Generating potent and specific soluble TCR-based T cell engagers via a high-throughput yeast-based platform

Saira Reyes[1], Vivien Qiao[1], Mrunal Sakharkar[1], Amanda Chan[2,3], Emme Pogue[2,3], Irina Burnina[1], Elizabeth McGurk[1], Cameron Henkel[1], Asparouh Lilov[1], Todd Boland[1], Elizabeth Parker[1], Michael Brown[1], Cory Ahonen[1], Arvind Sivasubramanian[1], James Geoghegan[1], Eric Krauland[1], Michael E. Birnbaum[2,3], C. Garrett Rappazzo[1]

[1] Adimab LLC, Lebanon, NH
[2] Department of Biological Engineering, Cambridge, MA
[3] Koch Institute for Integrative Cancer Research, Cambridge, MA

ADIMAB

Background – Soluble TCR-based T cell engagers

pHLA-targeting therapeutics

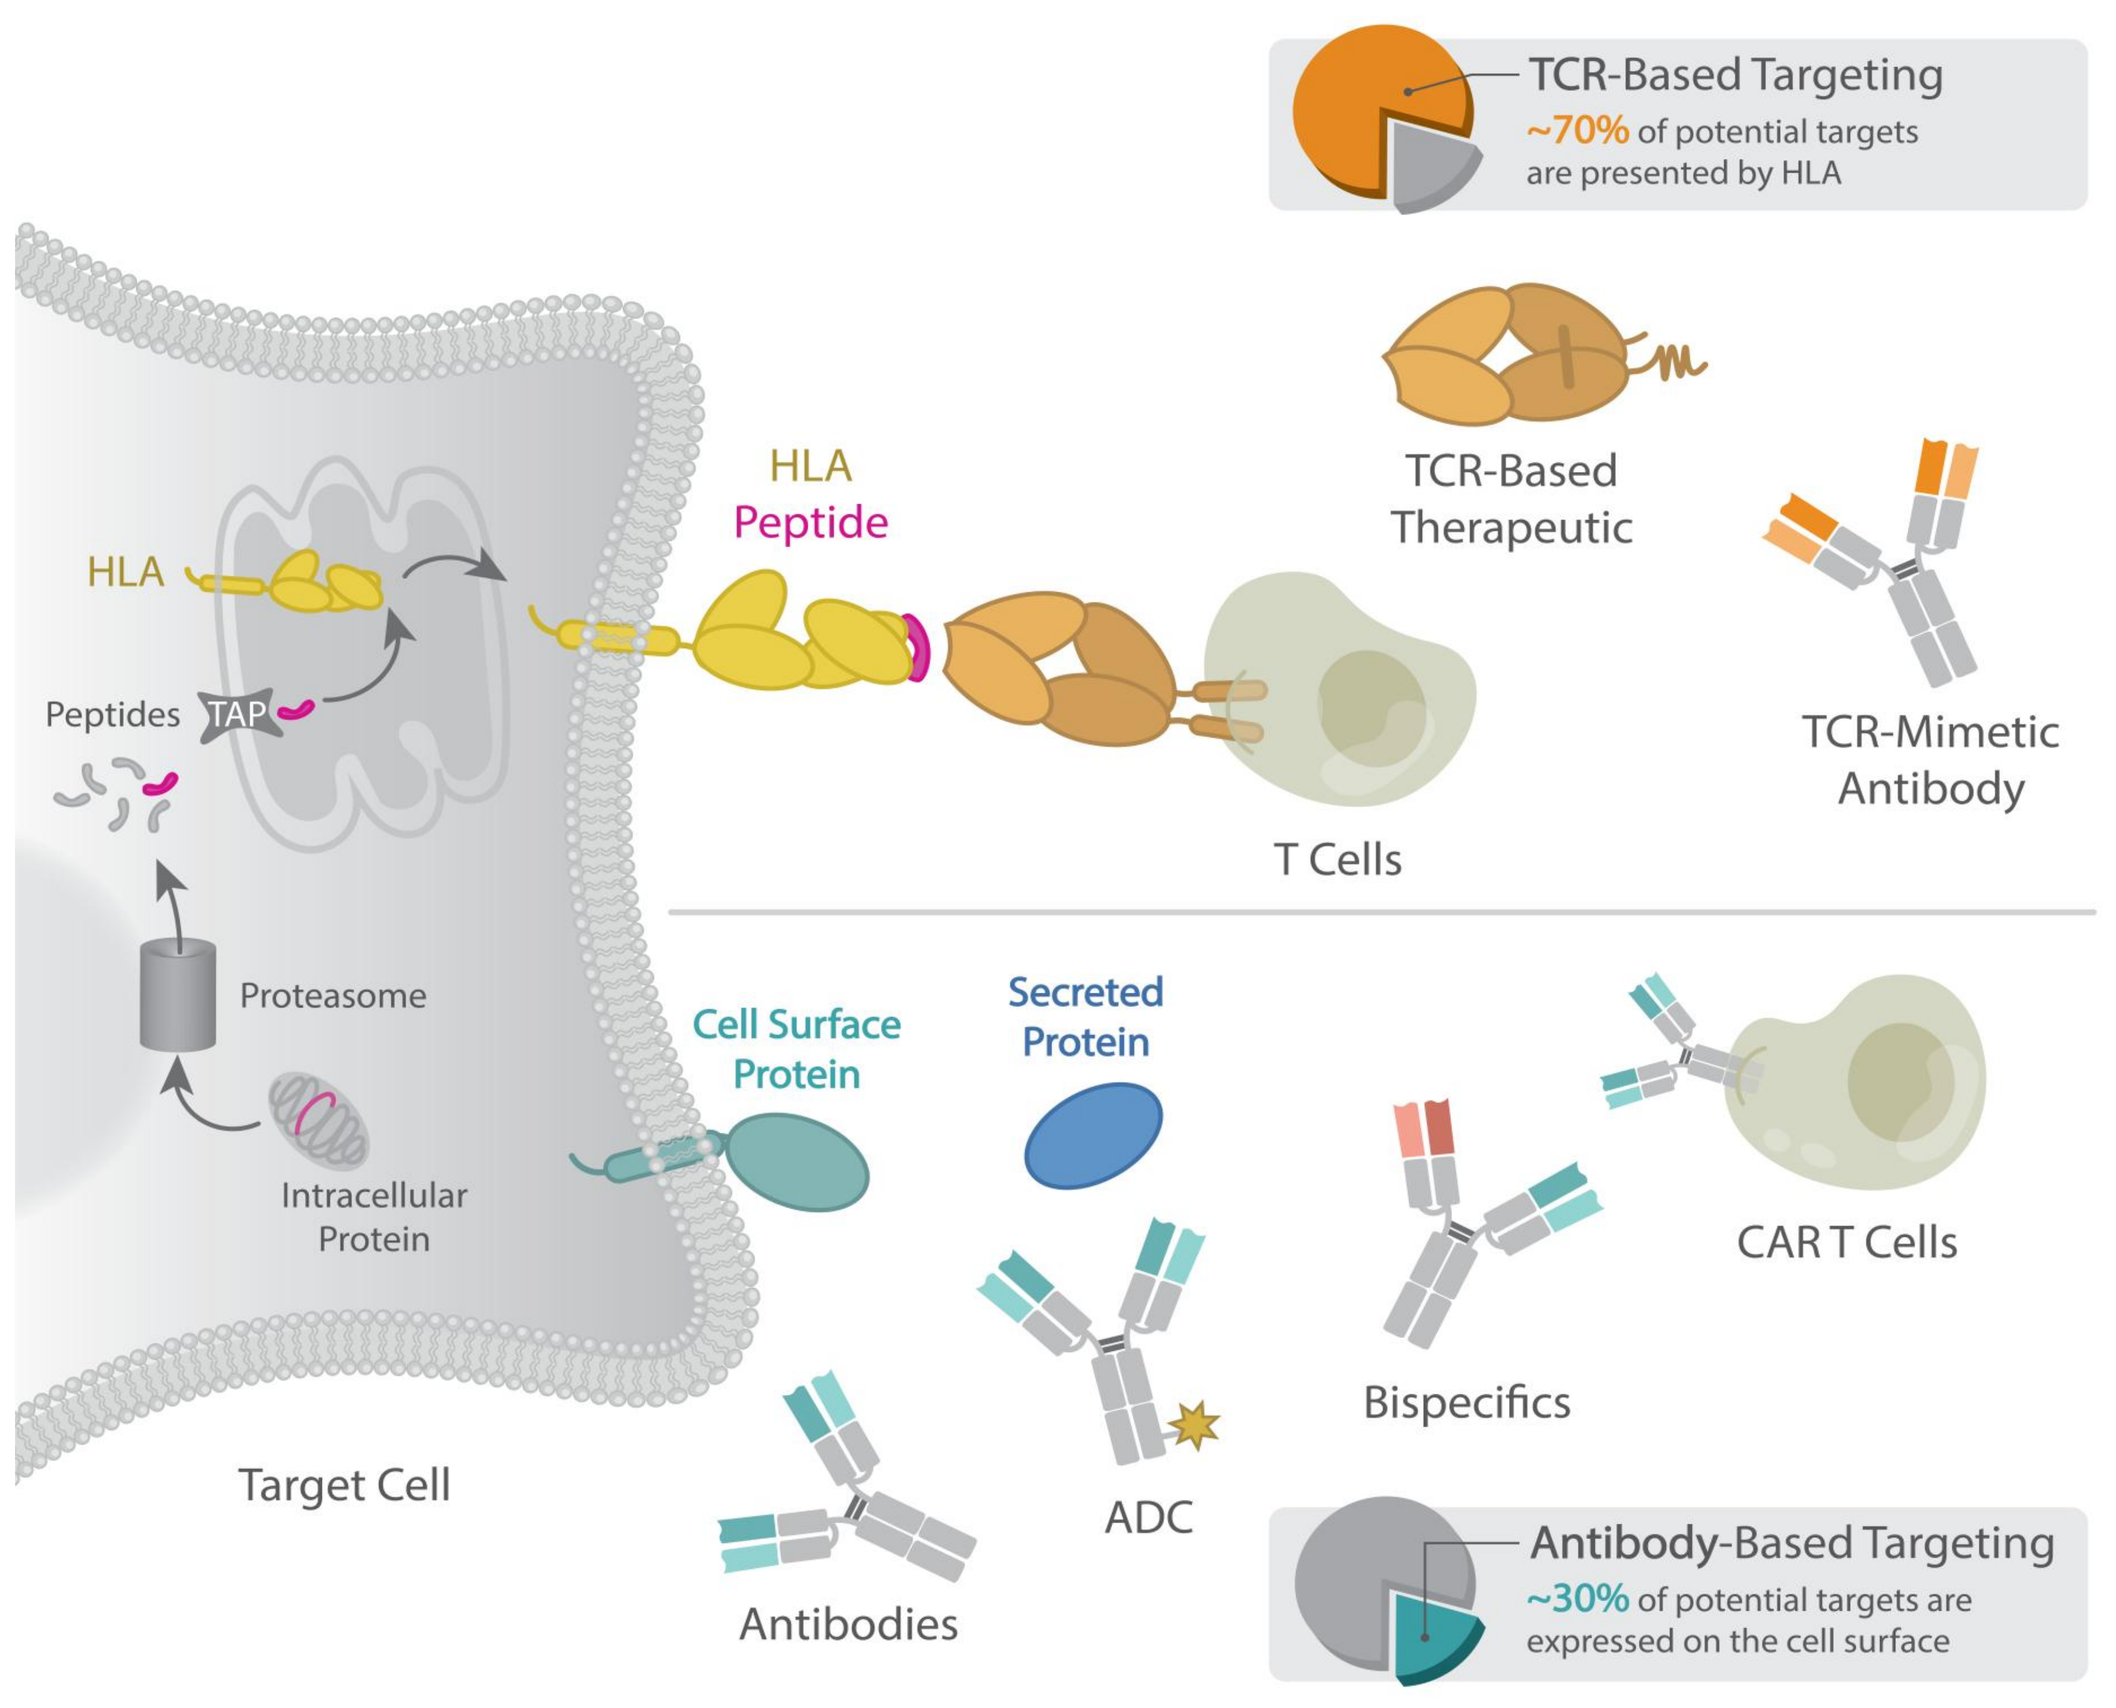

TCR-Based Targeting
~70% of potential targets are presented by HLA
HLA
Peptide
HLA
Peptides
TAP
Proteasome
Intracellular Protein
TCR-Based Therapeutic
TCR-Mimetic Antibody
T Cells
Cell Surface Protein
Secreted Protein
CAR T Cells
Bispecifics
ADC
Antibodies
Target Cell
Antibody-Based Targeting
~30% of potential targets are expressed on the cell surface

Cites data from Chandran, S.S. & Klebanoff, C.A. (2019) Immunol Rev

TCR-based T cell engagers

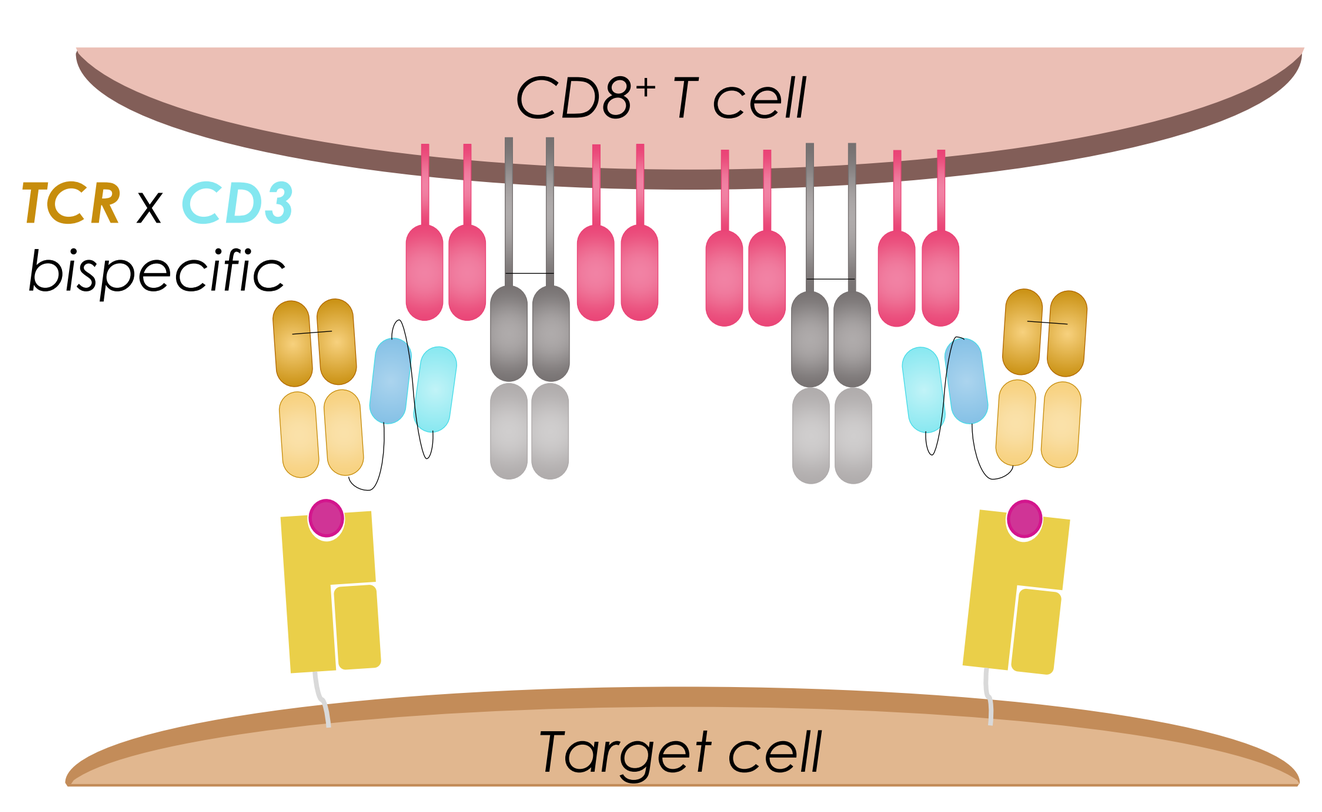

CD8+ T cell
TCR x CD3
bispecific
Target cell

TCR affinity drives TCE activity

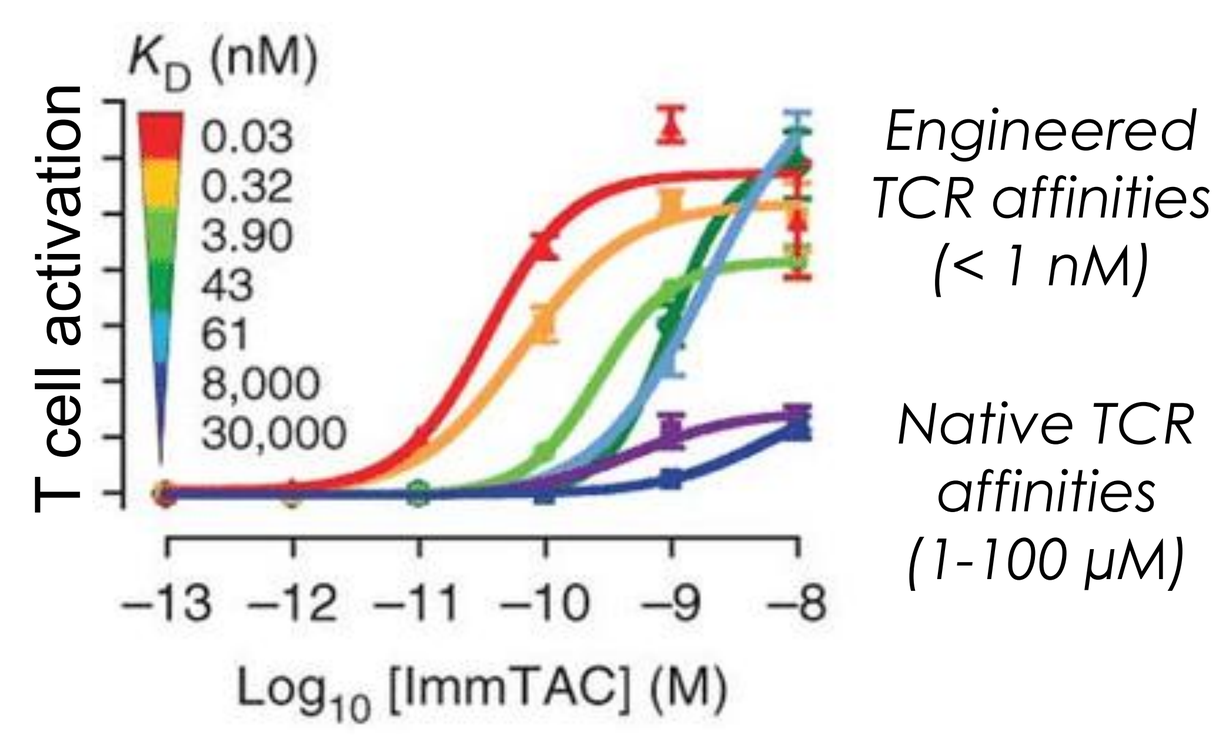

K_D (nM)
0.03
0.32
3.90
43
61
8,000
30,000
T cell activation
Engineered TCR affinities (< 1 nM)
Native TCR affinities (1-100 μM)
−13 −12 −11 −10 −9 −8
Log_{10} [ImmTAC] (M)

Adapted from Liddy, N. et al (2012) Nat Med

Soluble TCR-based T cell engagers enable potent and specific targeting of otherwise inaccessible intracellular antigens via peptide-HLA recognition and redirected T cell killing yet require extensive affinity maturation for function in clinically validated formats.

Background – Cancer-targeting 1G4 TCR

1G4 TCR recognition

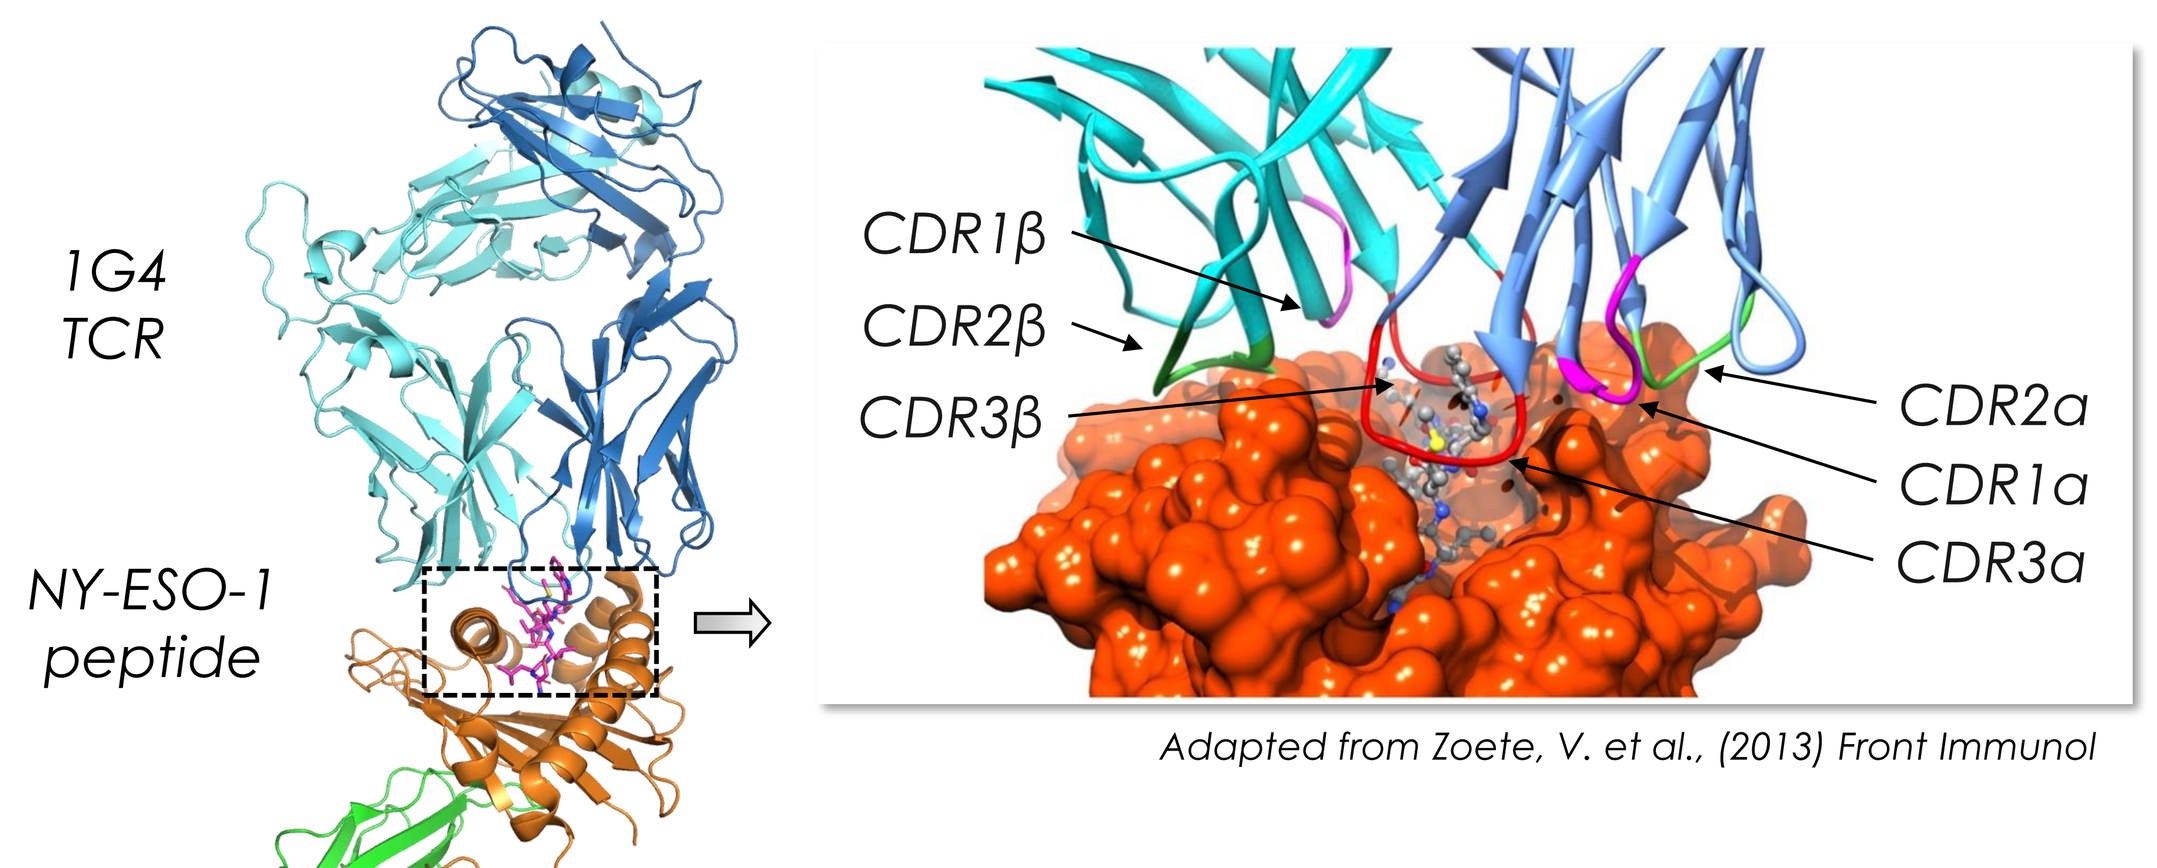

1G4 TCR
NY-ESO-1 peptide
HLA-A2
CDR1β
CDR2β
CDR3β
CDR2α
CDR1α
CDR3α

Adapted from Zoete, V. et al., (2013) Front Immunol

Chain	Germline	CDR1	CDR2	CDR3
Alpha	TRAV21	DSAIYN	IQSSQRE	AVRPTSGGSYIPT
Beta	TRBV6-5	MNHEY	SVGAGI	ASSYVGNTGELF

High-affinity variant in the clinic

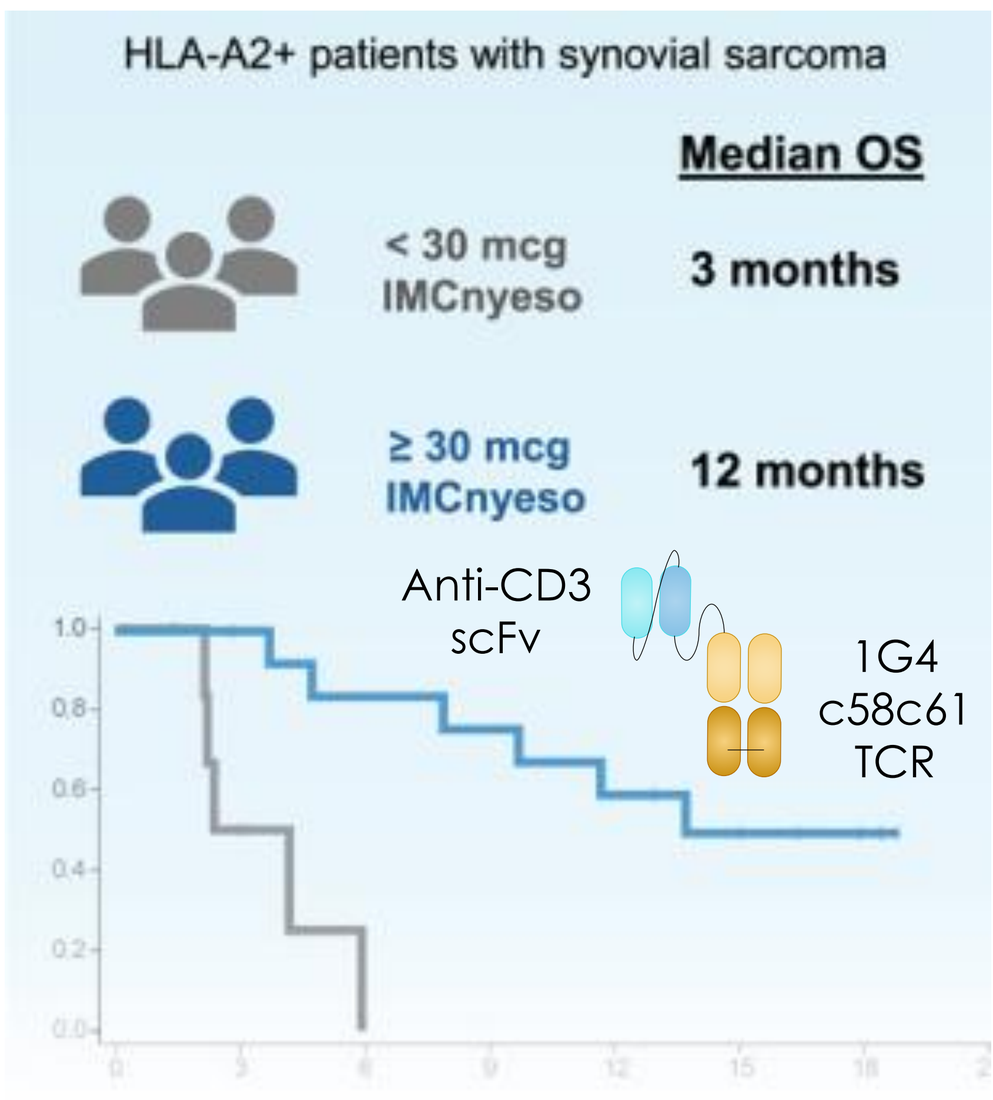

HLA-A2+ patients with synovial sarcoma
Median OS
< 30 mcg IMCnyeso
3 months
≥ 30 mcg IMCnyeso
12 months
Anti-CD3 scFv
1G4 c58c61 TCR

Adapted from Lopez, J.S. et al. (2025) Cell Rep Med

Patient-derived 1G4 TCR targets a cancer-restricted NY-ESO-1 peptide presented by HLA-A2 through specific interactions with its complementarity determining regions (CDRs) and has been affinity matured for use in a soluble TCR-based T cell engager currently in clinical trials.

Methods – Yeast-based TCR expression platform

TCR-CD3 complex / TCR-Fc / High-throughput yeast-based platform

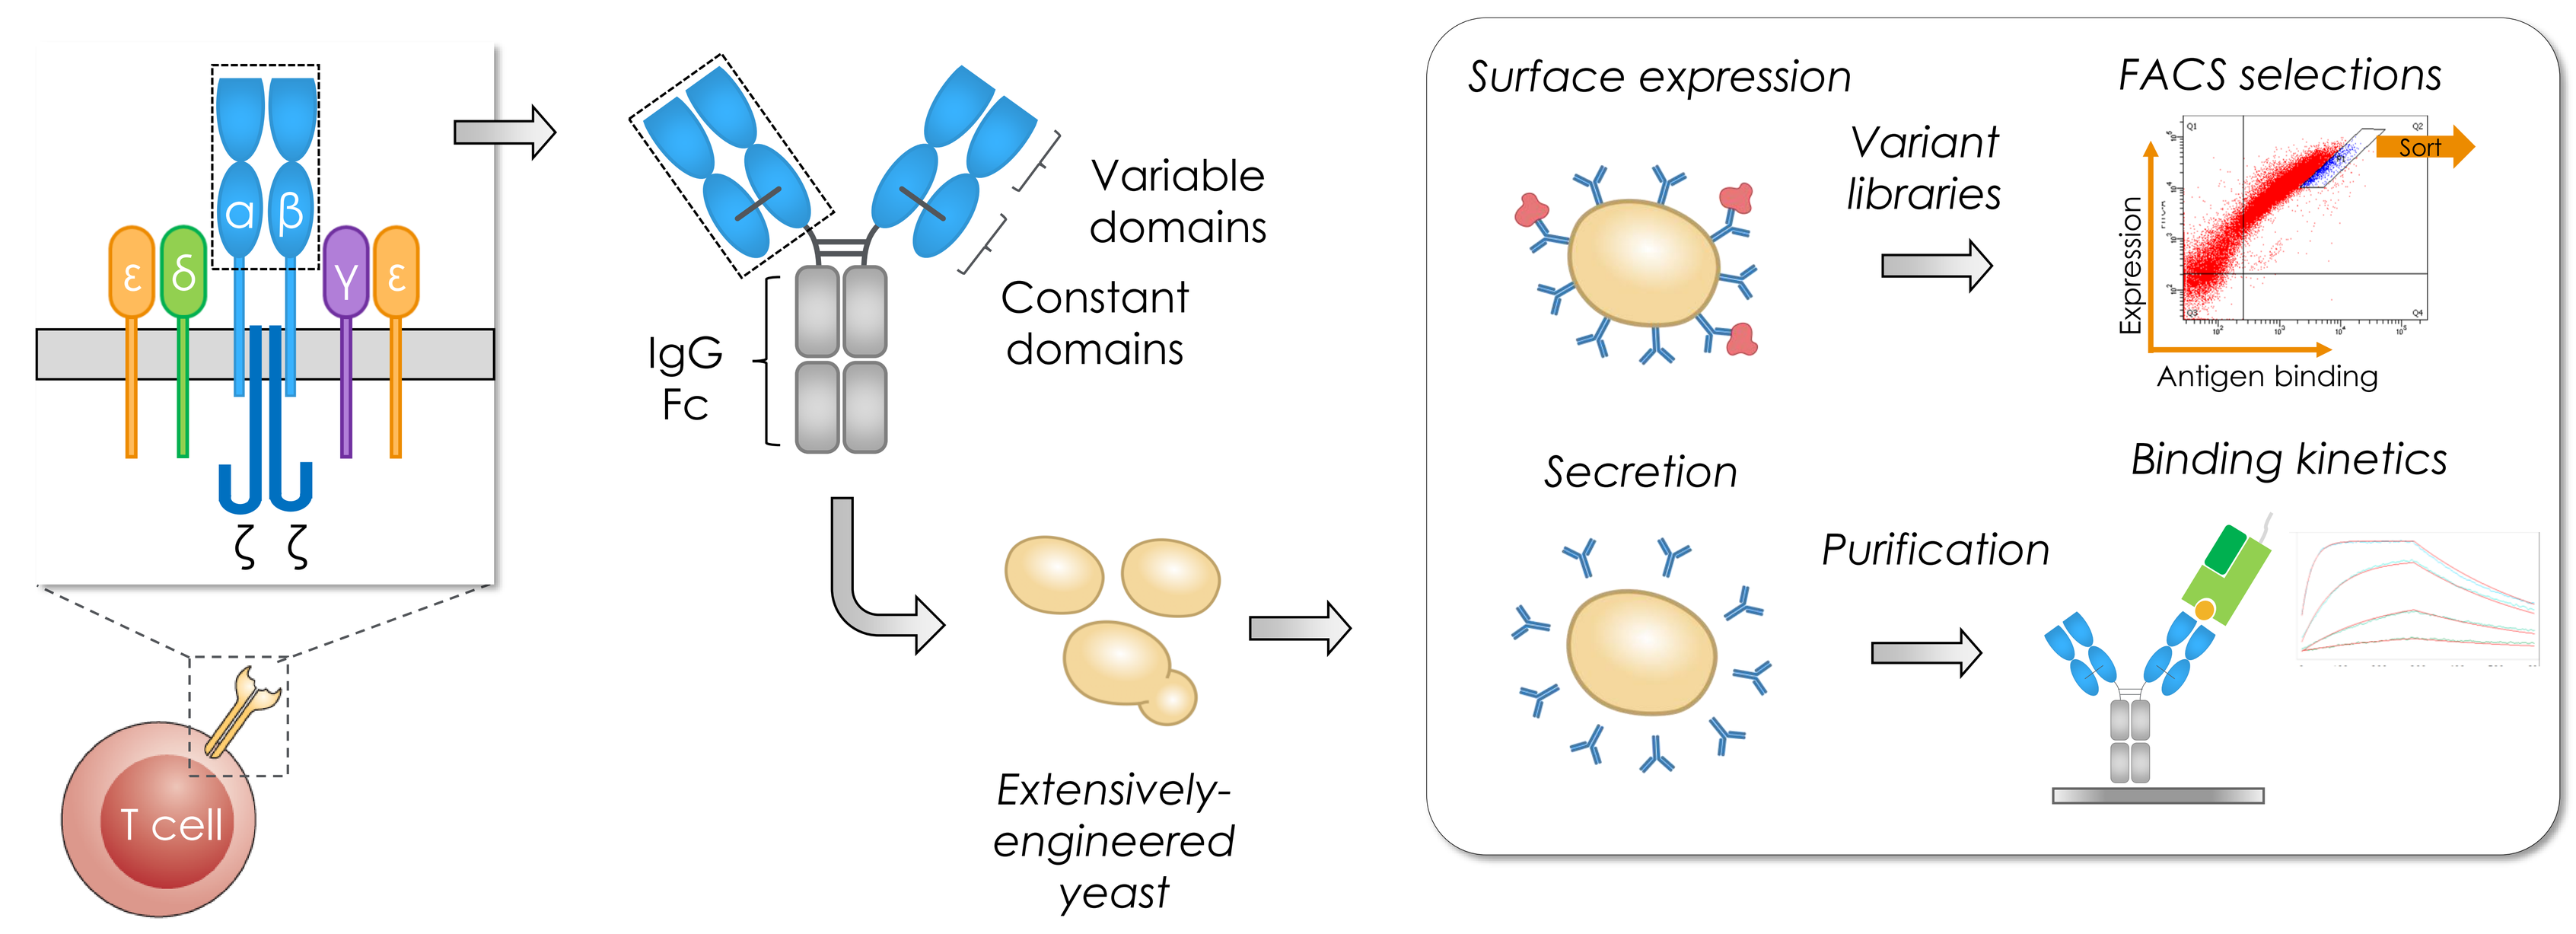

ε δ α β γ ε
ζ ζ
T cell
Variable domains
Constant domains
IgG Fc
Extensively-engineered yeast
Surface expression
Variant libraries
FACS selections
Sort
Expression
Antigen binding
Secretion
Purification
Binding kinetics

Yeast-based platform for high-throughput discovery and characterization of soluble TCRs expressed on the yeast surface or secreted in an Fc-linked bivalent format.

Methods – Affinity maturation of 1G4 TCR

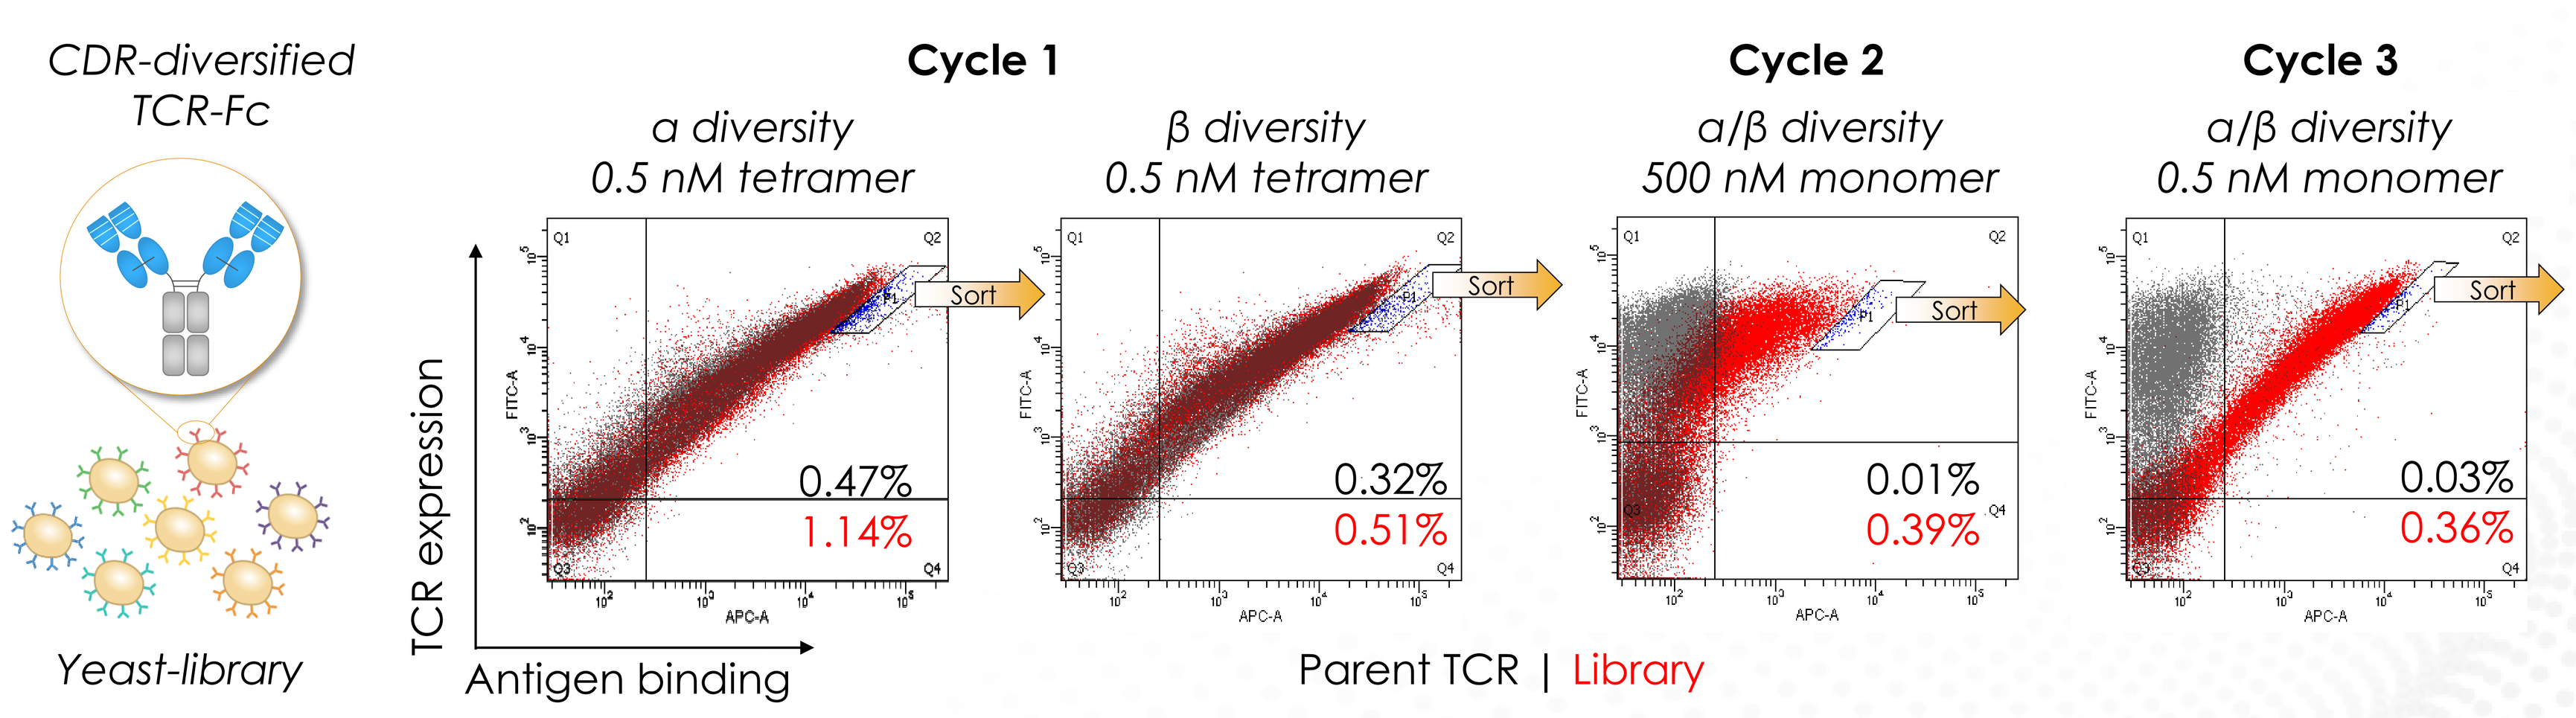

CDR-diversified TCR-Fc
Yeast-library
Cycle 1
α diversity
0.5 nM tetramer
β diversity
0.5 nM tetramer
Cycle 2
α/β diversity
500 nM monomer
Cycle 3
α/β diversity
0.5 nM monomer
Sort
0.47%
1.14%
0.32%
0.51%
0.01%
0.39%
0.03%
0.36%
TCR expression
Antigen binding
Parent TCR | Library

Affinity maturation of 1G4 TCR across iterative cycles of alpha- and beta-chain CDR diversification followed by FACS-based selections to enrich high-affinity TCR variants

Results – Minimally mutated high-affinity TCRs

Greatly enhanced affinity over parent / Minimally-mutated TCRs

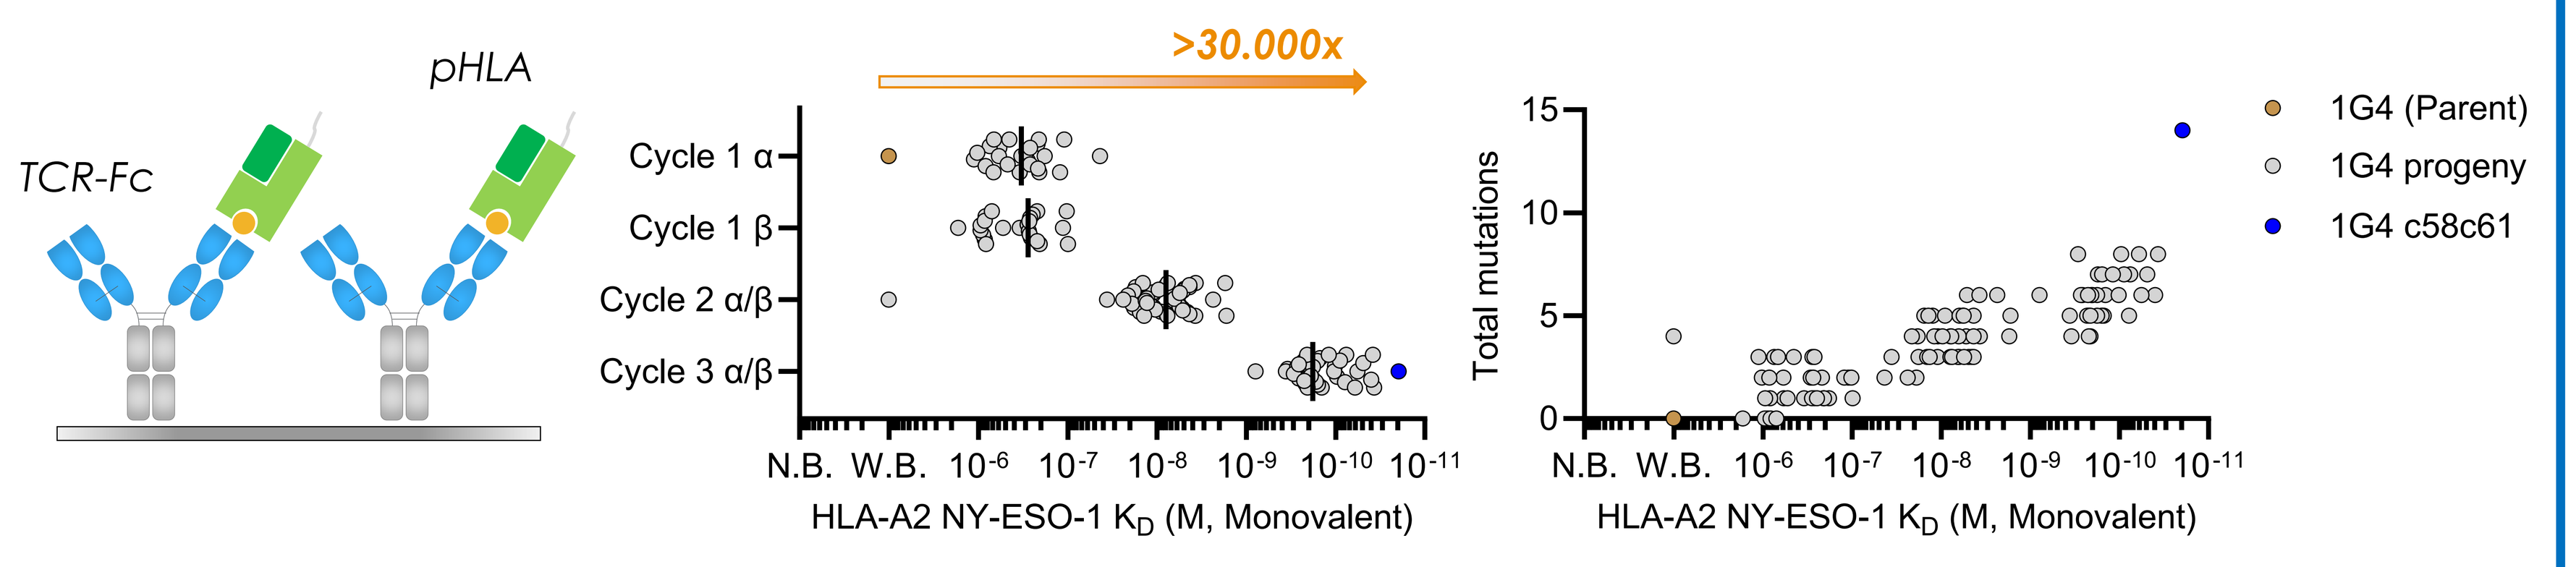

pHLA
TCR-Fc
>30.000x
Cycle 1 α
Cycle 1 β
Cycle 2 α/β
Cycle 3 α/β
N.B. W.B. 10^{-6} 10^{-7} 10^{-8} 10^{-9} 10^{-10} 10^{-11}
HLA-A2 NY-ESO-1 K_D (M, Monovalent)
Total mutations
1G4 (Parent)
1G4 progeny
1G4 c58c61

Top clones parallel high-affinity clinical control

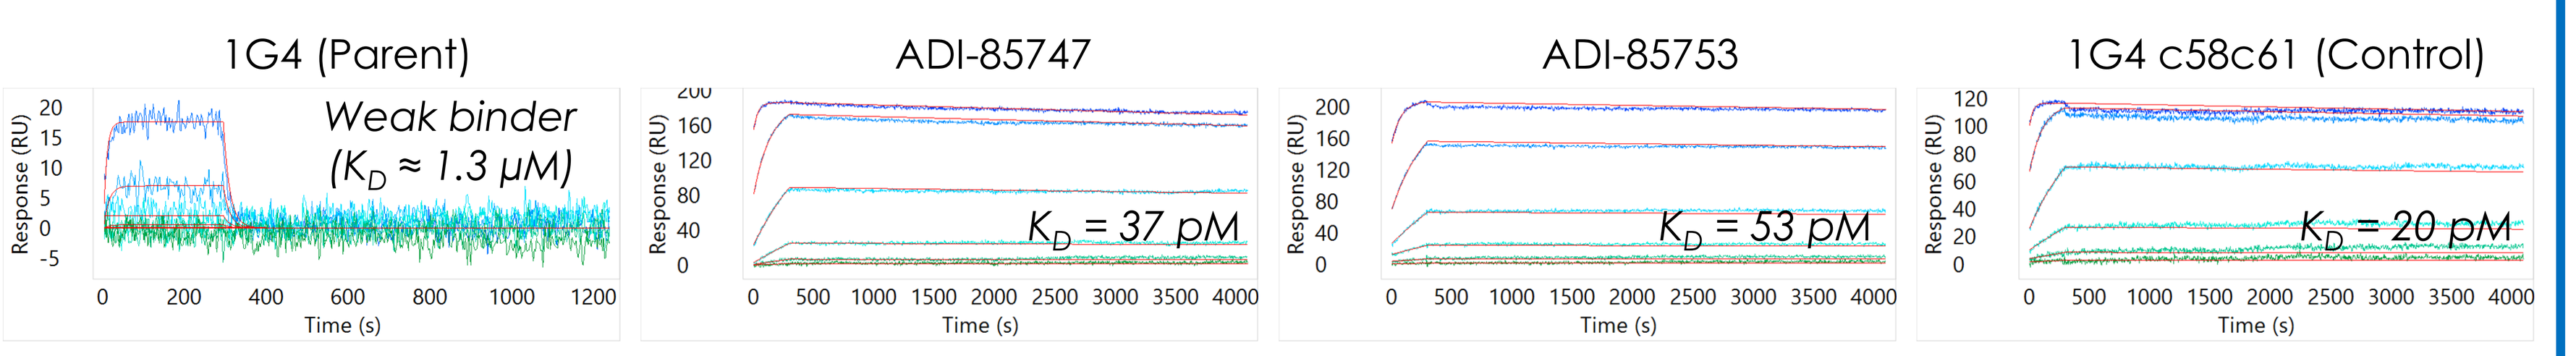

1G4 (Parent)
Weak binder (K_D ≈ 1.3 μM)
ADI-85747
K_D = 37 pM
ADI-85753
K_D = 53 pM
1G4 c58c61 (Control)
K_D = 20 pM
Response (RU)
Time (s)

1G4 TCR affinity maturation yields progeny with greatly enhanced affinities but far fewer mutations than a related high-affinity clinical control.

Results – Enhanced specificity over clinical control

High-affinity TCRs retain selective peptide recognition

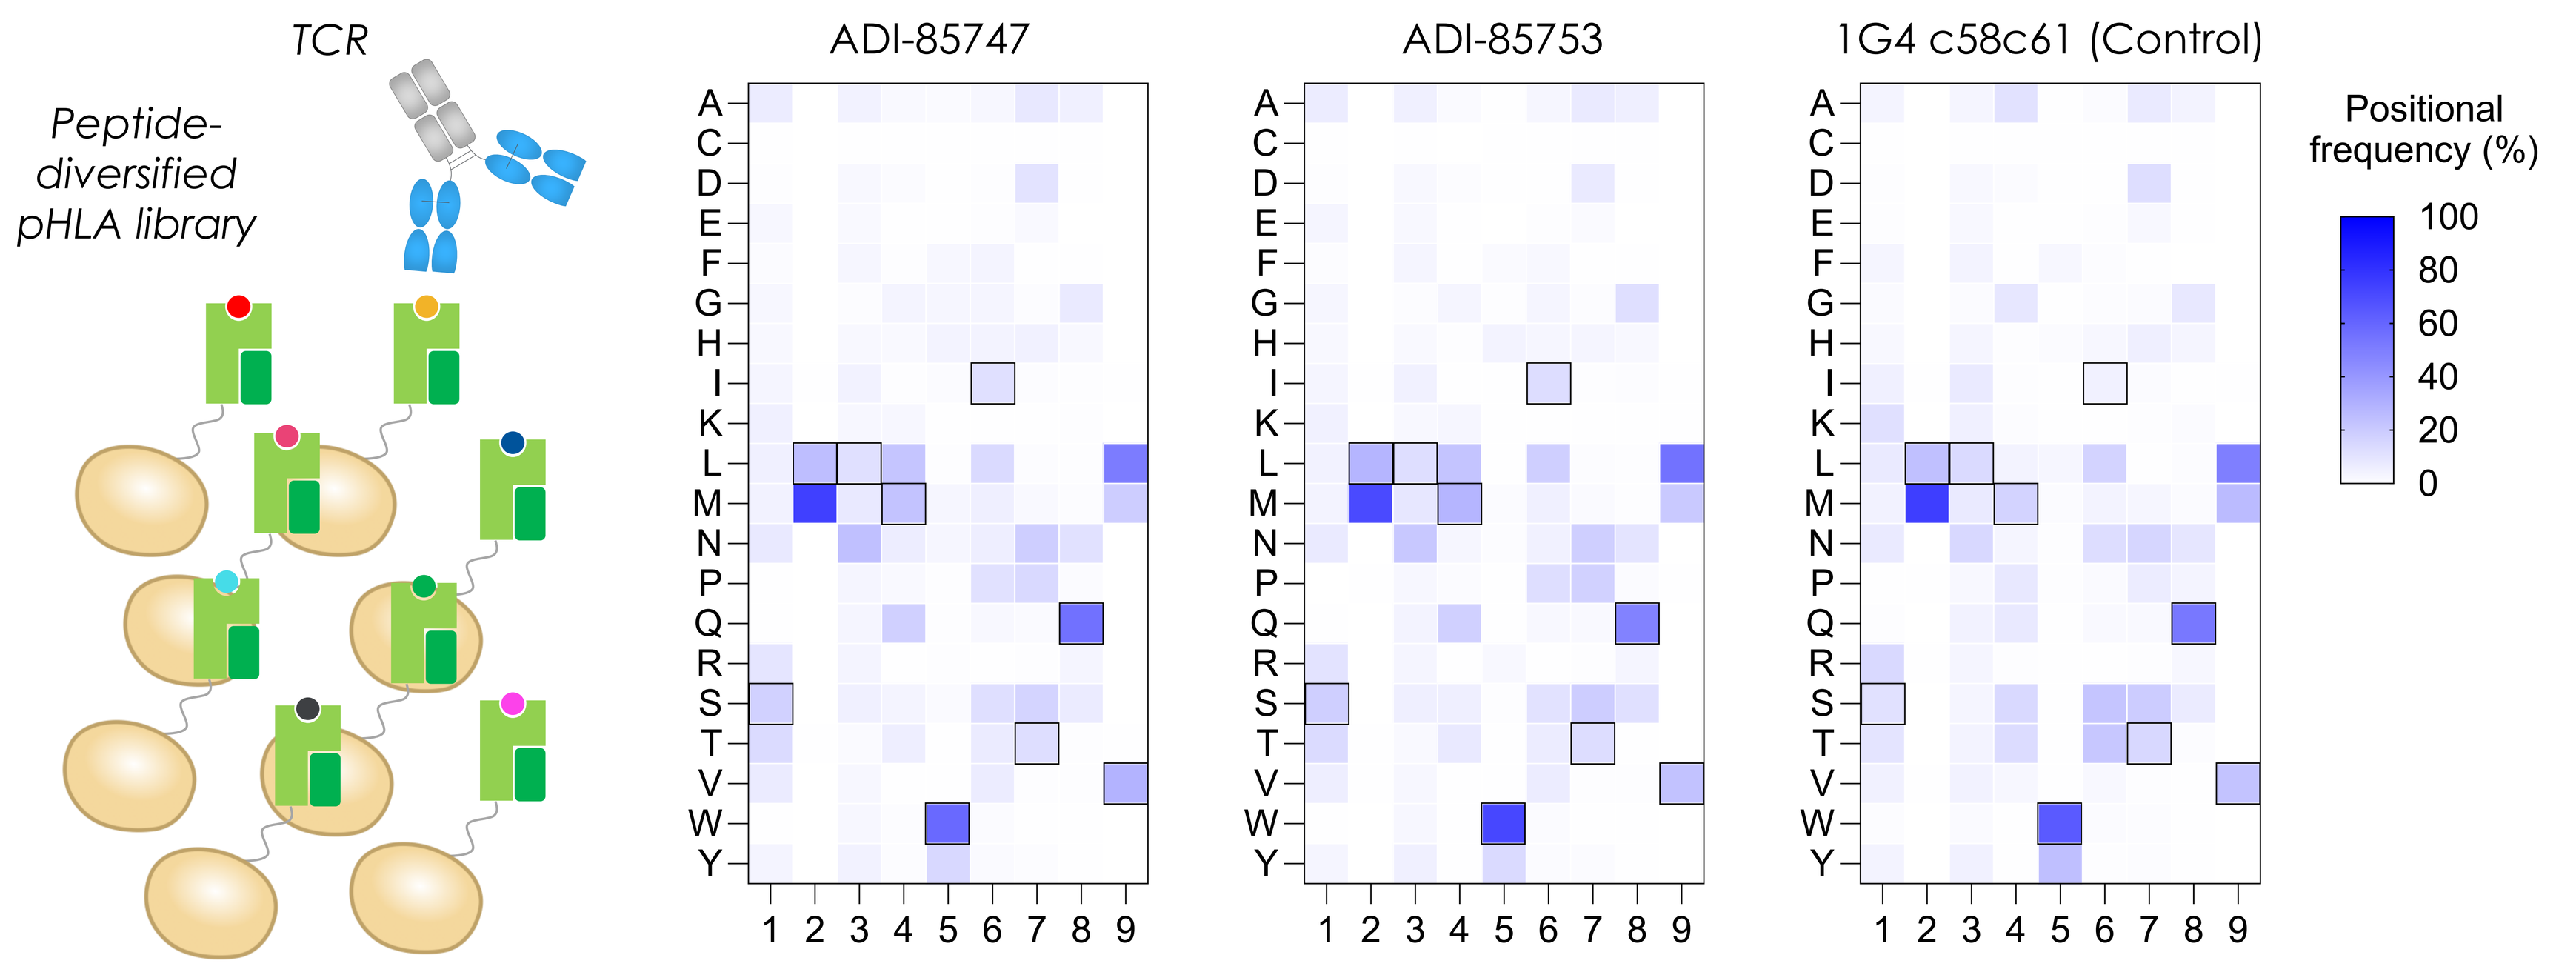

TCR
Peptide-diversified pHLA library
ADI-85747
ADI-85753
1G4 c58c61 (Control)
Positional frequency (%)

Library-derived TCRs exceed specificity of clinical control

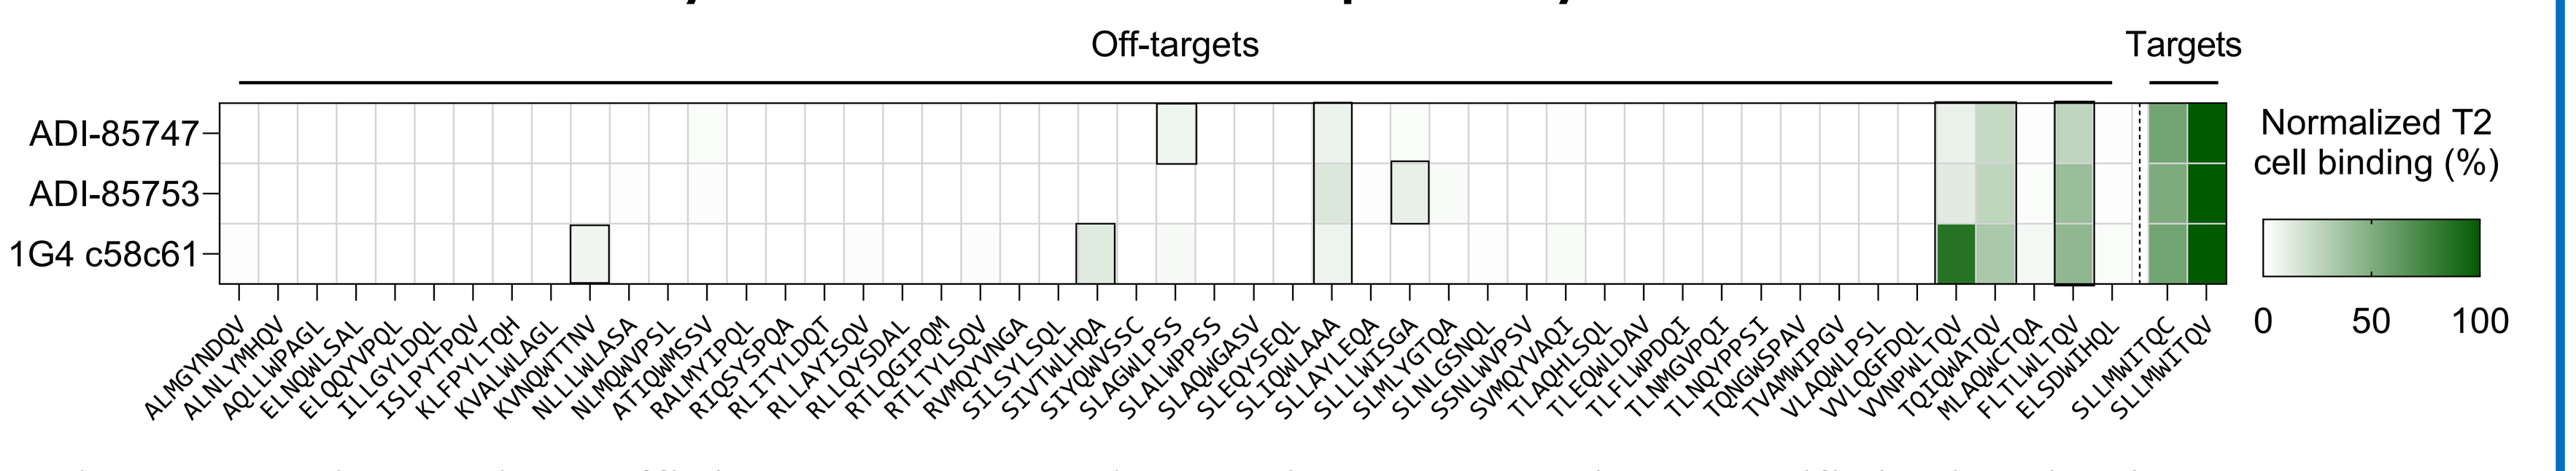

Off-targets
Targets
ADI-85747
ADI-85753
1G4 c58c61
Normalized T2 cell binding (%)
0 50 100

Library-derived high-affinity 1G4 TCR variants display superior specificity in binding to a panel of human off-target peptides predicted by pHLA-library-derived recognition motifs.

Results – Potent TCE-mediated target cell killing

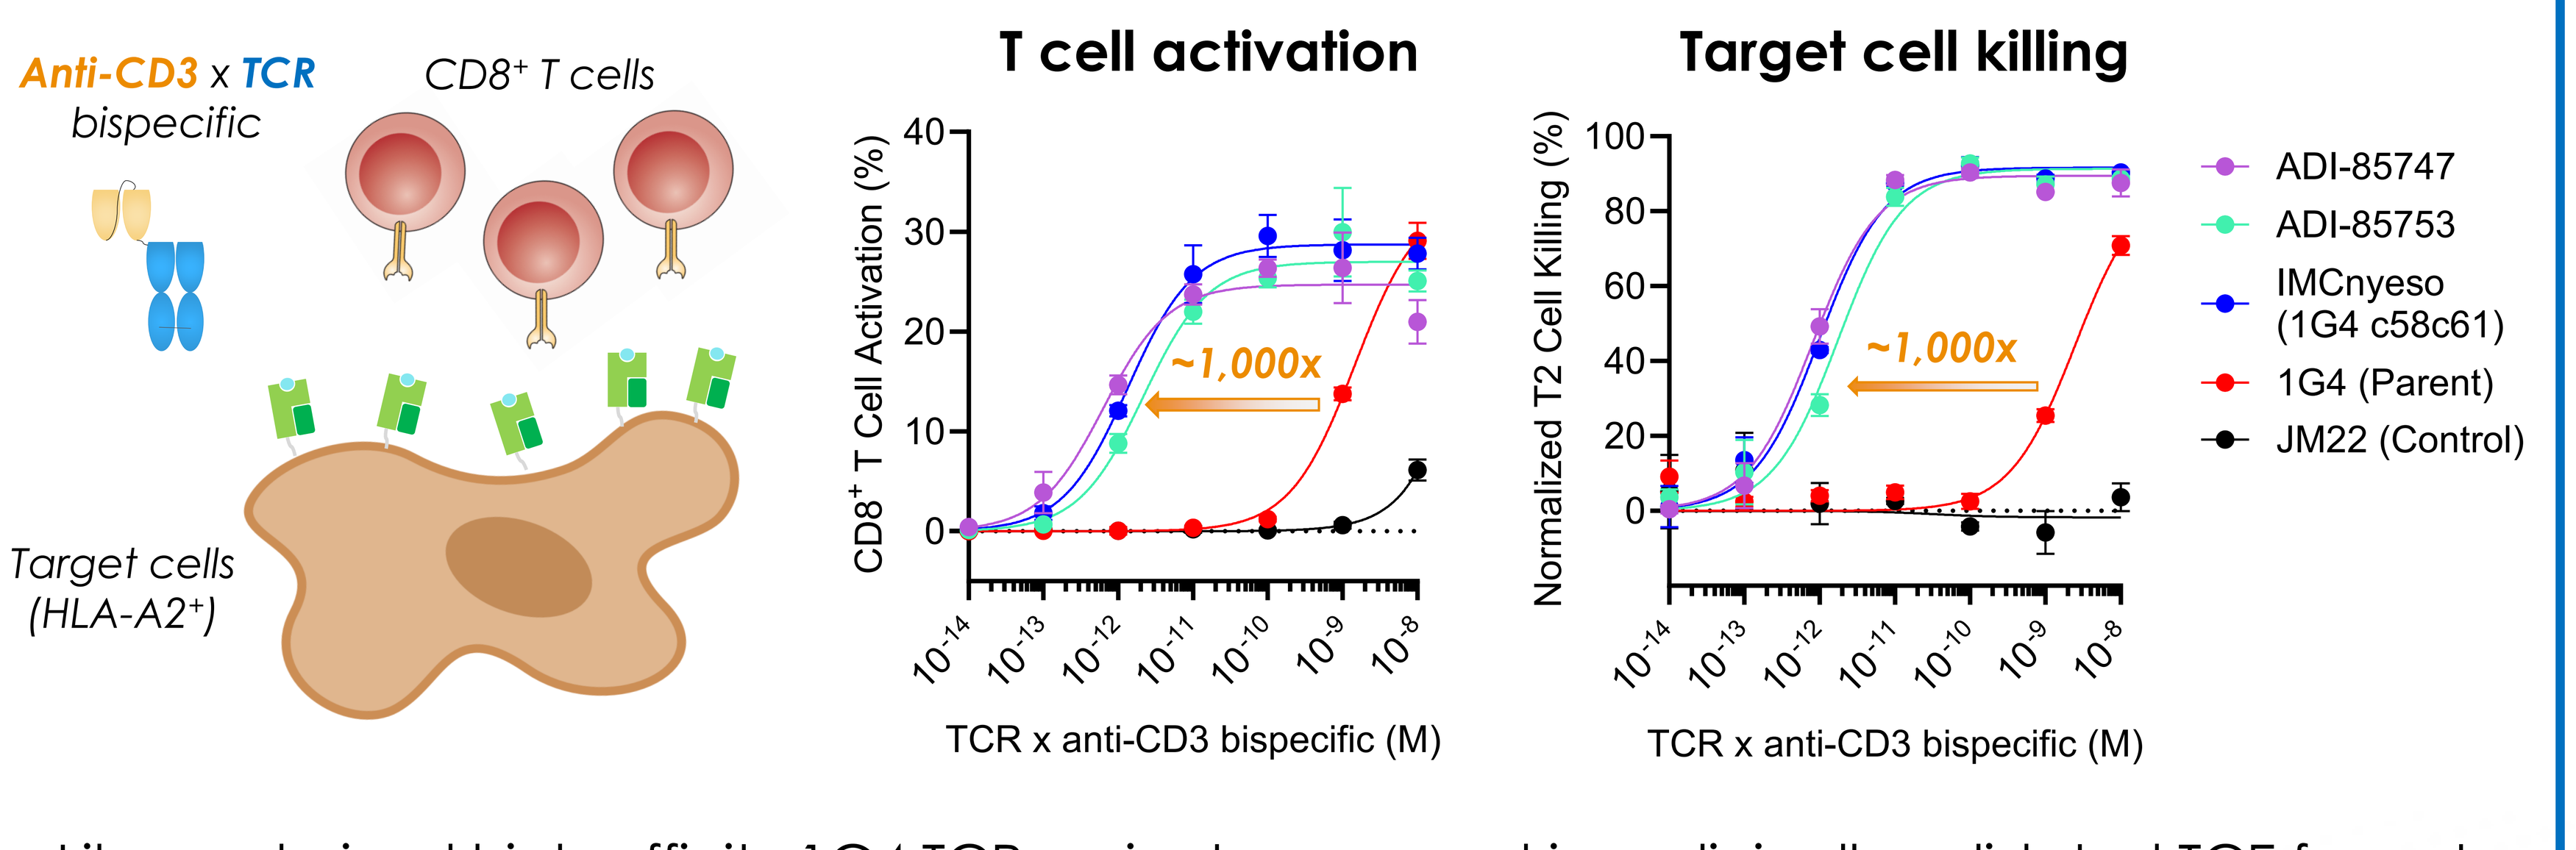

Anti-CD3 x TCR bispecific
CD8+ T cells
Target cells (HLA-A2+)
T cell activation
Target cell killing
CD8+ T Cell Activation (%)
Normalized T2 Cell Killing (%)
~1,000x
TCR x anti-CD3 bispecific (M)
ADI-85747
ADI-85753
IMCnyeso (1G4 c58c61)
1G4 (Parent)
JM22 (Control)

Library-derived high-affinity 1G4 TCR variants expressed in a clinically validated TCE format mediate highly-potent T cell activation and target cell killing equivalent to a clinical control.

Summary

- Adimab's high-throughput yeast-based platform enables rapid generation and characterization of high-affinity soluble TCR for use in pHLA-targeting therapeutics.
- Affinity maturation of a cancer-targeting TCR yielded variants with >30,000-fold improved affinities and far fewer mutations than a high-affinity clinical control.
- Library-derived high-affinity TCR variants exhibited highly specific peptide binding, with fewer human off-target interactions than a related clinical control.
- Library-derived high-affinity TCR variants potently induced target cell killing equivalent to a clinical control when reformatted into a clinically validated TCE format.

Acknowledgements

We thank the members of Adimab's High Throughput Expression Yeast, Mammalian, and CORE teams for their contributions to this work.

Scan for downloadable version