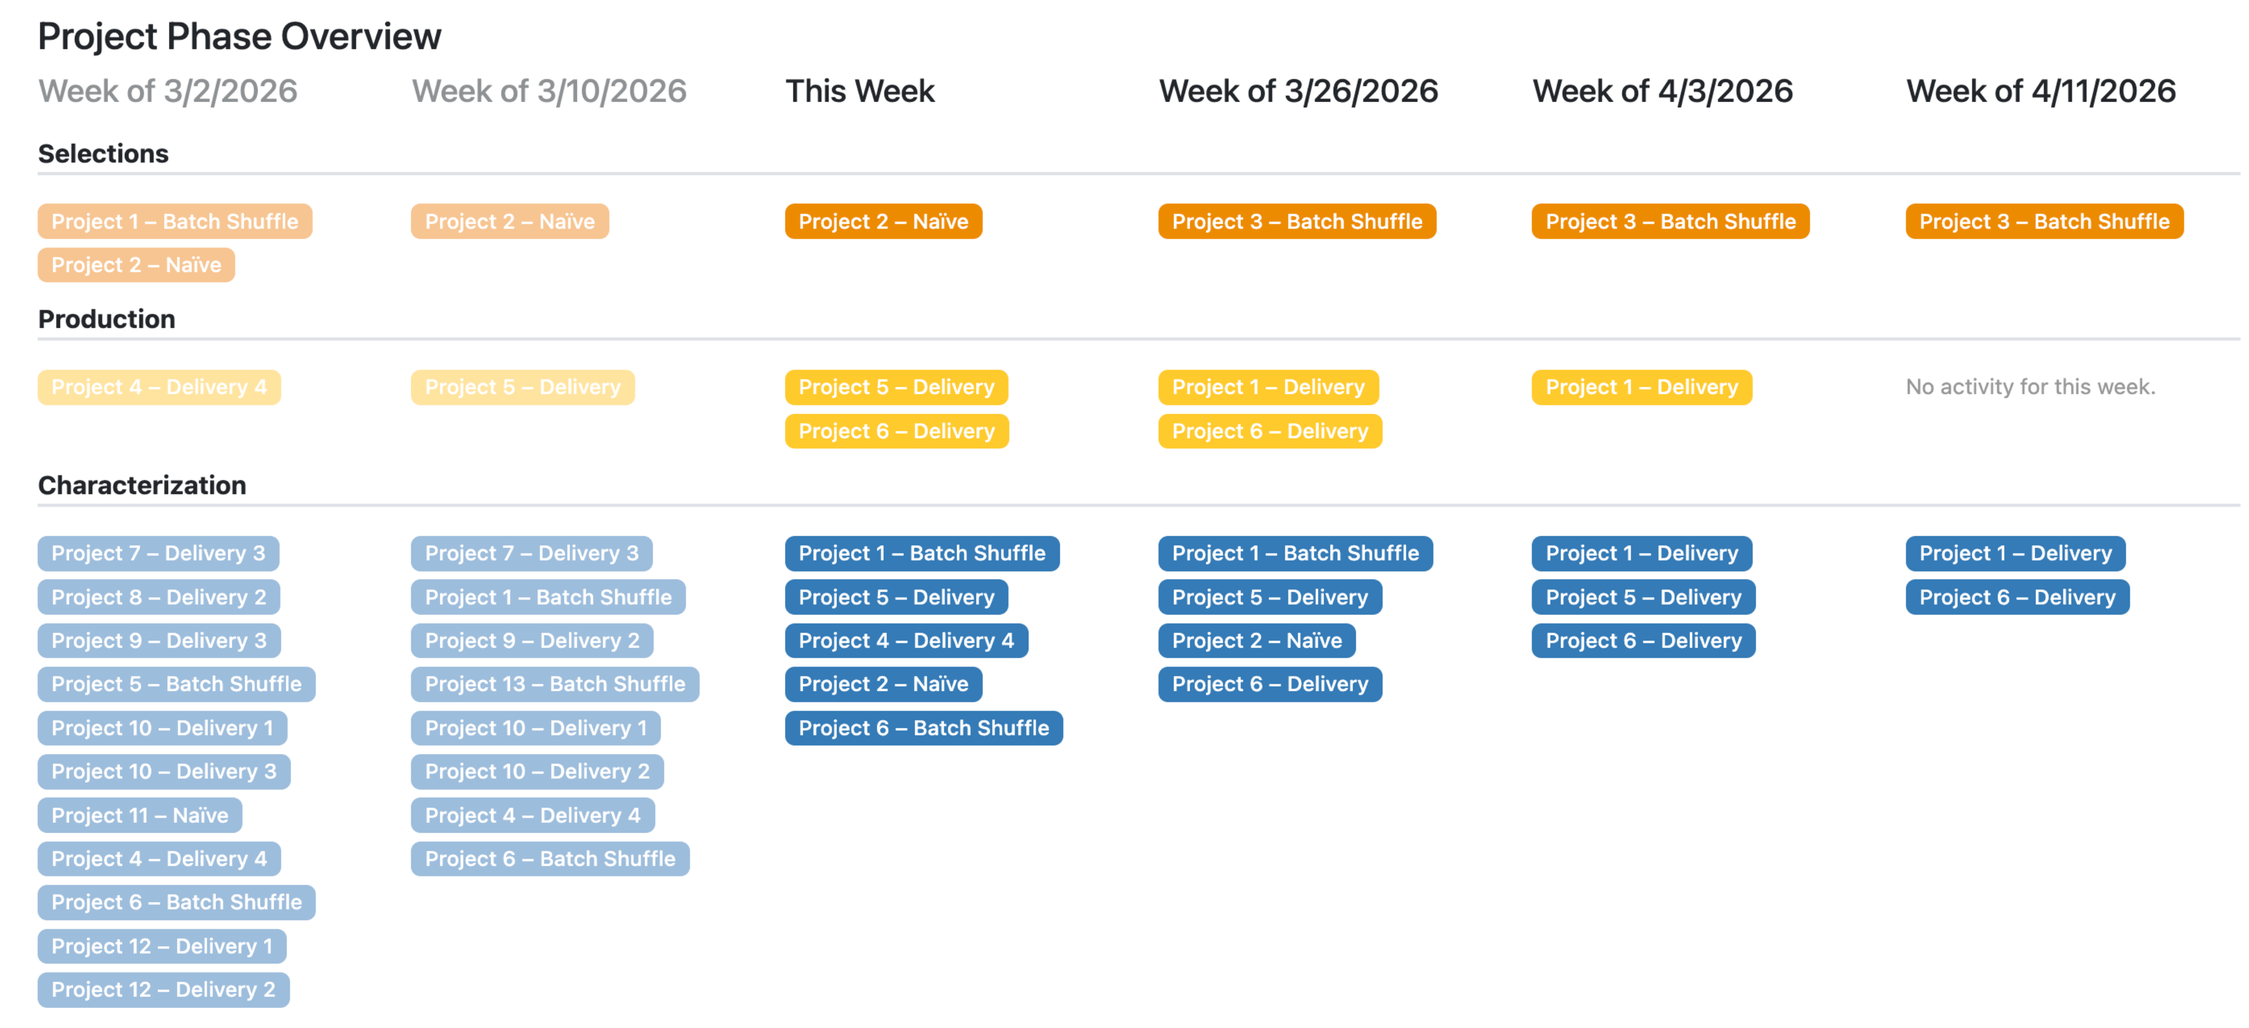
Project Phase Overview
Week of 3/2/2026
Week of 3/10/2026
This Week
Week of 3/26/2026
Week of 4/3/2026
Week of 4/11/2026
Selections
Production
Characterization
No activity for this week.

THE CONNECTED CLONE JOURNEY

A central feature of Atlas is end-to-end lineage tracking. Each clone record is automatically linked to its source library, selection history, characterization data, and delivery status at the time of creation.

This traceability is bidirectional: users can trace forward from a library to all derived clones, or backward from a delivered clone to its original library composition. Six stages are tracked: Library Design, Selection Campaigns, Clone Identification, Sample Production, Characterization Assays, and Delivery.

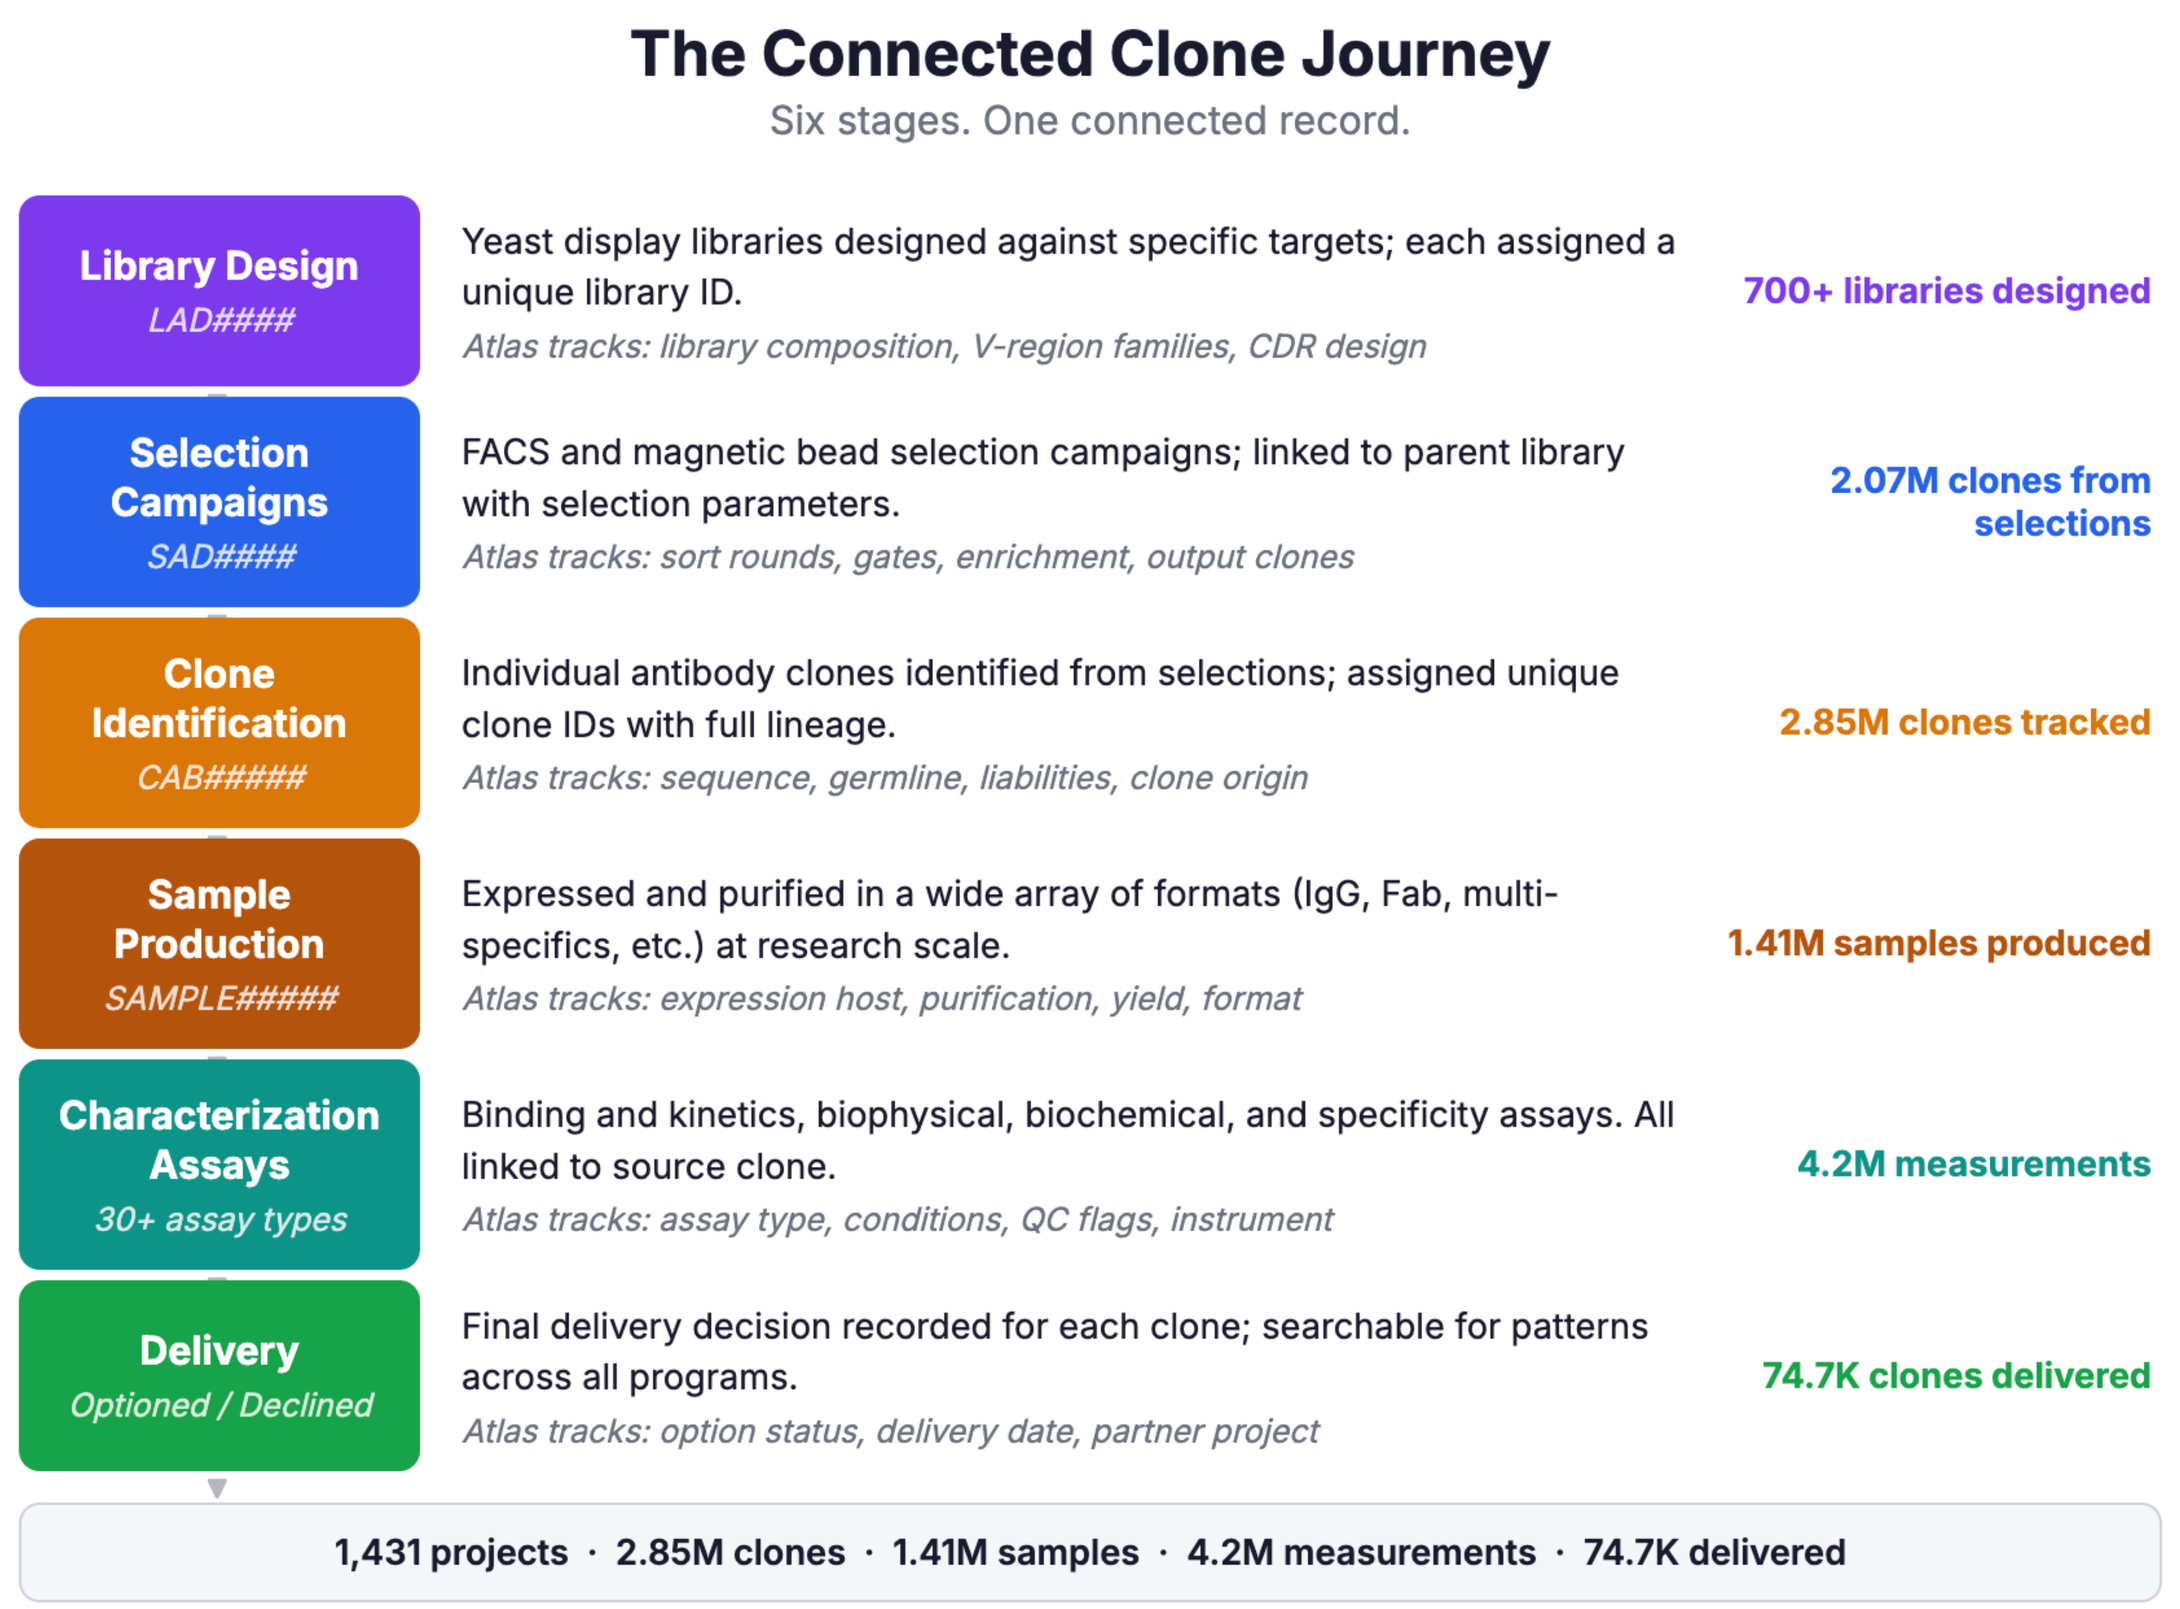
The Connected Clone Journey
Six stages. One connected record.
Library Design
LAD####
Yeast display libraries designed against specific targets; each assigned a unique library ID.
Atlas tracks: library composition, V-region families, CDR design
700+ libraries designed
Selection Campaigns
SAD####
FACS and magnetic bead selection campaigns; linked to parent library with selection parameters.
Atlas tracks: sort rounds, gates, enrichment, output clones
2.07M clones from selections
Clone Identification
CAB#####
Individual antibody clones identified from selections; assigned unique clone IDs with full lineage.
Atlas tracks: sequence, germline, liabilities, clone origin
2.85M clones tracked
Sample Production
SAMPLE#####
Expressed and purified in a wide array of formats (IgG, Fab, multi-specifics, etc.) at research scale.
Atlas tracks: expression host, purification, yield, format
1.41M samples produced
Characterization Assays
30+ assay types
Binding and kinetics, biophysical, biochemical, and specificity assays. All linked to source clone.
Atlas tracks: assay type, conditions, QC flags, instrument
4.2M measurements
Delivery
Optioned / Declined
Final delivery decision recorded for each clone; searchable for patterns across all programs.
Atlas tracks: option status, delivery date, partner project
74.7K clones delivered
1,431 projects · 2.85M clones · 1.41M samples · 4.2M measurements · 74.7K delivered

THE CHARACTERIZATION DATA WAREHOUSE

Atlas contains 4.1 million characterization measurements across 16 assay platforms, each linked to its source clone and project. This structured data warehouse supports cross-program analysis: HIC retention time predictions, developability assessments, and other ML applications draw on the full history of measurements accumulated over 15 years of routine operational use.

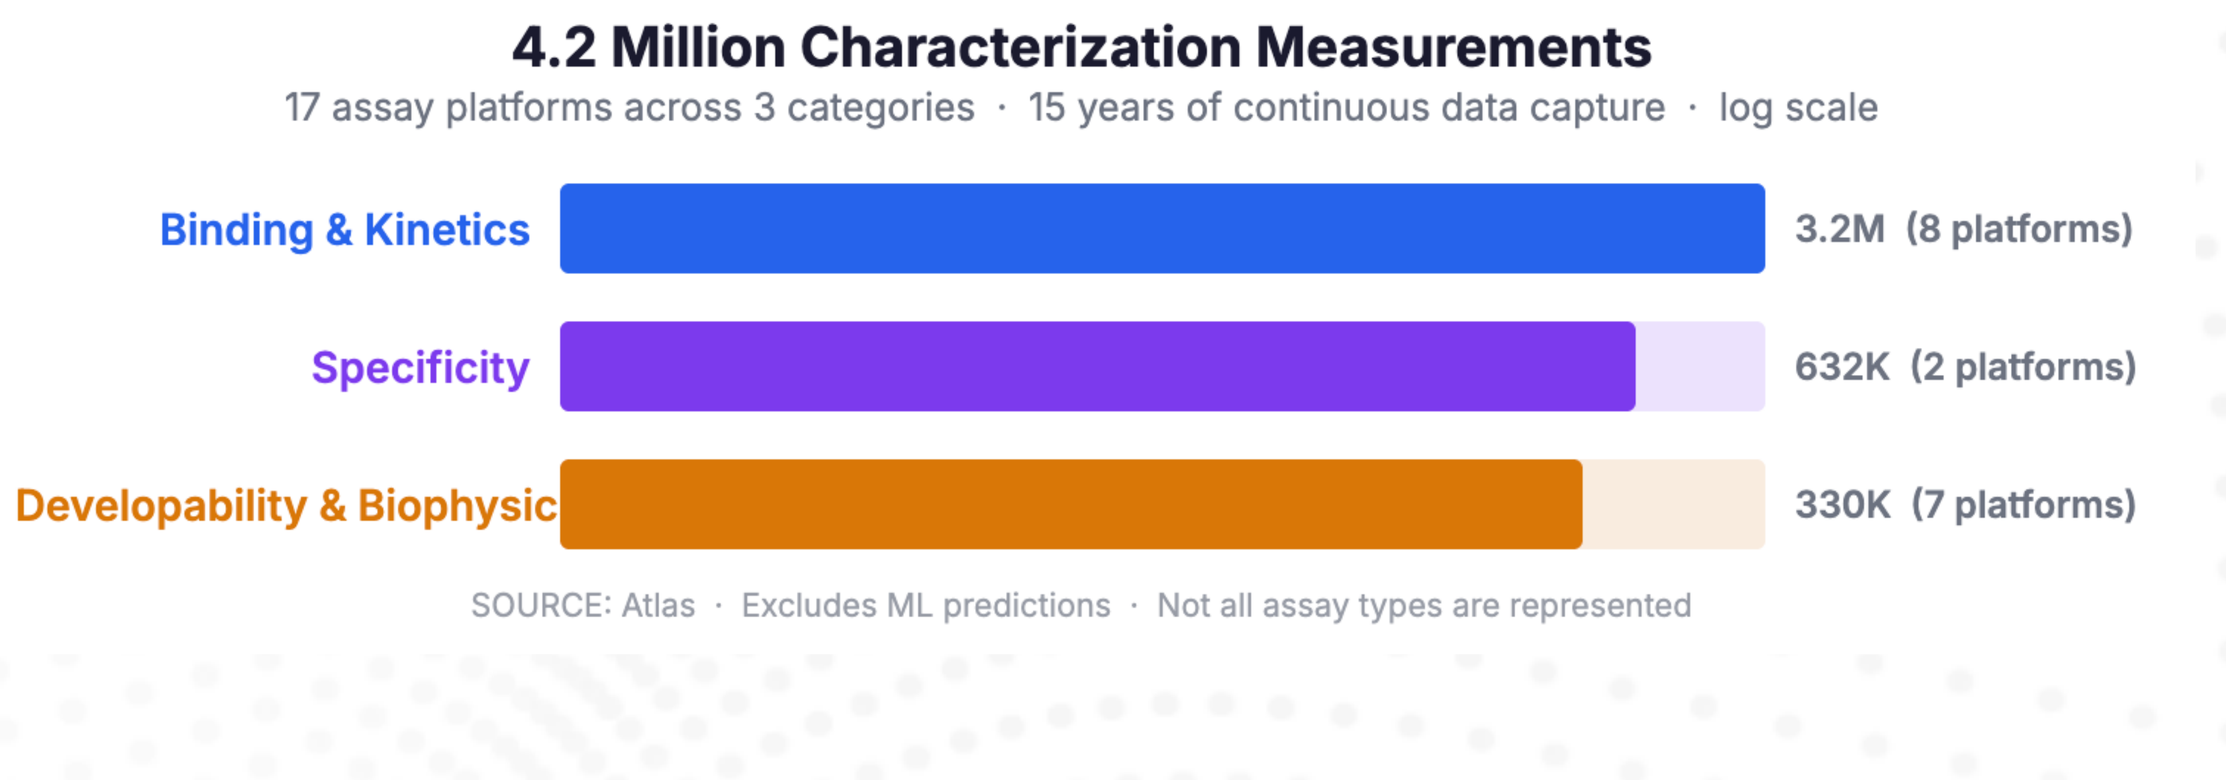
4.2 Million Characterization Measurements
17 assay platforms across 3 categories · 15 years of continuous data capture · log scale
Binding & Kinetics
3.2M (8 platforms)
Specificity
632K (2 platforms)
Developability & Biophysic
330K (7 platforms)
SOURCE: Atlas · Excludes ML predictions · Not all assay types are represented

ML ENABLED DEVELOPABILITY PREDICTION

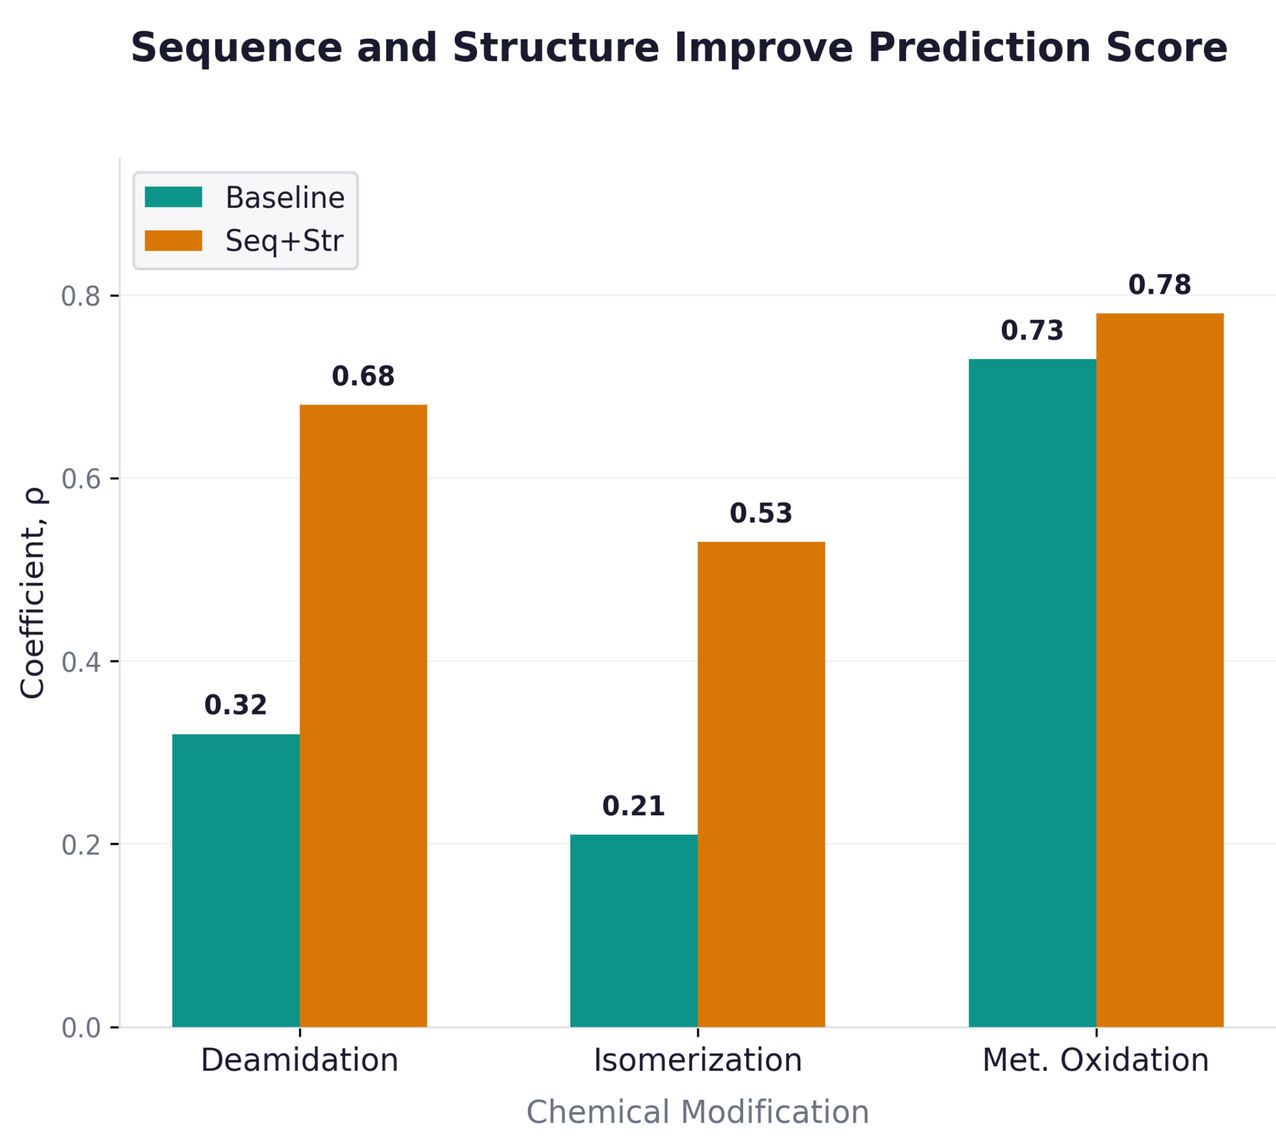
Sequence and Structure Improve Prediction Score
Baseline
Seq+Str
Coefficient, ρ
0.32
0.68
0.21
0.53
0.73
0.78
Deamidation
Isomerization
Met. Oxidation
Chemical Modification

Atlas warehouses 11,000+ site-specific accelerated stress measurements across 700+ antibodies, enabling ML models that combine sequence motifs with structural context. Prediction scores (ρ) improve 2–2.5× over baseline for deamidation and isomerization liabilities.

HIC retention time is a key developability indicator reflecting antibody hydrophobicity. Atlas's linked sequence and characterization data, with 103K experimental measurements accumulated over 11 years, provided the training foundation for a predictive model deployed on AWS SageMaker in 2022.

Predicted values are stored in Atlas alongside experimental results, enabling direct comparison and iterative model refinement. The integration of predictions back into the same system that generated the training data creates a closed feedback loop: each experimentally validated prediction improves future model iterations.

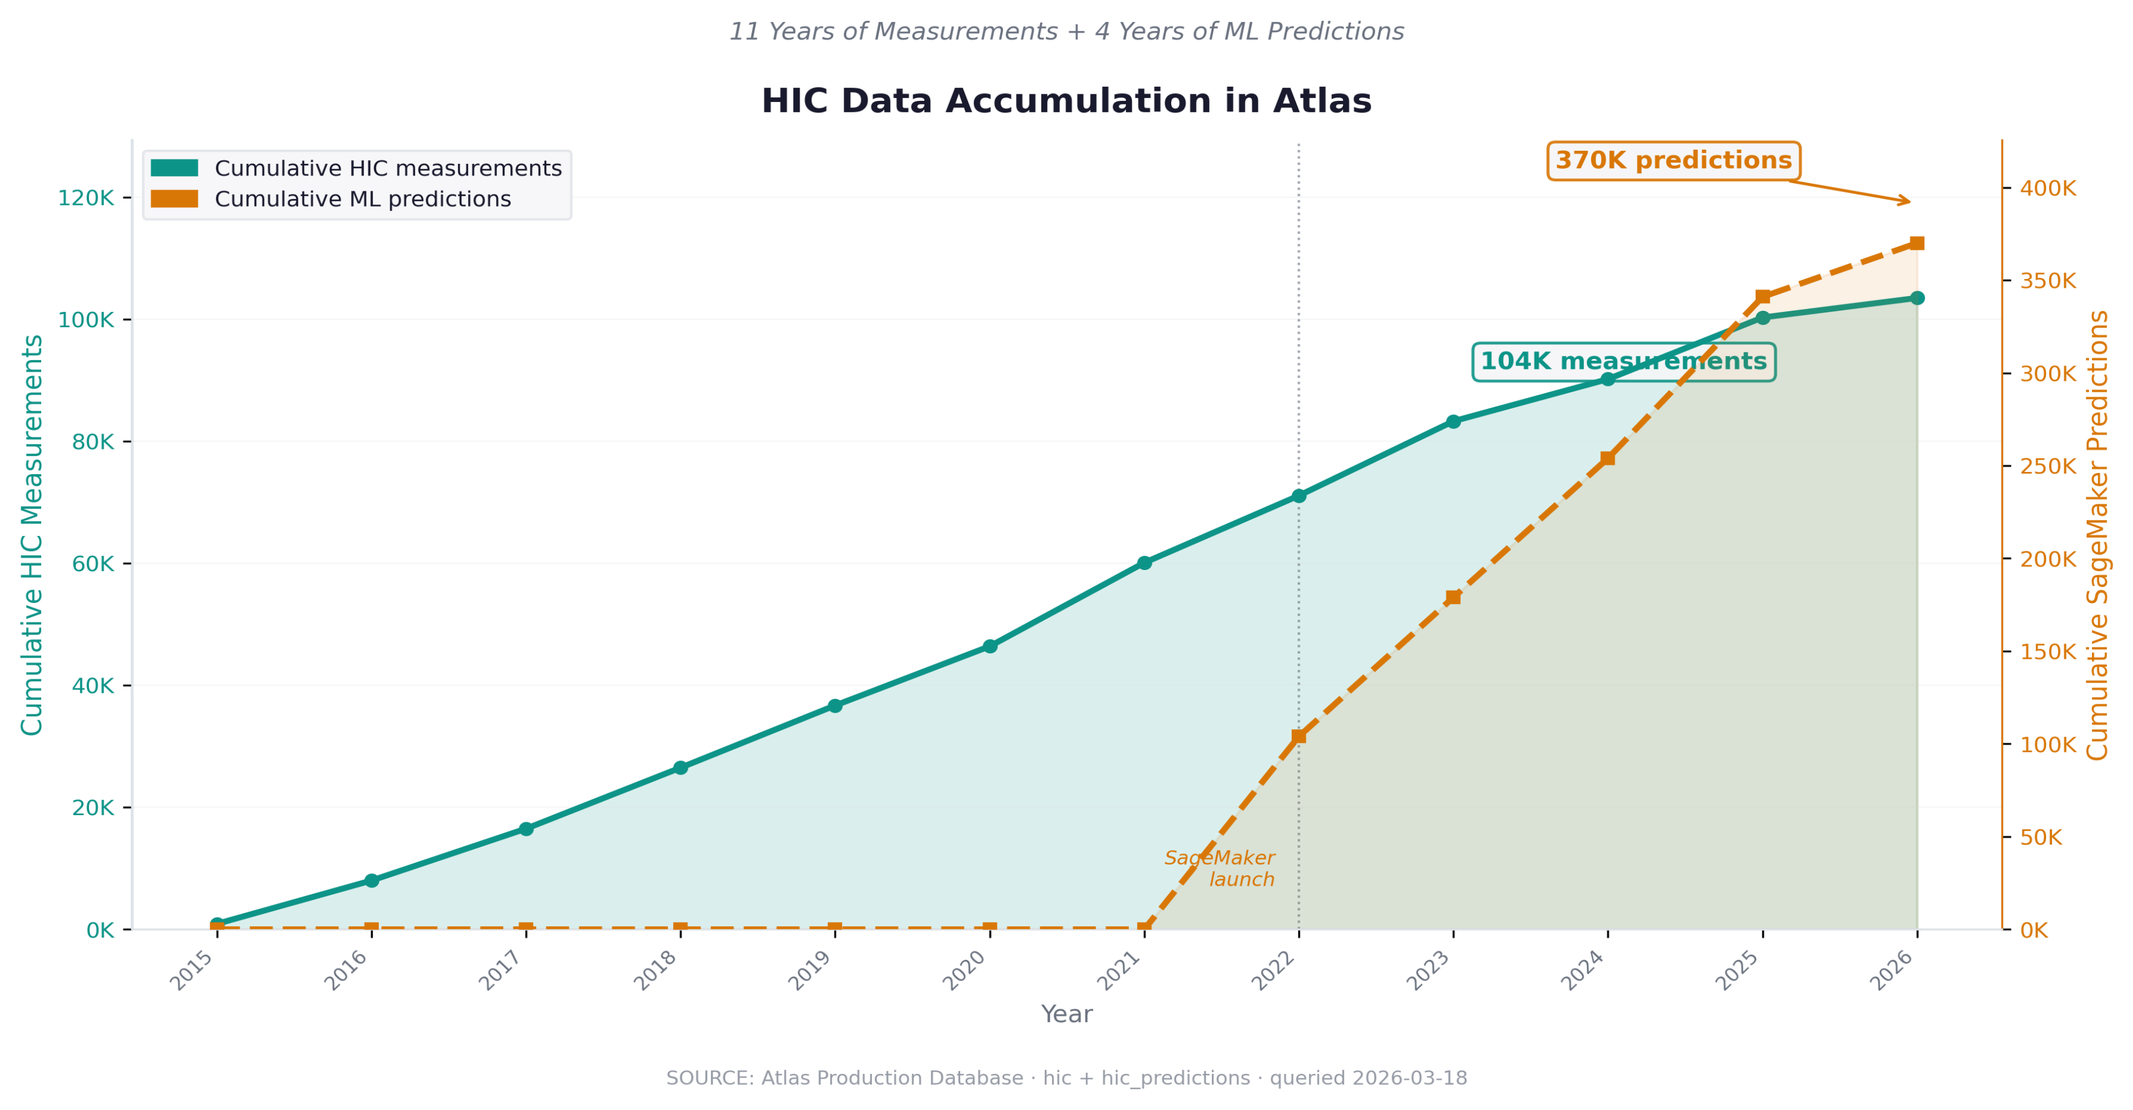
11 Years of Measurements + 4 Years of ML Predictions
HIC Data Accumulation in Atlas
Cumulative HIC measurements
Cumulative ML predictions
370K predictions
104K measurements
SageMaker launch
Cumulative HIC Measurements
Cumulative SageMaker Predictions
Year
SOURCE: Atlas Production Database · hic + hic_predictions · queried 2026-03-18

NATURAL LANGUAGE ACCESS VIA MCP→ MODEL PERFORMANCE

Atlas is accessible via the Model Context Protocol (MCP), which translates natural language queries into validated SQL and returns structured results. This enables researchers without database expertise to query 17 years of linked experimental data directly.

The query pathway is: Claude Desktop → Atlas MCP Server → SQL generation and validation → structured results. All queries are read-only and audit-logged. For example, a researcher can ask "What are the HIC retention times for clones from project X?" and receive tabulated results without writing SQL.

The utility of this interface depends directly on the consistency and completeness of the underlying data model. These qualities result from Atlas's long-term use as an operational system.

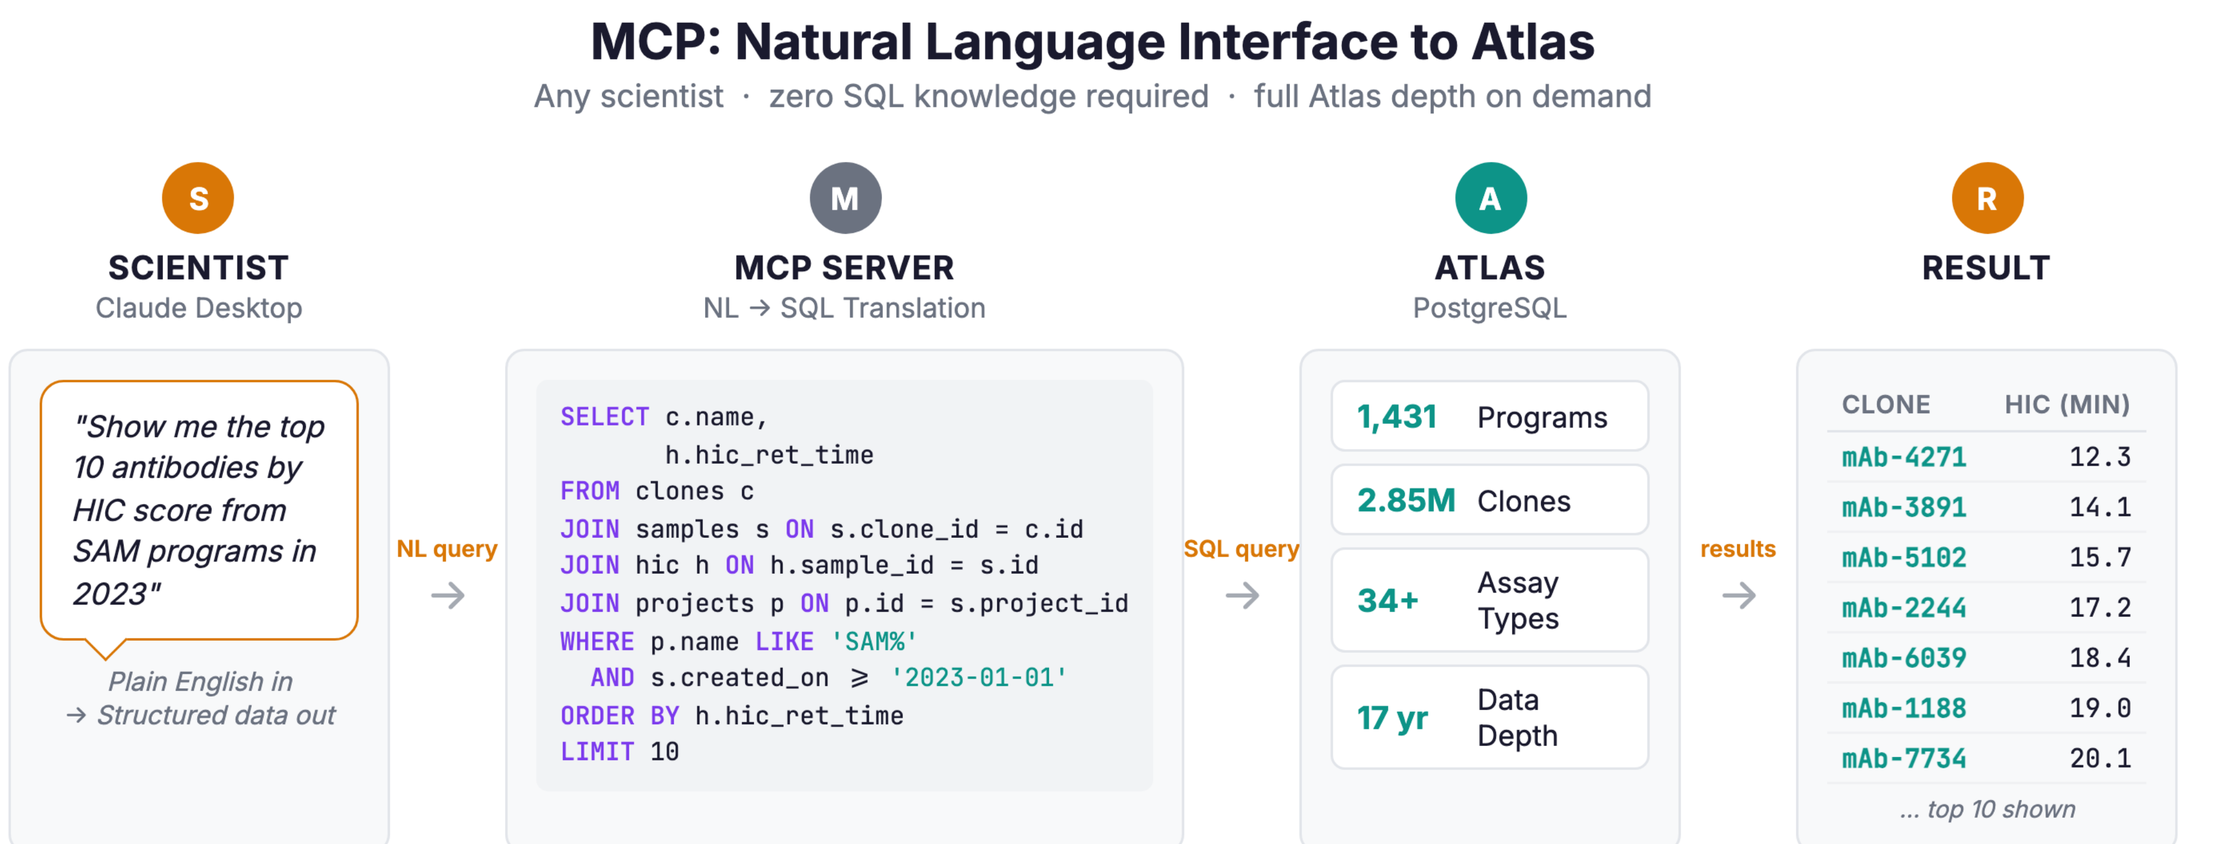
MCP: Natural Language Interface to Atlas
Any scientist · zero SQL knowledge required · full Atlas depth on demand
SCIENTIST
Claude Desktop
"Show me the top 10 antibodies by HIC score from SAM programs in 2023"
Plain English in → Structured data out
NL query
MCP SERVER
NL → SQL Translation
SELECT c.name, h.hic_ret_time
FROM clones c
JOIN samples s ON s.clone_id = c.id
JOIN hic h ON h.sample_id = s.id
JOIN projects p ON p.id = s.project_id
WHERE p.name LIKE 'SAM%'
AND s.created_on ≥ '2023-01-01'
ORDER BY h.hic_ret_time
LIMIT 10
SQL query
ATLAS
PostgreSQL
1,431 Programs
2.85M Clones
34+ Assay Types
17 yr Data Depth
results
RESULT
CLONE HIC (MIN)
mAb-4271 12.3
mAb-3891 14.1
mAb-5102 15.7
mAb-2244 17.2
mAb-6039 18.4
mAb-1188 19.0
mAb-7734 20.1
... top 10 shown

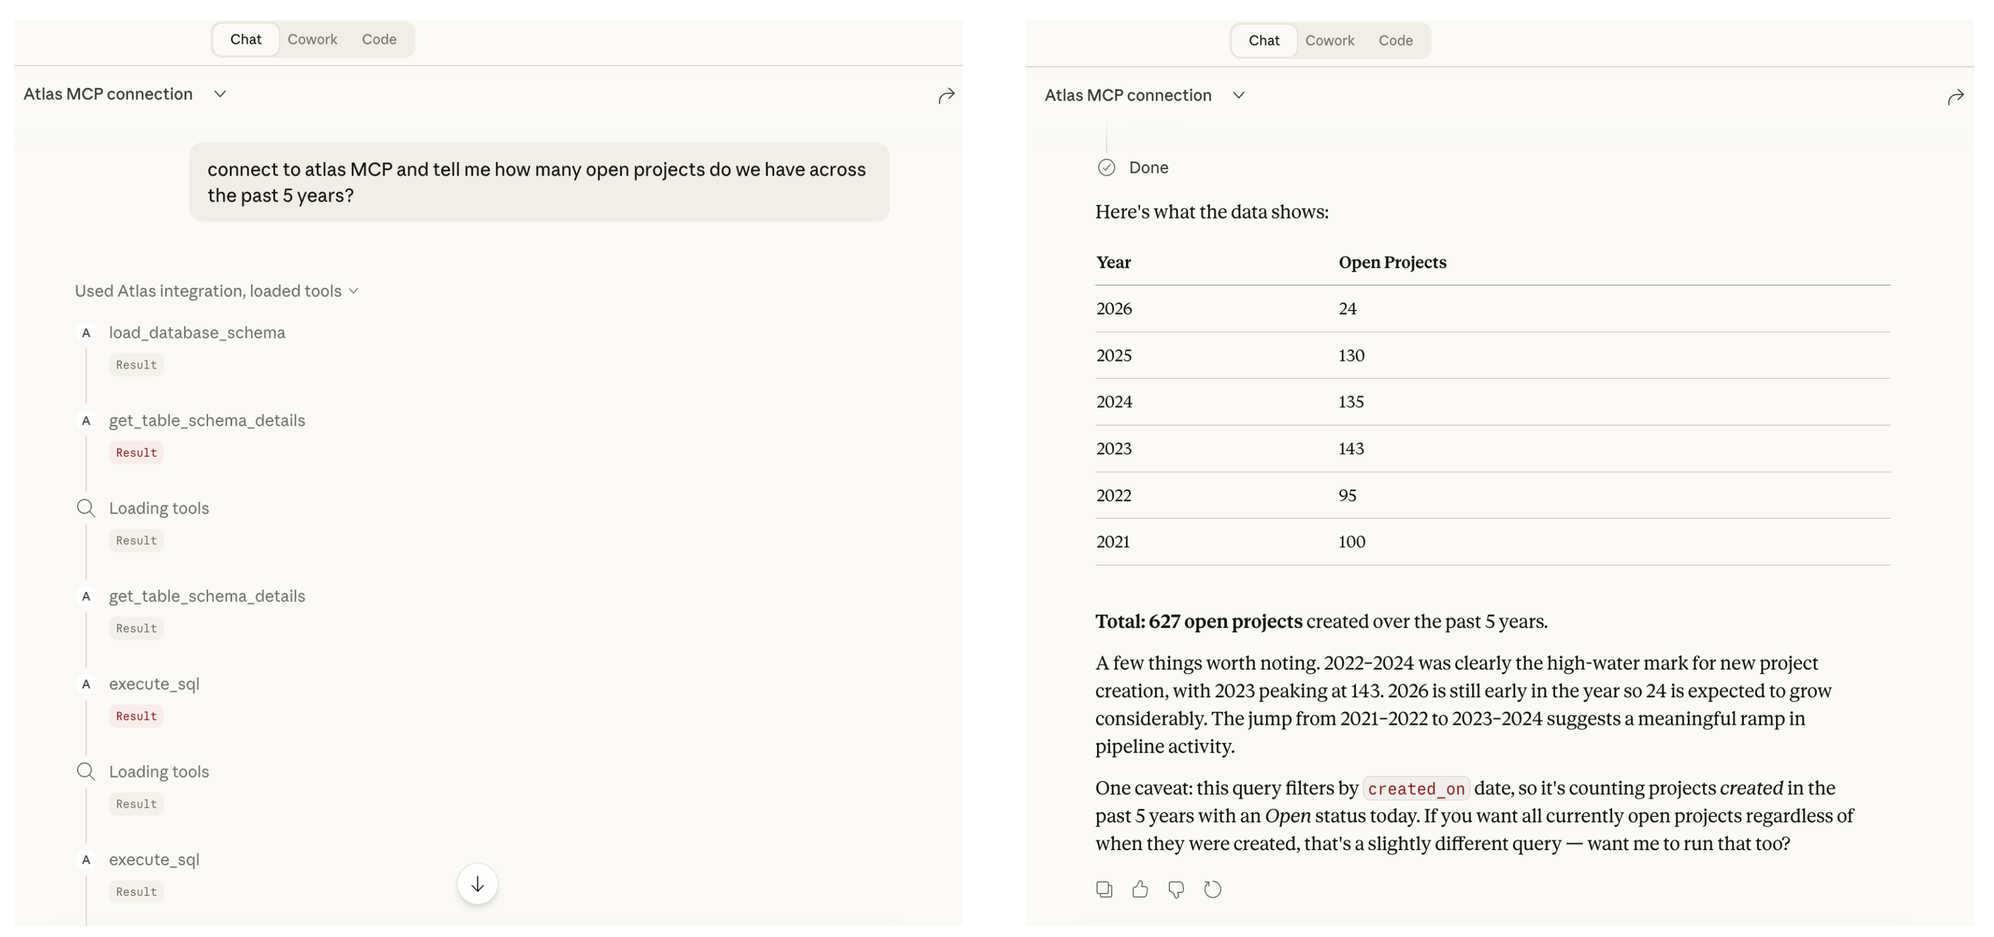
Chat Cowork Code
Atlas MCP connection
connect to atlas MCP and tell me how many open projects do we have across the past 5 years?
Used Atlas integration, loaded tools
load_database_schema
get_table_schema_details
Loading tools
execute_sql
Done
Here's what the data shows:
Year Open Projects
2026 24
2025 130
2024 135
2023 143
2022 95
2021 100
Total: 627 open projects created over the past 5 years.

CONCLUSIONS

1 Data architecture decisions precede and constrain ML capability

Schema and workflow design choices made in 2010 enabled machine learning applications not deployed until 2022. Retrospective integration of fragmented data systems is substantially more difficult than prospective design.

2 Domain-specific relational models support cumulative analysis

Atlas's schema is organized by scientific workflow rather than sample type, and links 1,334 programs across 17 years. Each new project is immediately queryable in the context of all prior data.

3 Natural language interfaces lower the barrier to institutional data access

MCP enables researchers to query the full Atlas database without SQL expertise, making 17 years of linked experimental data accessible to all team members.

4 ML prediction throughput scales with accumulated experimental data

SageMaker-based predictions exceed experimental measurement throughput by approximately 10× (104K predictions vs. 11K measurements per year). Predicted values are stored alongside experimental data, supporting iterative model evaluation.

Contact Adimab
If you are interested in partnering with Adimab, please reach out to our Business Development department at bd@adimab.com.
The QR code on the right links to Adimab posters and other resources.