HIGH-THROUGHPUT DISCOVERY OF RARE ANTI-TUMOR TCRs VIA SYNTHETIC YEAST-BASED LIBRARIES

Joseph Brouillard, Tse-Han Kuo, Vivien Qiao, Akila Katuwawala, Nicholas Luey, Jonas Spaulding, Mrunal Sakharkar, Patricia Sackett, Whitney Renaud, Morgan Morrill, Allie LeMay, Hannah Green, John Horangic, Shu Lin, Beth Sharkey, Christin Strong, Elizabeth Parker, Tushar Jain, James Geoghegan, Arvind Sivasubramanian, Eric Krauland, C. Garrett Rappazzo

ADIMAB

BACKGROUND – TCR-BASED T CELL ENGAGERS

pHLA-targeting therapeutics

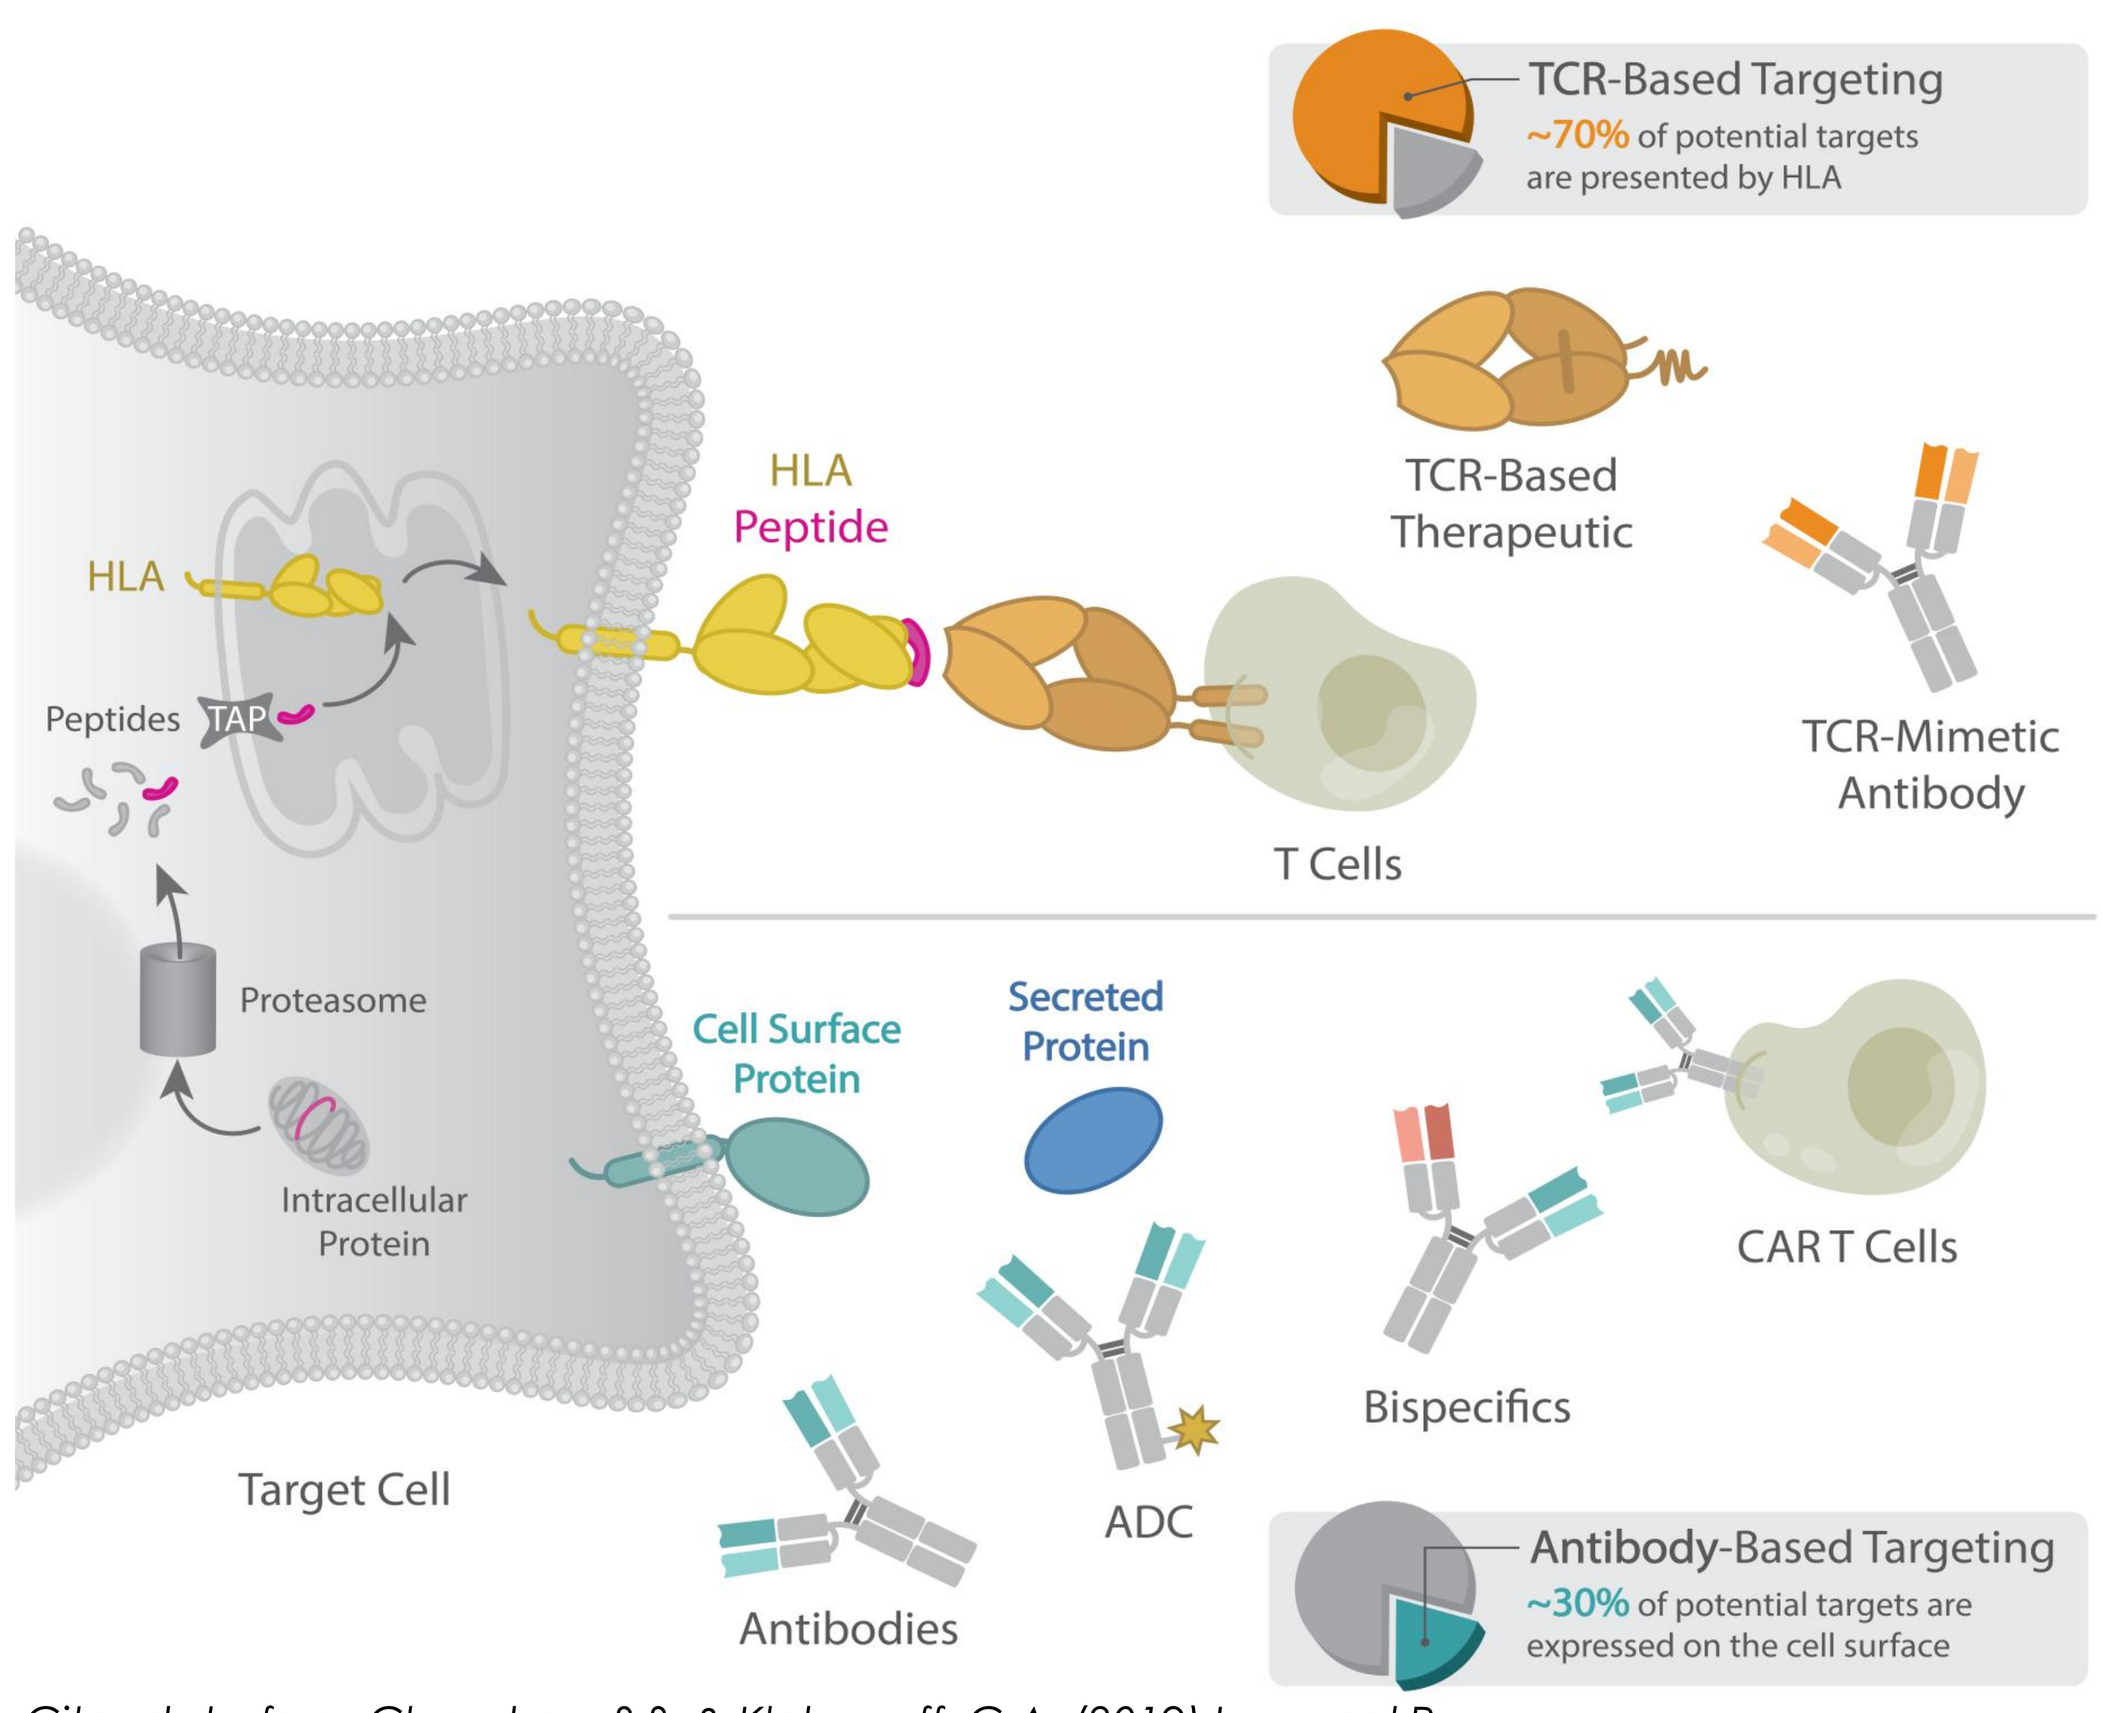
TCR-Based Targeting
~70% of potential targets are presented by HLA
HLA
Peptide
HLA
Peptides
TAP
TCR-Based Therapeutic
TCR-Mimetic Antibody
T Cells
Proteasome
Intracellular Protein
Cell Surface Protein
Secreted Protein
CAR T Cells
Bispecifics
ADC
Antibodies
Target Cell
Antibody-Based Targeting
~30% of potential targets are expressed on the cell surface

Cites data from Chandran, S.S. & Klebanoff, C.A. (2019) Immunol Rev

TCR-based T Cell Engagers

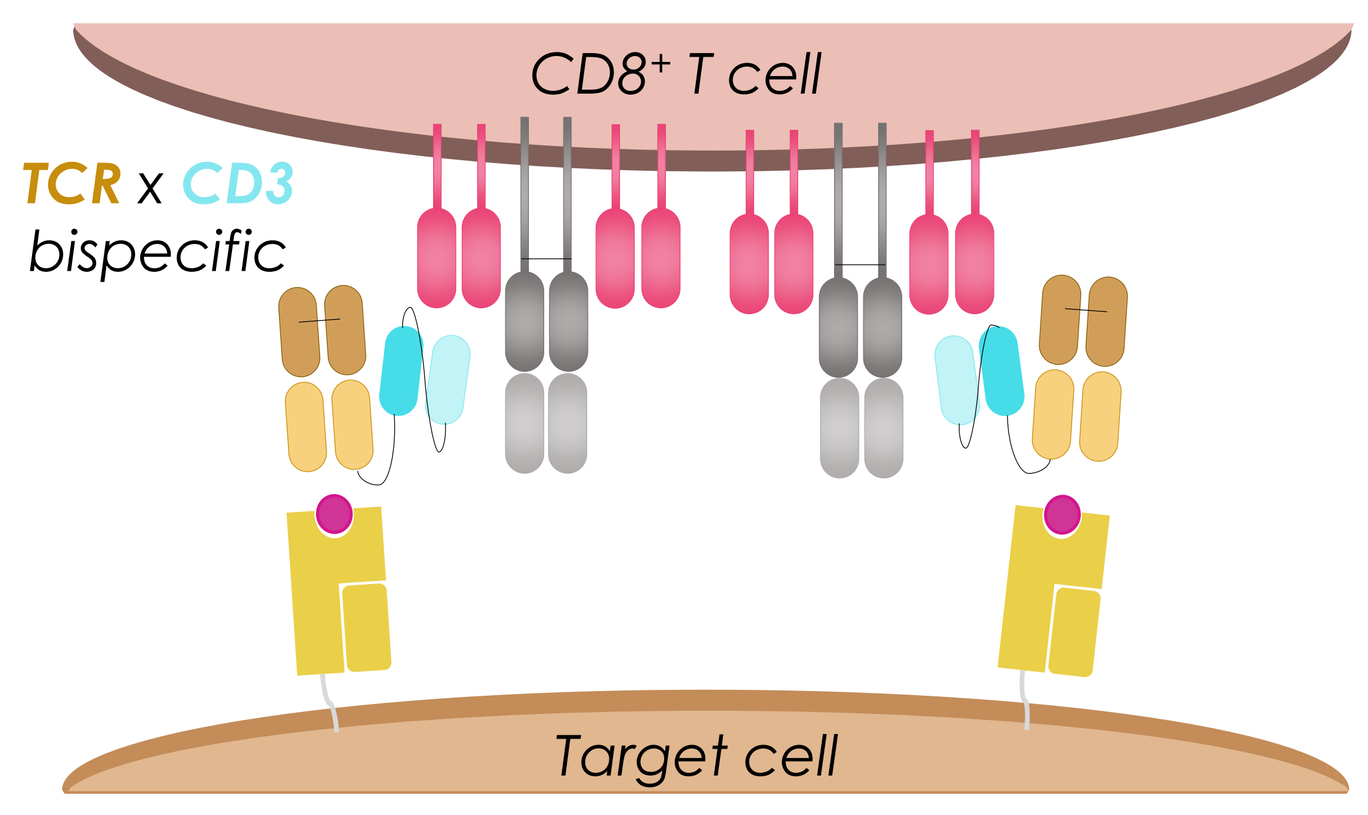
CD8+ T cell
TCR x CD3 bispecific
Target cell

Soluble TCR-based therapeutics

✓ *Evolutionarily-engrained specificity*

✓ *Approved pHLA-targeting benchmark*

★ ***Affinity and expression remain challenges***

Soluble TCR-based T cell engagers enable potent and specific targeting of otherwise inaccessible intracellular antigens via peptide-HLA recognition and redirected T cell killing, yet low affinity and poor expression complicate their development.

BACKGROUND – MULTI-SPECIFIC ANTI-TUMOR TCRs

Structural overview of MEL5 TCR multi-specificity

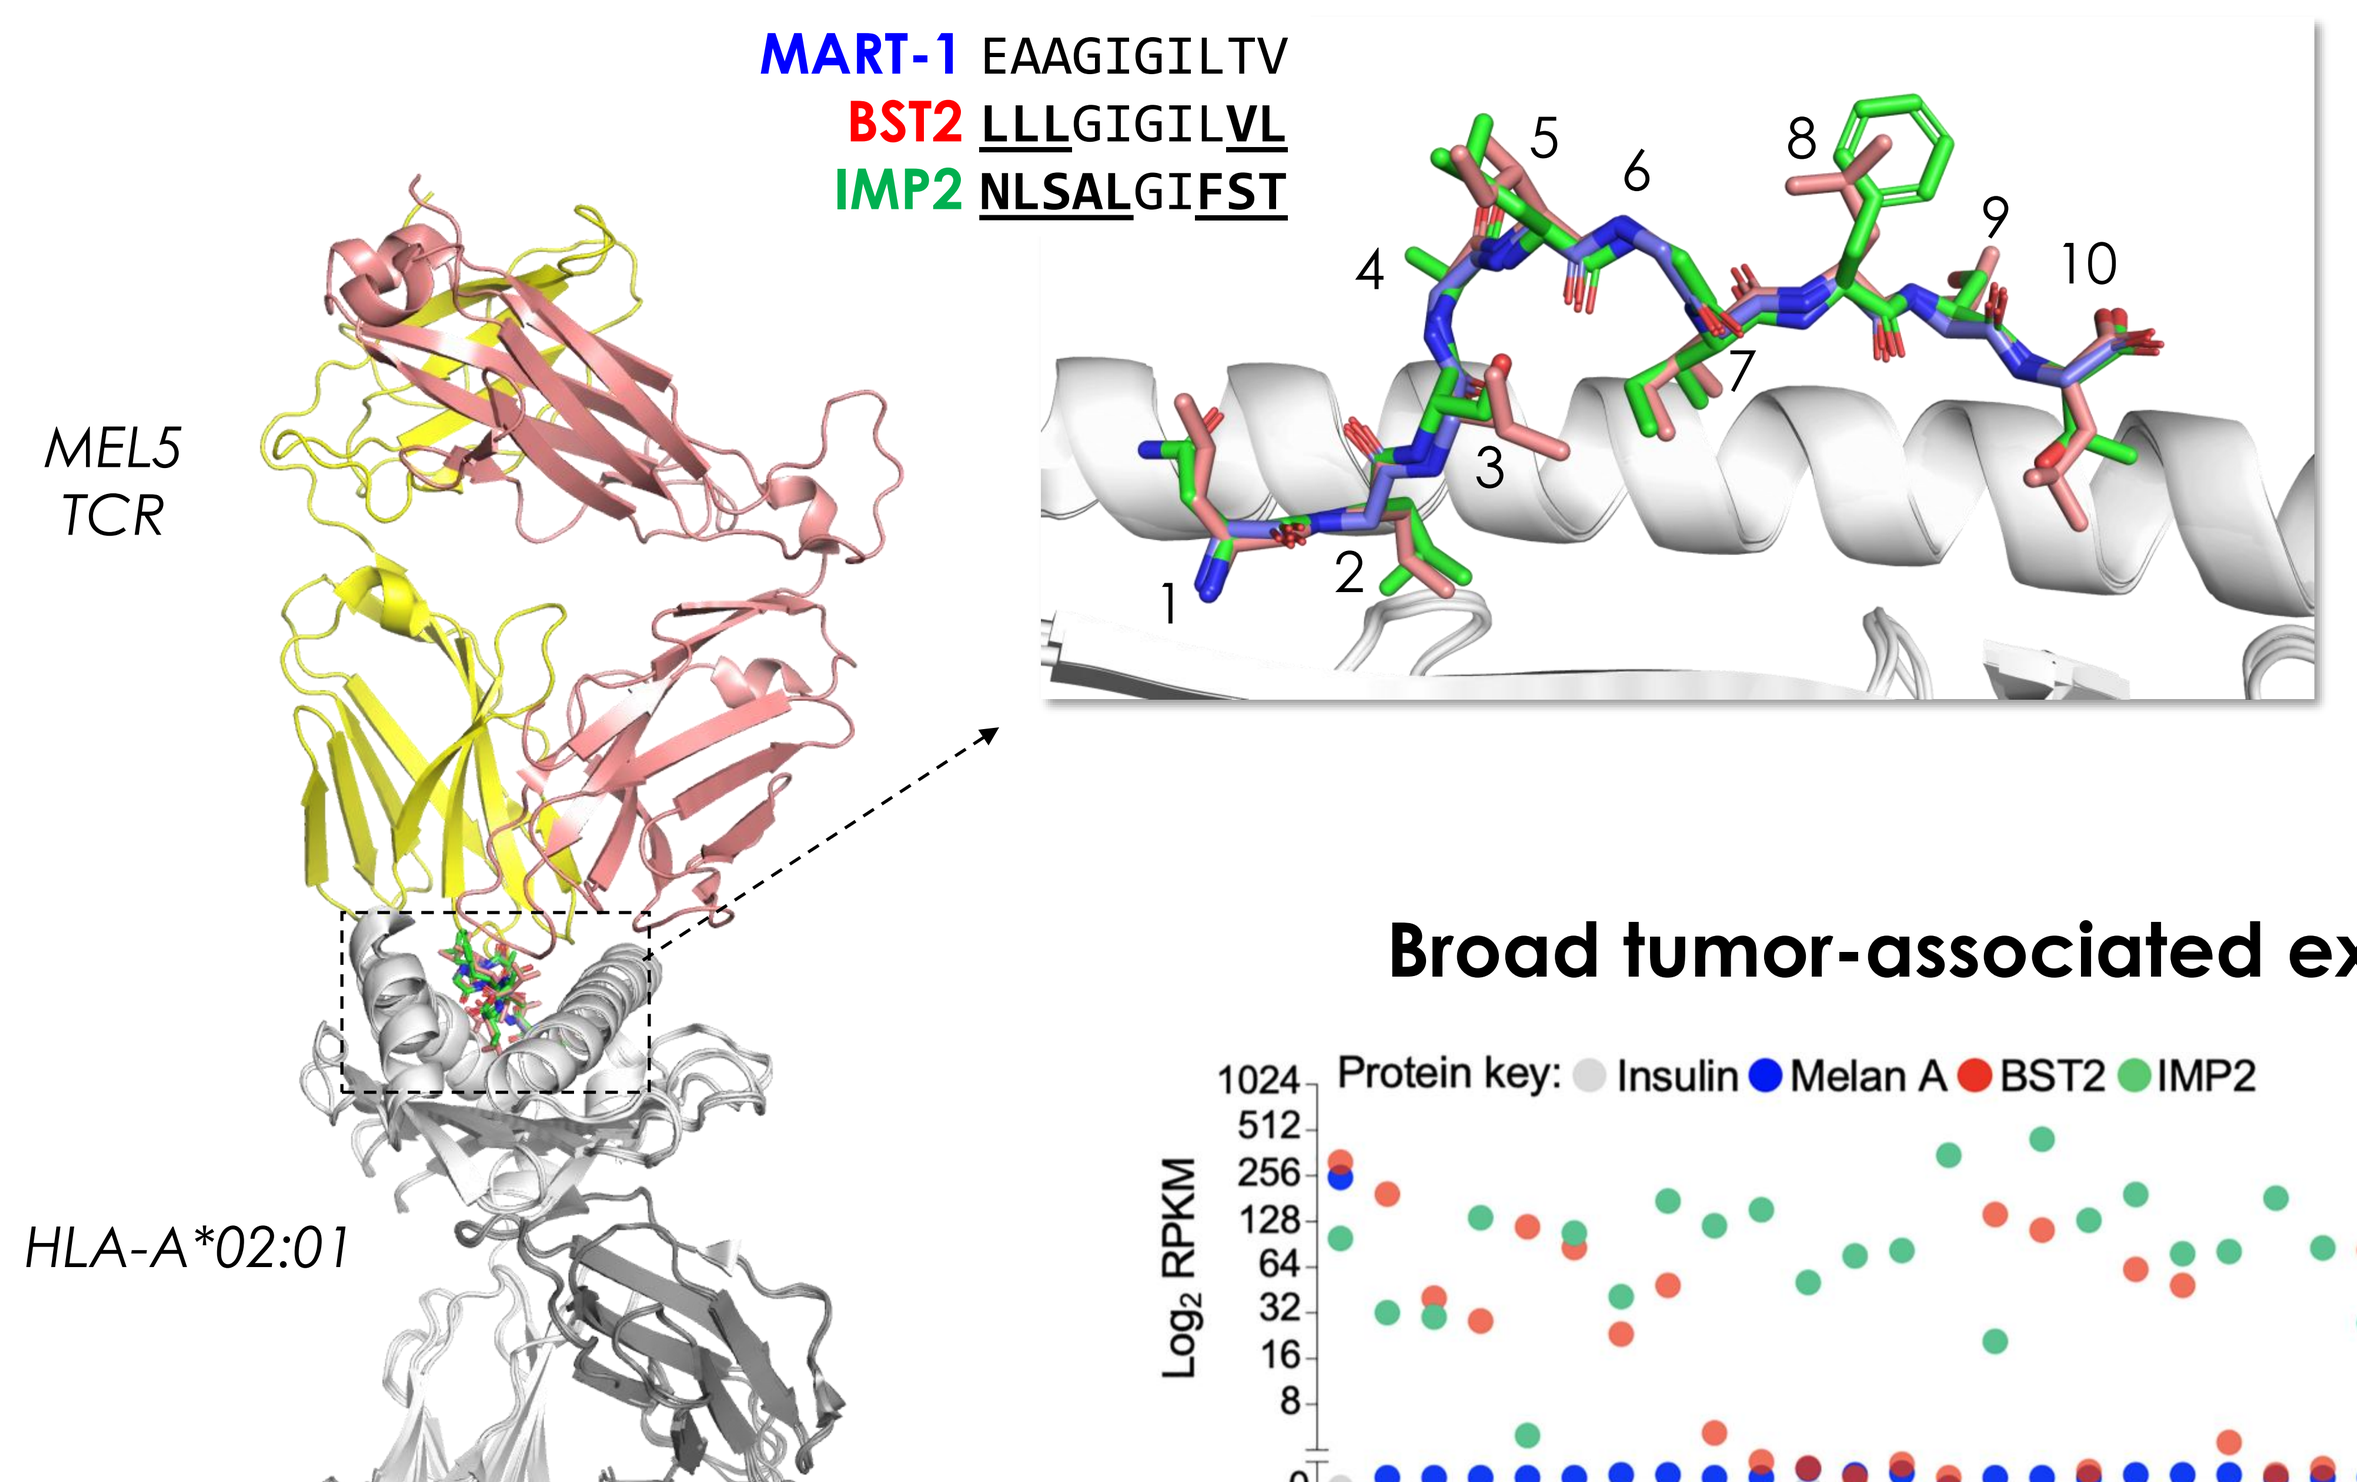
MART-1 EAAGIGILTV
BST2 LLLGIGILVL
IMP2 NLSALGIFST
1 2 3 4 5 6 7 8 9 10
MEL5 TCR
HLA-A*02:01
B2M

PDB: 4QQK, 7Q99, 7Q9A

Weak multi-specific binding

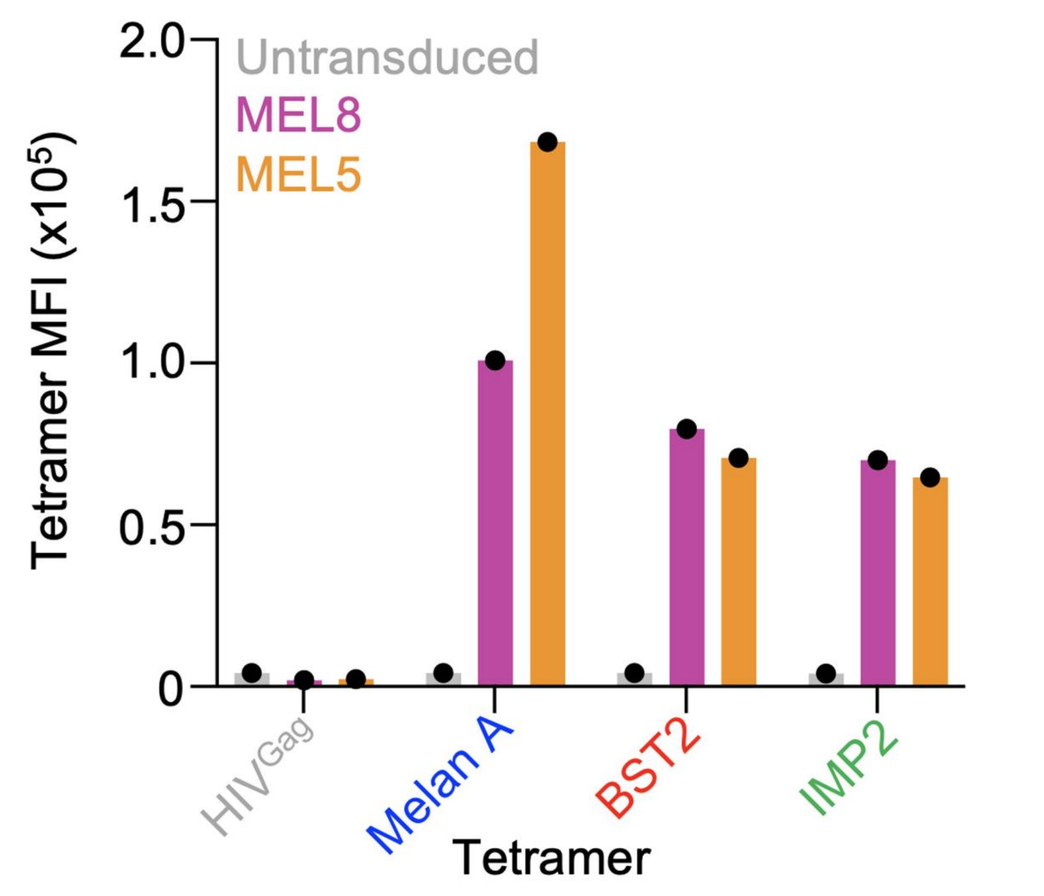
Untransduced
MEL8
MEL5
Tetramer MFI (x10^5)
2.0
1.5
1.0
0.5
0
HIVGag
Melan A
BST2
IMP2
Tetramer

Broad tumor-associated expression

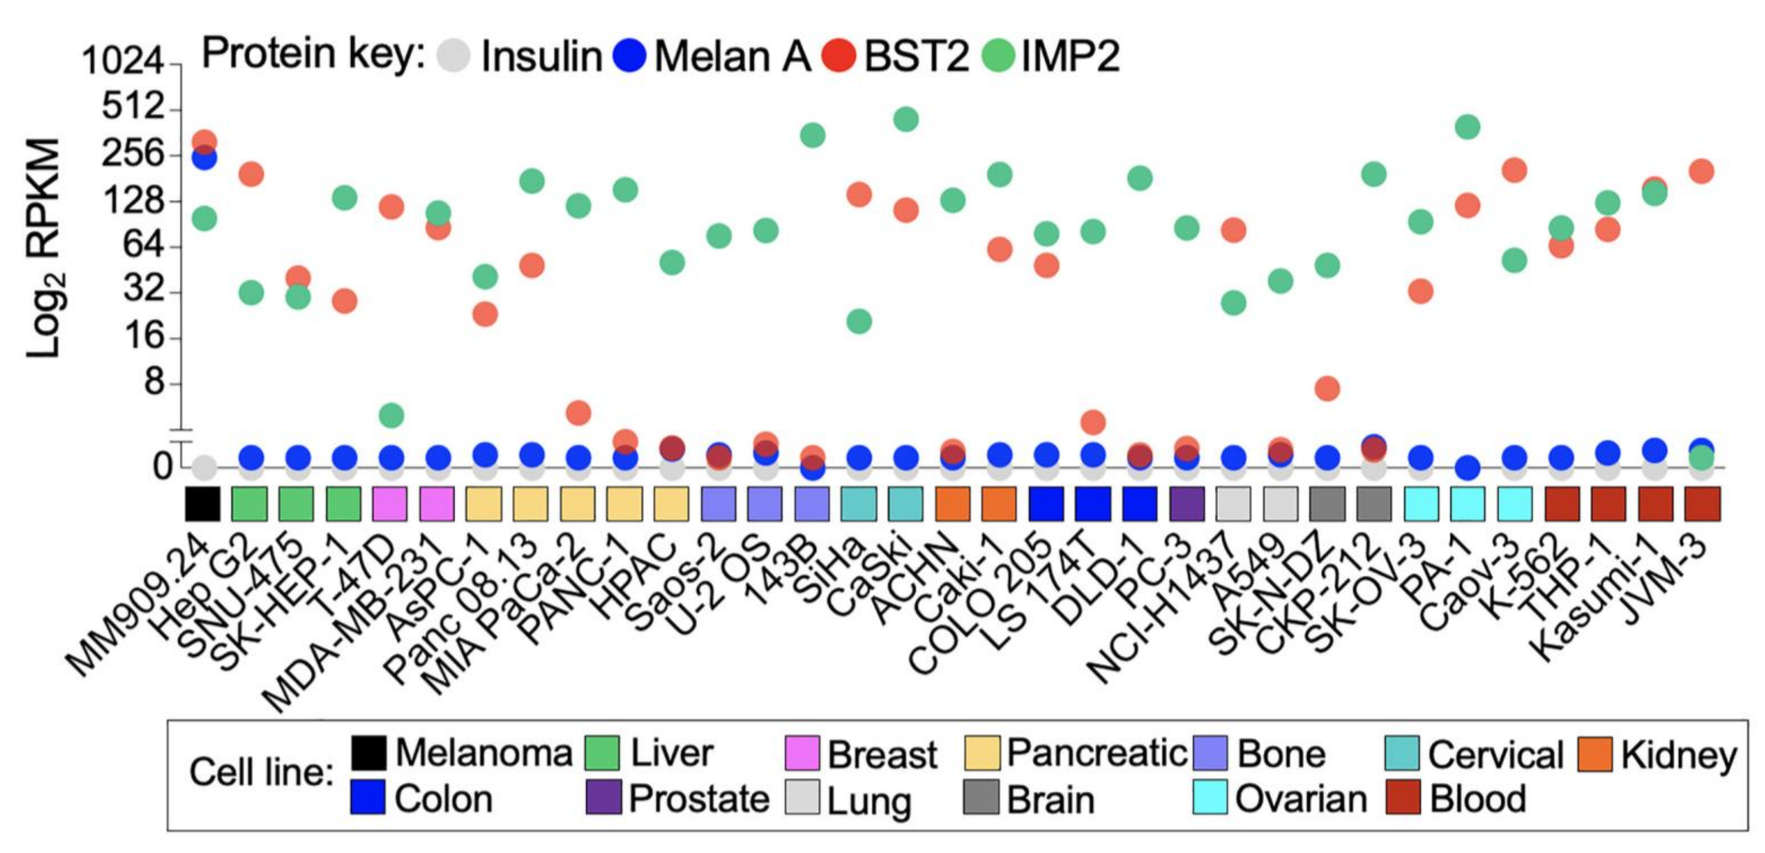
Protein key: Insulin Melan A BST2 IMP2
Log2 RPKM
1024
512
256
128
64
32
16
8
0
MM909.24
Hep G2
SNU-475
SK-HEP-1
T-47D
MDA-MB-231
AsPC-1
Panc 08.13
MIA PaCa-2
PANC-1
HPAC
Saos-2
U-2 OS
143B
SiHa
CaSki
ACHN
Caki-1
COLO 205
LS 174T
DLD-1
PC-3
NCI-H1437
A549
SK-N-DZ
CKP-212
SK-OV-3
PA-1
Caov-3
K-562
THP-1
Kasumi-1
JVM-3
Cell line: Melanoma Liver Breast Pancreatic Bone Cervical Kidney Colon Prostate Lung Brain Ovarian Blood

Adapted from Dolton, G. et al. (2023) Cell

Rare patient-derived TCRs (e.g. MEL5) display weak binding to three broadly-expressed HLA-A2 restricted tumor-associated antigens MART-1 (Melan A), BST2, and IMP2, motivating discovery of high-affinity TCRs for broad and potent multi-epitope tumor targeting.

METHODS – YEAST-BASED SYNTHETIC TCR LIBRARIES

TCR-CD3 complex / TCR-Fc / High-throughput yeast-based platform

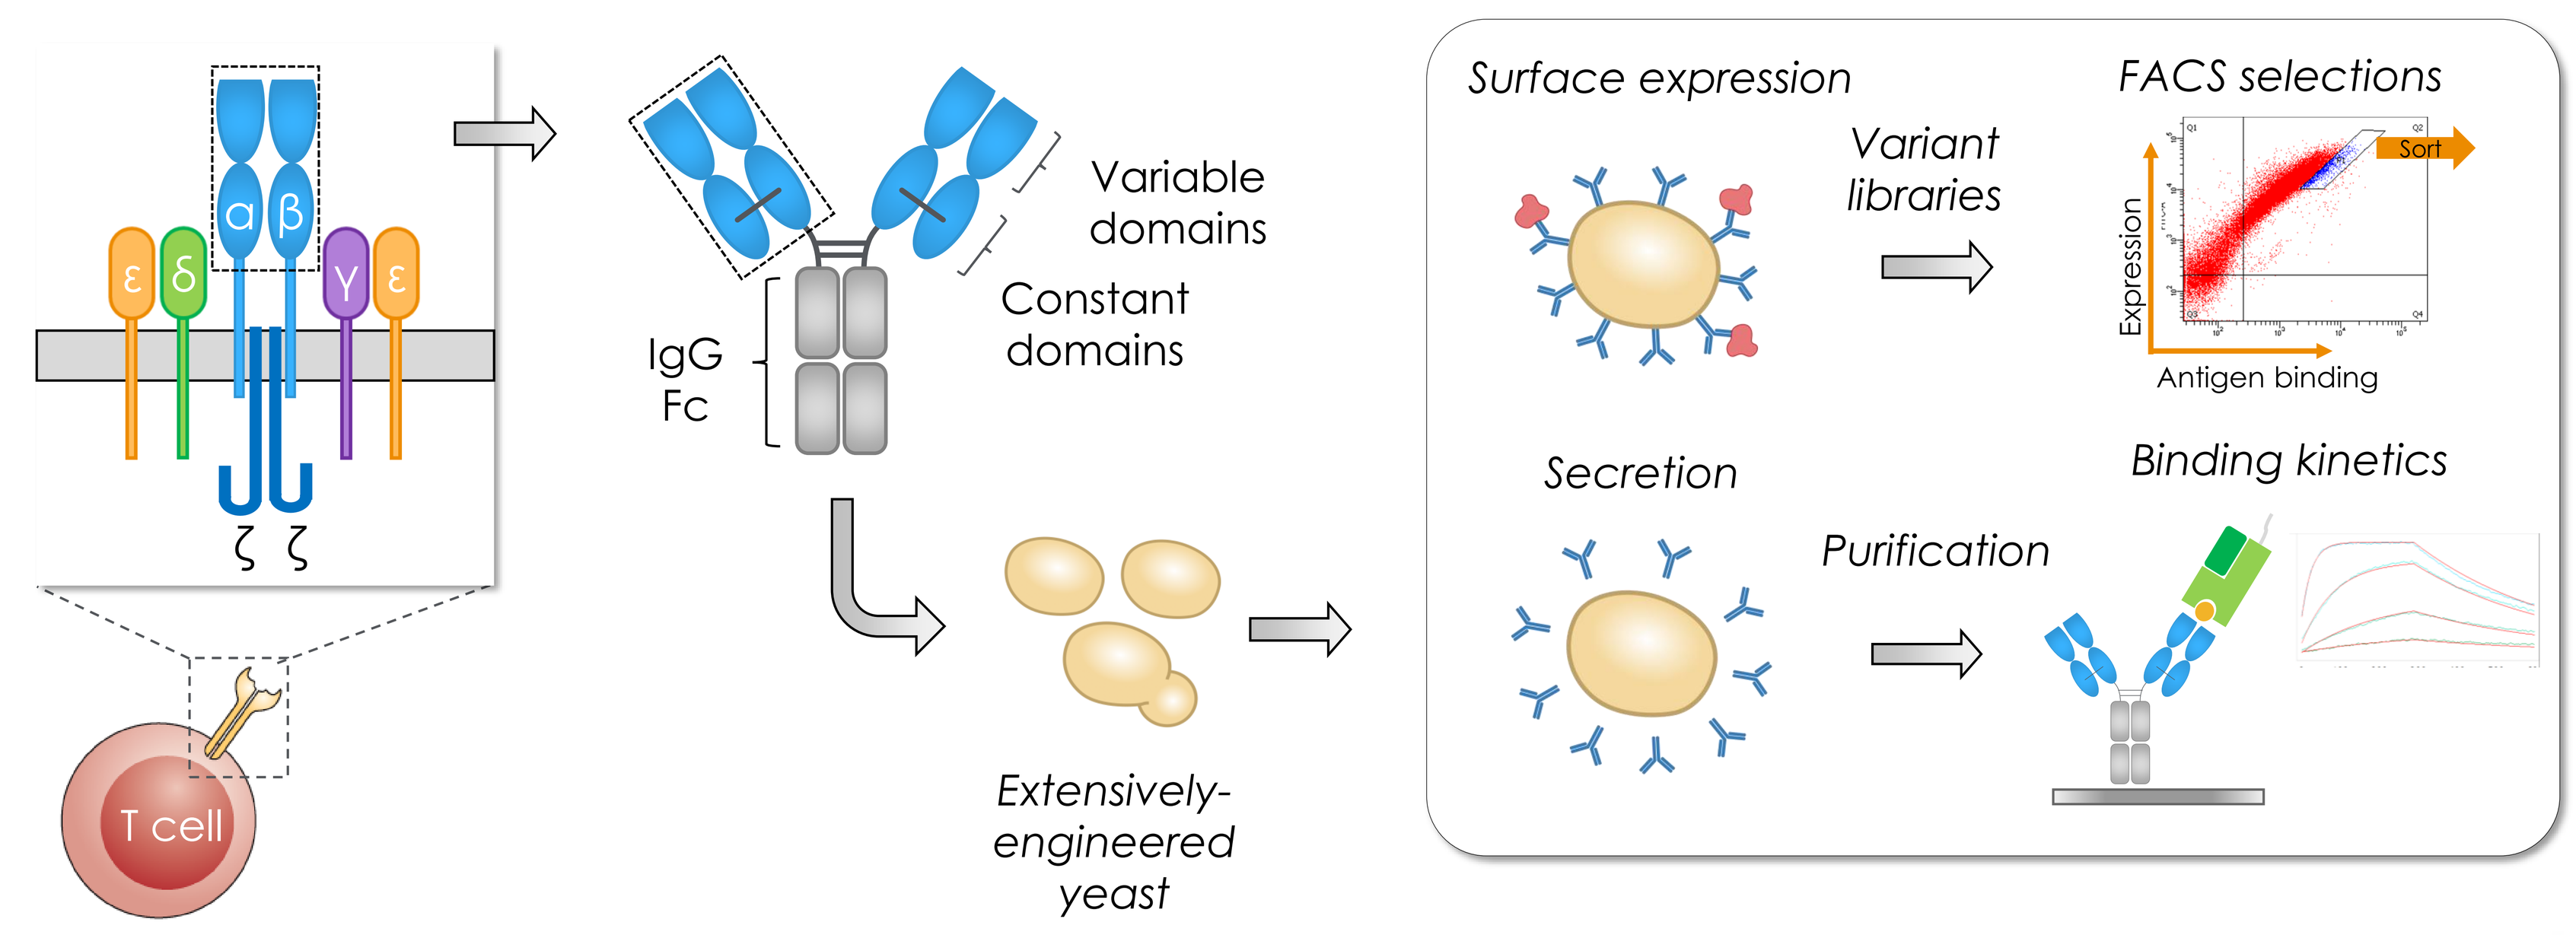
ε δ α β γ ε
ζ ζ
T cell
Variable domains
Constant domains
IgG Fc
Extensively-engineered yeast
Surface expression
Variant libraries
FACS selections
Sort
Expression
Antigen binding
Secretion
Purification
Binding kinetics

TCR library design / Synthetic TCR libraries

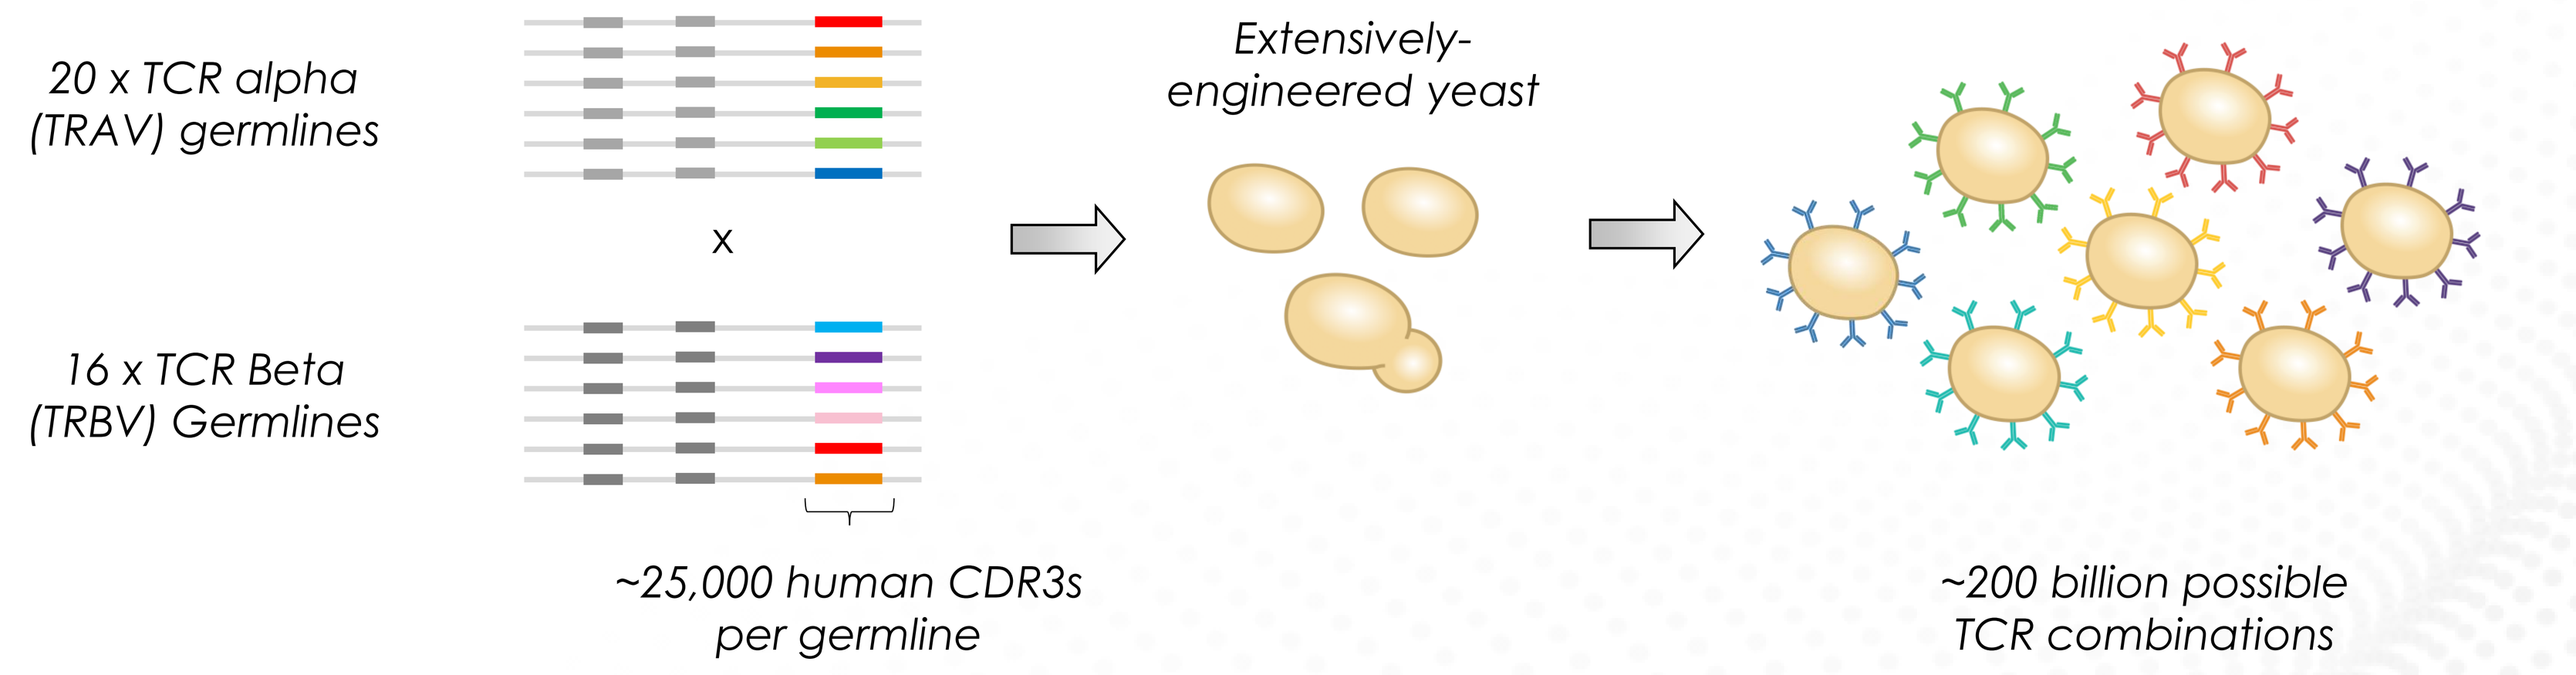
20 x TCR alpha (TRAV) germlines
x
16 x TCR Beta (TRBV) Germlines
~25,000 human CDR3s per germline
Extensively-engineered yeast
~200 billion possible TCR combinations

Generation of large and diverse fully-human synthetic TCR libraries built in yeast for rapid discovery and characterization of Fc-linked soluble TCRs in a high-throughput yeast platform.

METHODS – DISCOVERY OF RARE MULTI-SPECIFIC TCRs

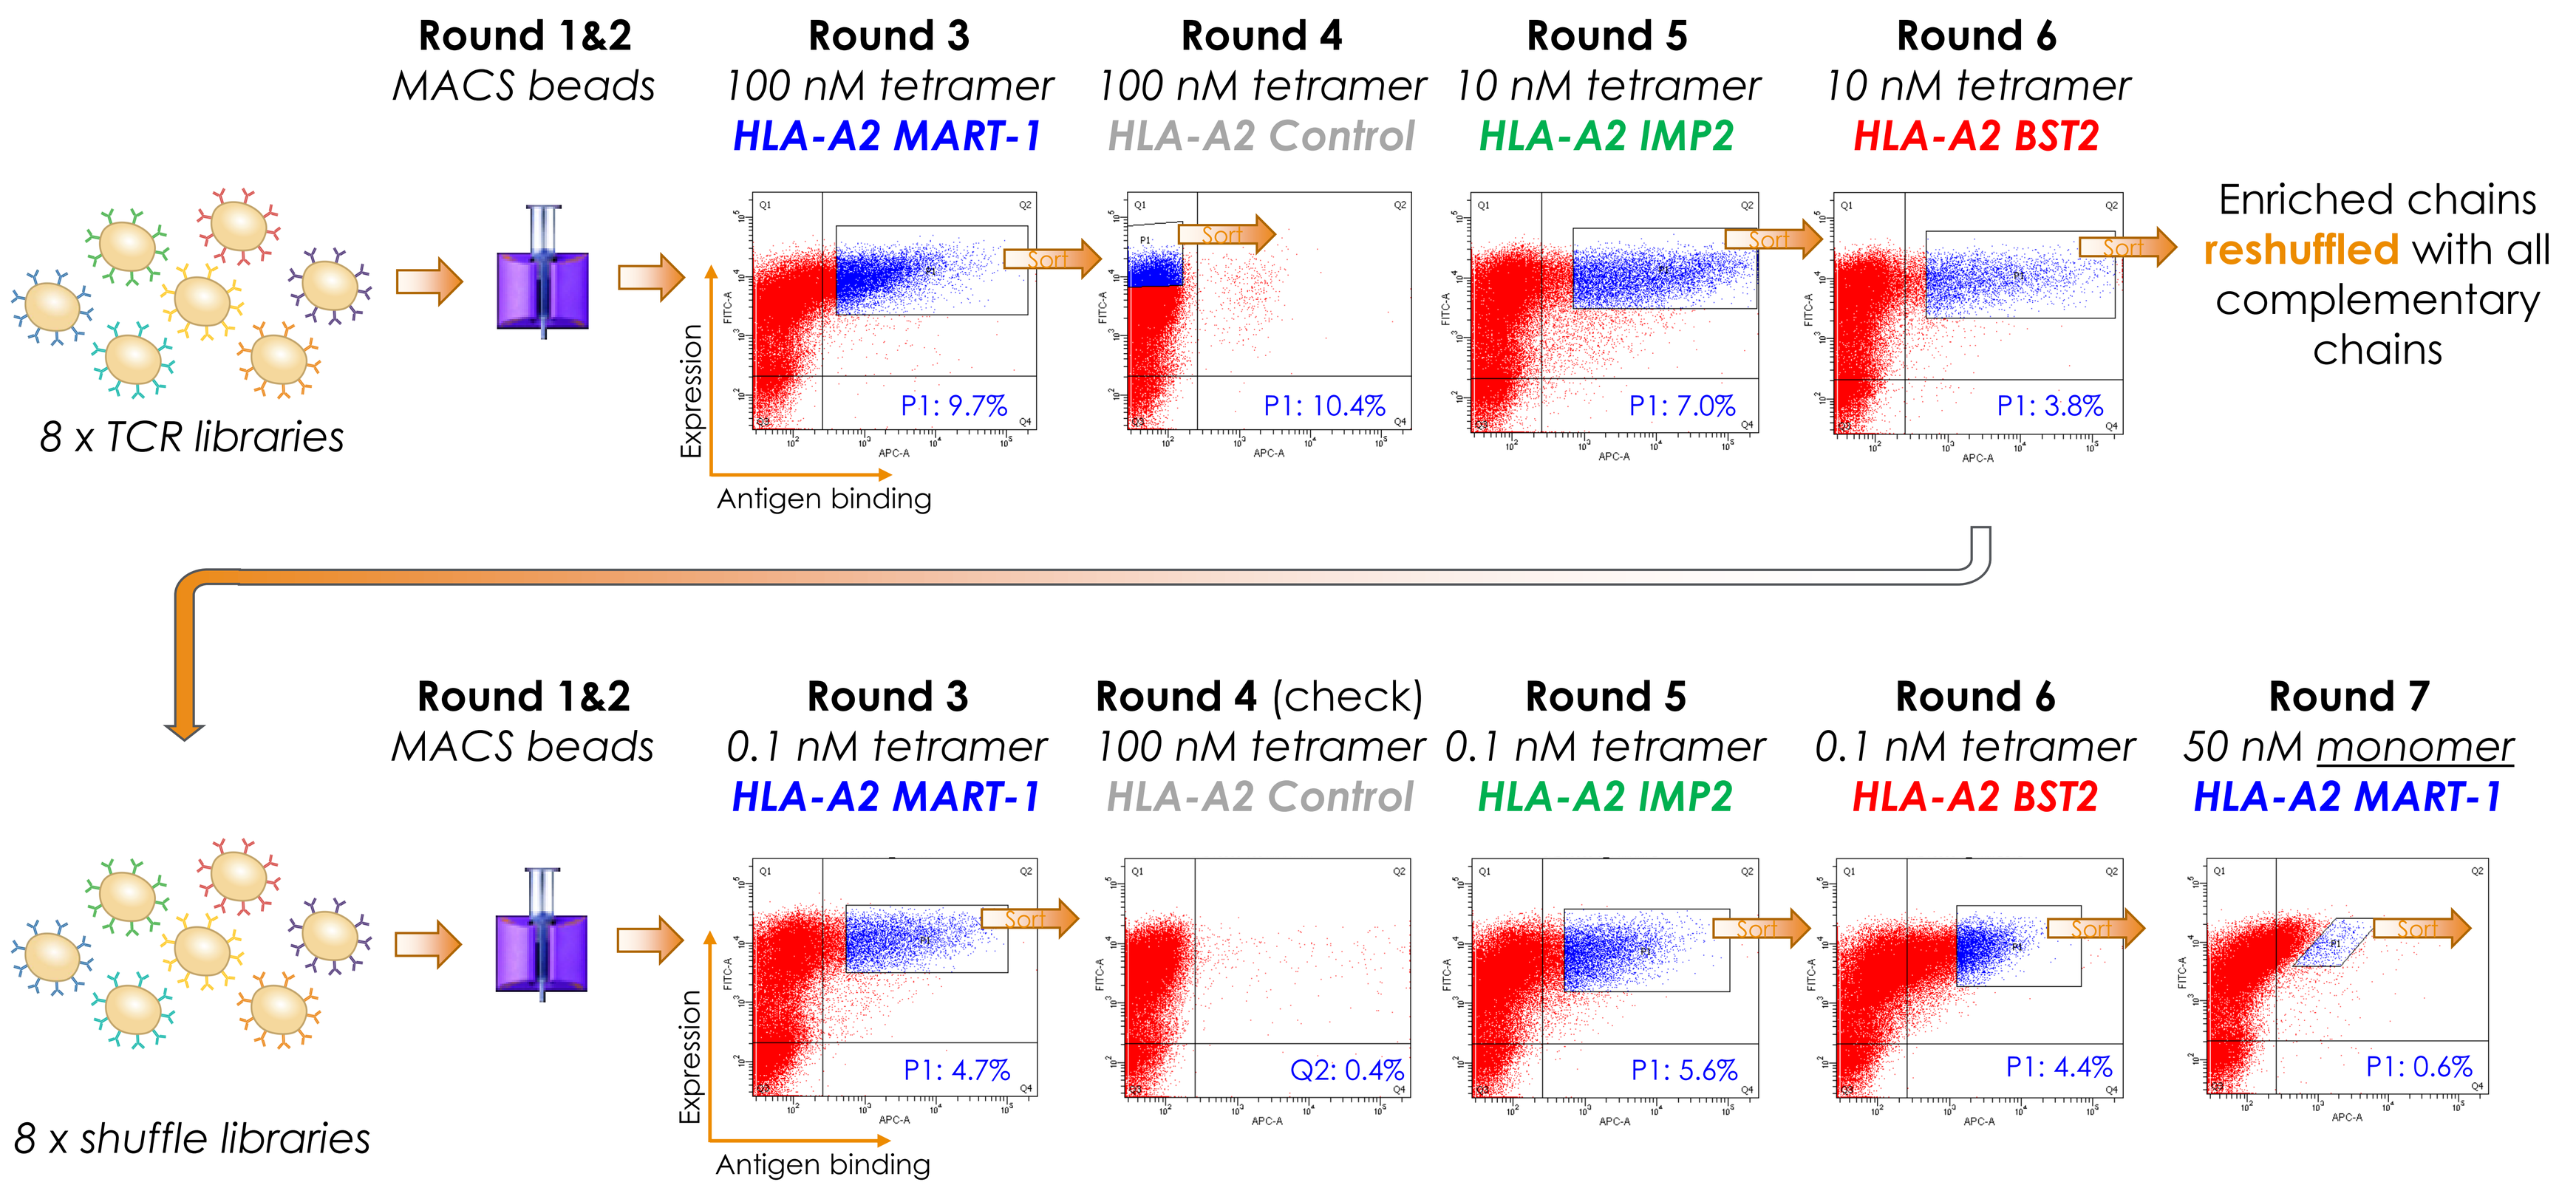
Round 1&2
MACS beads
Round 3
100 nM tetramer
HLA-A2 MART-1
Round 4
100 nM tetramer
HLA-A2 Control
Round 5
10 nM tetramer
HLA-A2 IMP2
Round 6
10 nM tetramer
HLA-A2 BST2
8 x TCR libraries
Expression
Antigen binding
P1: 9.7%
P1: 10.4%
P1: 7.0%
P1: 3.8%
Sort
Enriched chains reshuffled with all complementary chains
Round 1&2
MACS beads
Round 3
0.1 nM tetramer
HLA-A2 MART-1
Round 4 (check)
100 nM tetramer
HLA-A2 Control
Round 5
0.1 nM tetramer
HLA-A2 IMP2
Round 6
0.1 nM tetramer
HLA-A2 BST2
Round 7
50 nM monomer
HLA-A2 MART-1
8 x shuffle libraries
P1: 4.7%
Q2: 0.4%
P1: 5.6%
P1: 4.4%
P1: 0.6%

Rare TCRs with high-affinity multi-specific binding to HLA-A2 MART-1, BST2, and IMP2 were isolated from synthetic TCR libraries via iterative rounds of selection and re-diversification.

RESULTS – HIGH-AFFINITY MULTI-SPECIFIC TCRs

TCR affinity and cross-reactivity

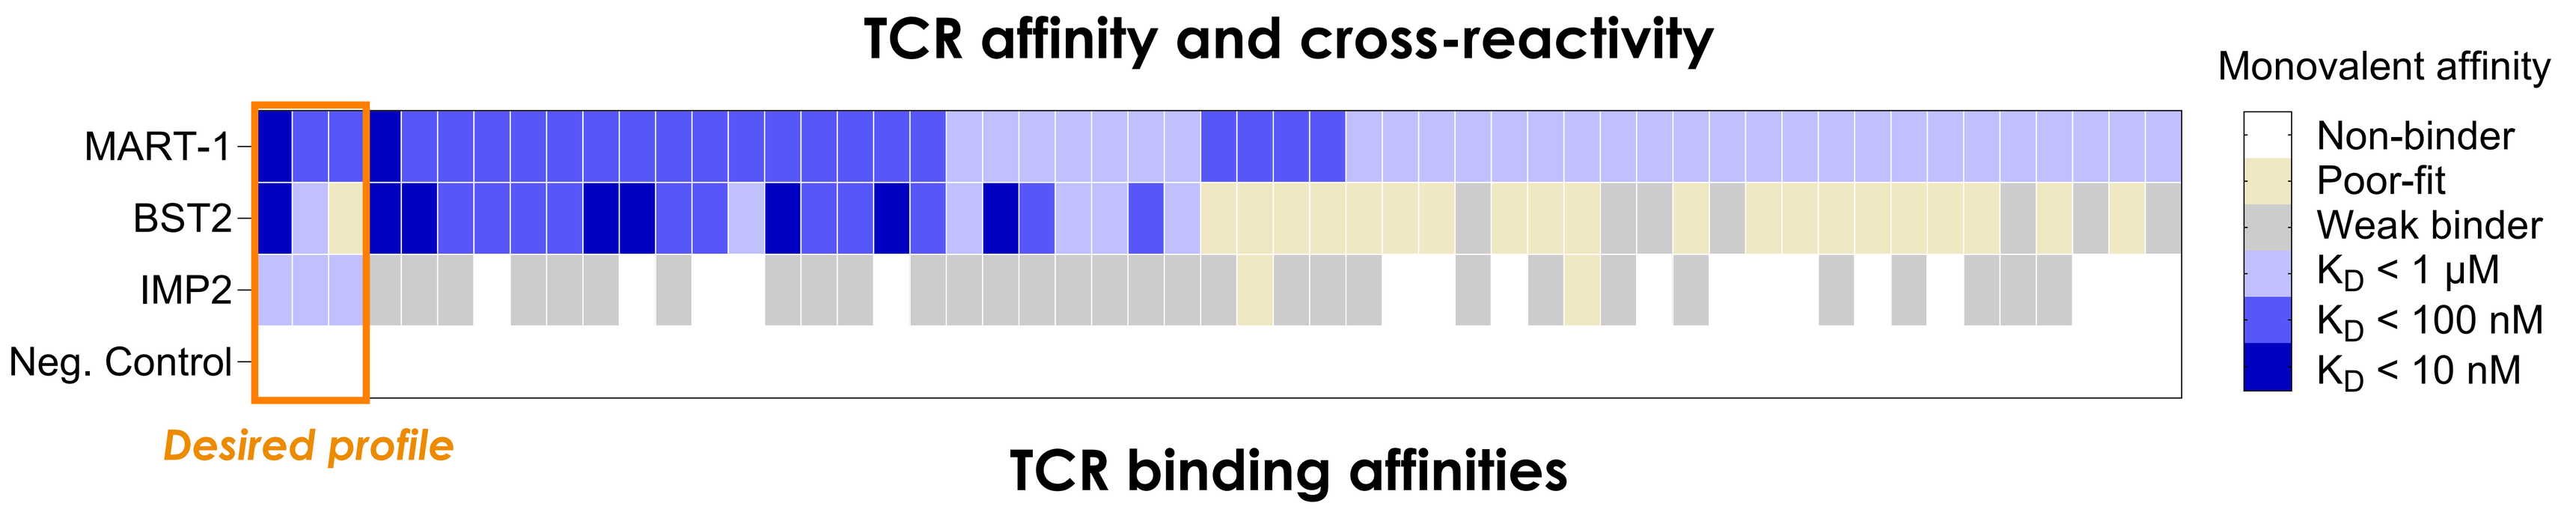
MART-1
BST2
IMP2
Neg. Control
Desired profile
Monovalent affinity
Non-binder
Poor-fit
Weak binder
KD < 1 μM
KD < 100 nM
KD < 10 nM

TCR binding affinities

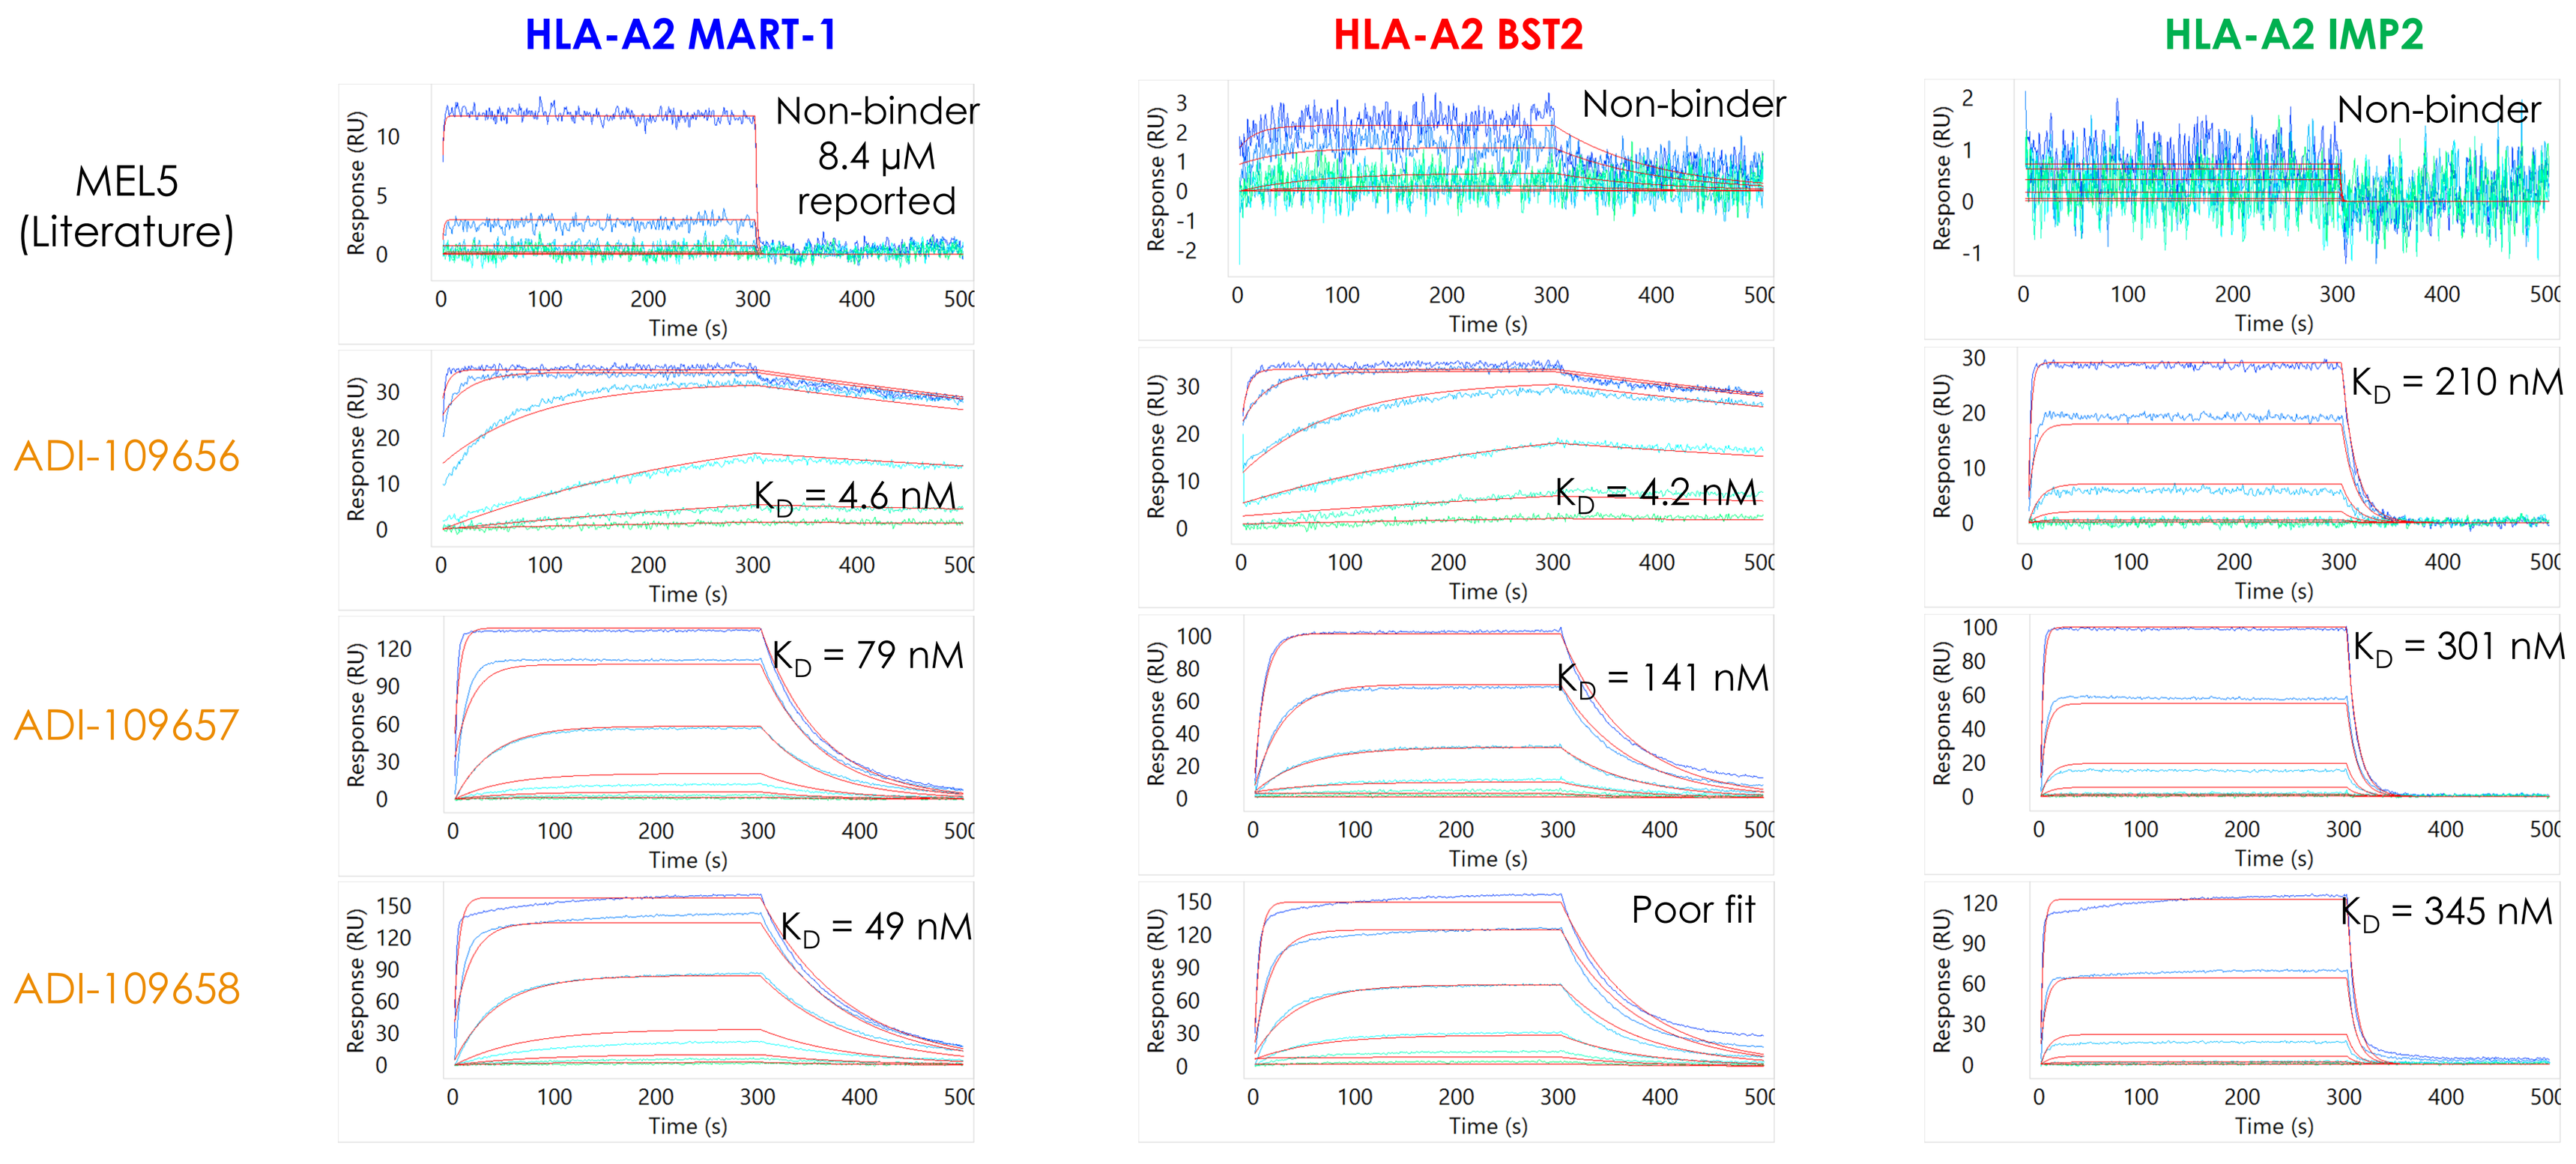
HLA-A2 MART-1
HLA-A2 BST2
HLA-A2 IMP2
MEL5 (Literature)
Non-binder 8.4 μM reported
Non-binder
Non-binder
ADI-109656
KD = 4.6 nM
KD = 4.2 nM
KD = 210 nM
ADI-109657
KD = 79 nM
KD = 141 nM
KD = 301 nM
ADI-109658
KD = 49 nM
Poor fit
KD = 345 nM
Response (RU)
Time (s)

Library-derived TCRs are enriched for multi-specific binding and display high affinity to all three tumor-associated pHLA targets but no binding to allele-matched negative controls.

NEXT STEPS – FINE SPECIFICITY ASSSESSMENTS

Peptide-diversified library / Peptide specificity motif / Off-target validation

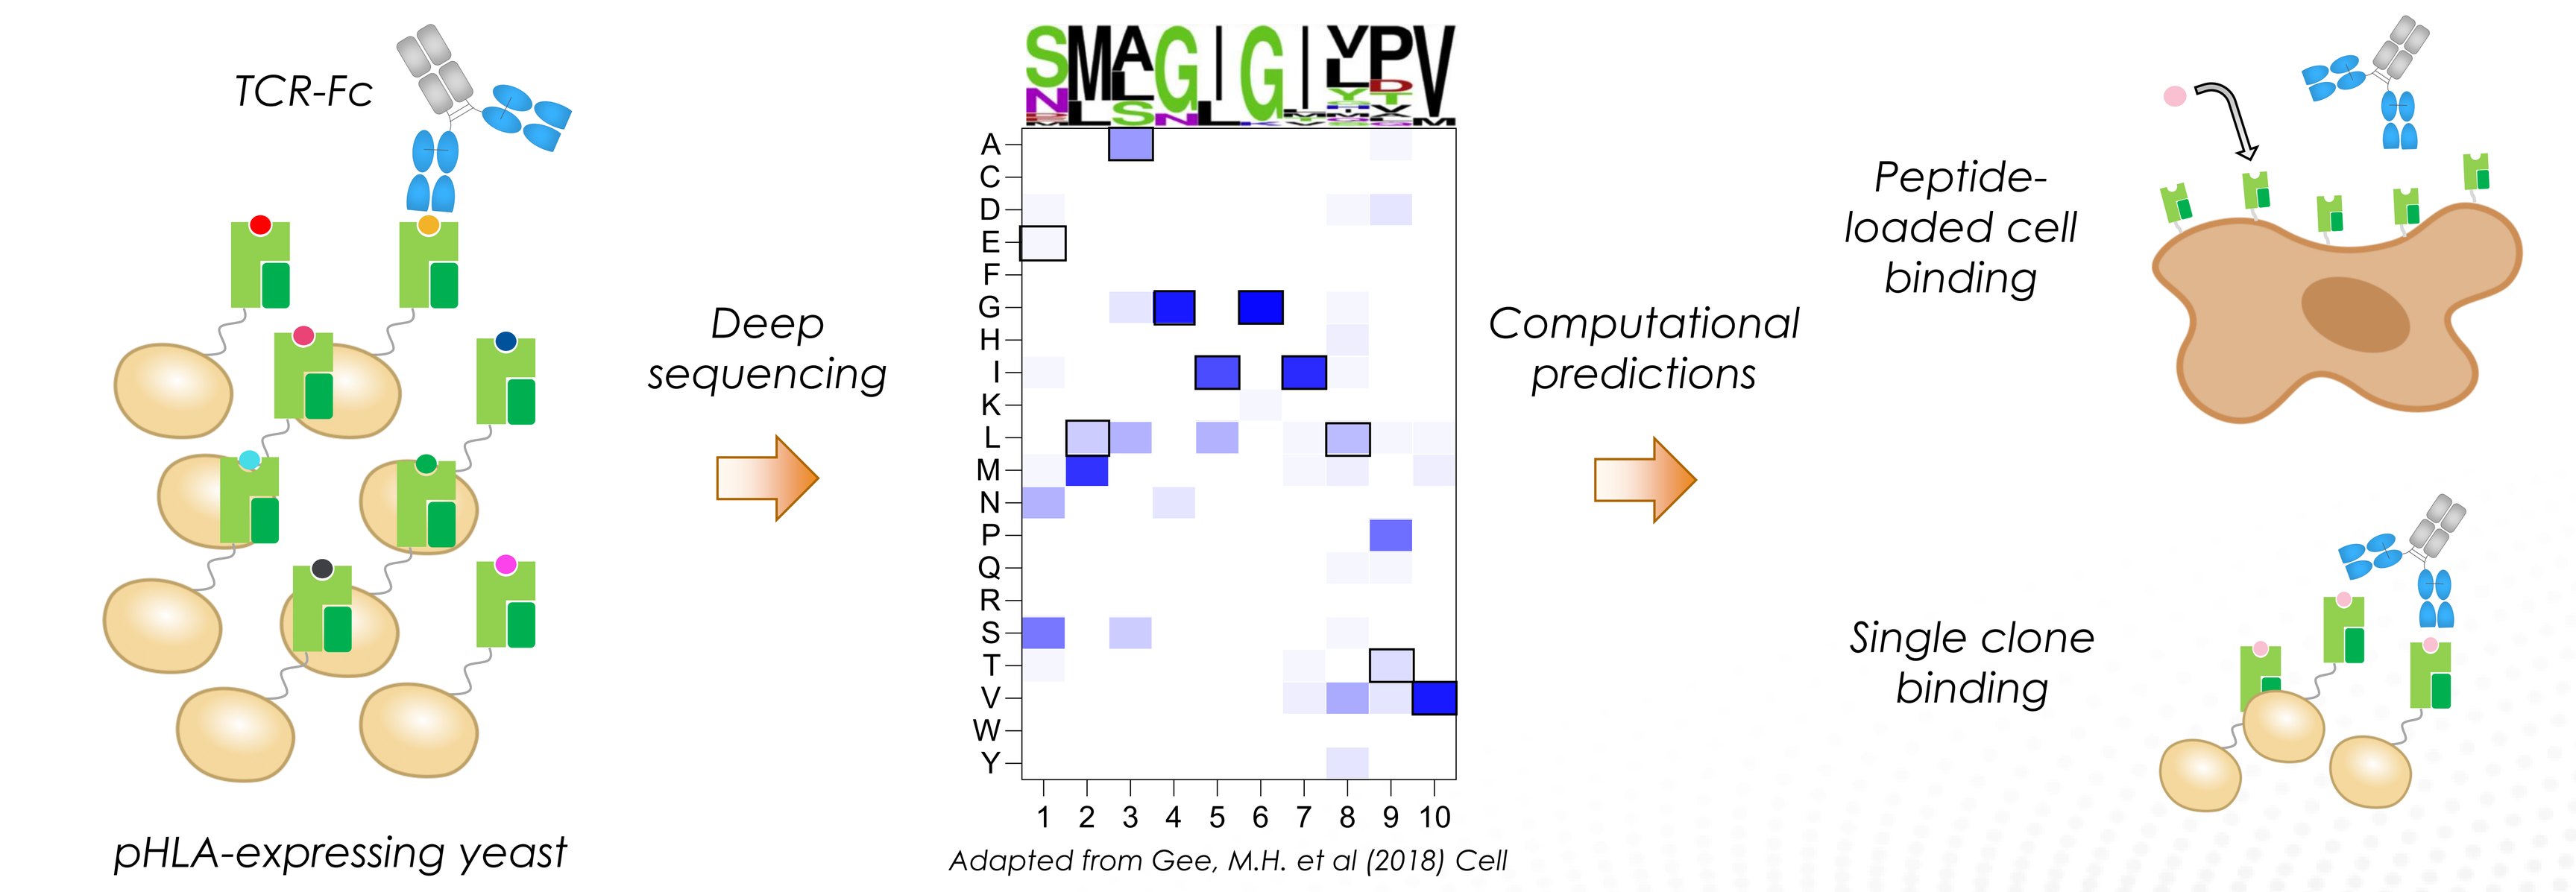
TCR-Fc
pHLA-expressing yeast
Deep sequencing
A C D E F G H I K L M N P Q R S T V W Y
1 2 3 4 5 6 7 8 9 10
Computational predictions
Peptide-loaded cell binding
Single clone binding

Adapted from Gee, M.H. et al (2018) Cell

Fine specificity of library-derived TCRs will be determined via selection of diversified pHLA libraries expressed in yeast, and predicted human off-targets will be validated individually.

CONCLUSIONS

- Rare patient-derived TCRs exhibit specificity for three broadly-expressed HLA-A2 restricted tumor-associated antigens but possess insufficient affinity for use in soluble TCR-based T cell engager therapeutics.
- Tailored selections of fully-human TCR libraries built in yeast yield synthetic TCRs with high-affinity multi-specific binding to all three tumor-associated pHLA targets, providing new opportunities for broad and potent multi-epitope tumor targeting.
- Future work will investigate the specificity of library-derived TCRs to ensure their selectivity.
- Identification of rare high-affinity anti-tumor TCRs against other pHLA targets is ongoing.

Scan for downloadable version