NOVEL ANTI-CD3 HEAVY CHAIN-ONLY ANTIBODIES FOR USE IN T-CELL ENGAGING THERAPEUTICS

Paul Khalifé, Hannah C. Watkins, Elizabeth R. Parker, Cameron M. R. Henkel, Todd M. Boland, Hailey M. Heston, Christin R. Strong, Jingfu Zhao, Carissa A. Metzger, Irina Burnina, Michael E. Brown, Jessica P. Dawson, Andrew D. Avery, C. Garrett Rappazzo, Beth H. Sharkey, Morgan B. Morrill, Cory L. Ahonen, Eric Krauland, Rob Pejchal, Michael B. Battles, and Noel T. Pauli

ADIMAB

INTRODUCTION TO T CELL ENGAGERS

T cell engagers (TCEs) are an increasingly validated class of multispecific antibodies. Through pairing an arm targeting CD3 with an arm targeting a tumor associated antigen, the TCE forces the T cell and tumor cell into close proximity, triggering T cell activation leading to tumor cell killing[1].

- Eight TCEs are currently FDA approved—six since 2022[1]
- TCEs can be engineered in multiple bi- and multispecific formats. Activation level can be influenced by format[1]

T cell engagers enable killing of target cells

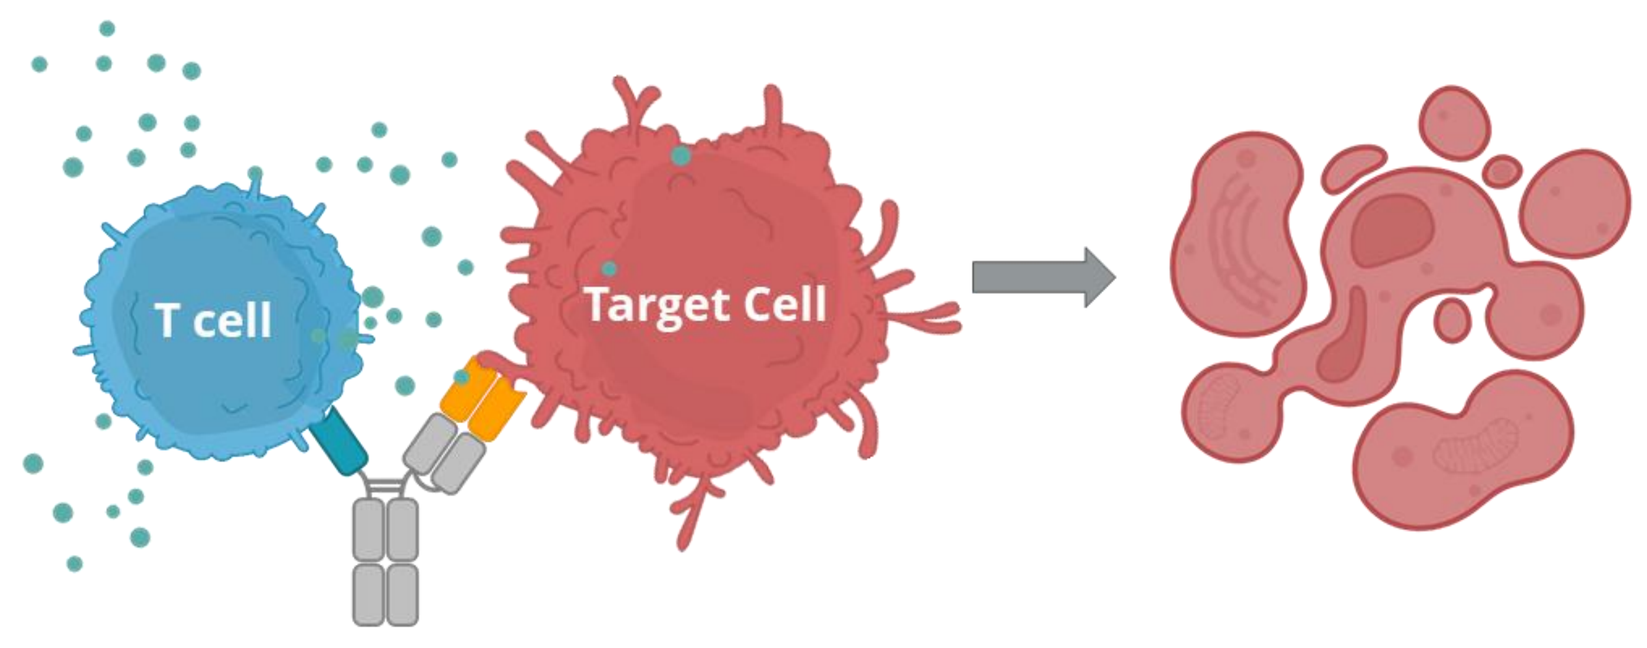
T cell
Target Cell

ADIMAB a-CD3 CONVENTIONAL IgG LINEAGE: HUMAN x CYNO CROSS-REACTIVTY AND TUNABLE POTENCY

Adimab's aCD3 IgG/FAb/scFv panel spans a wide range of affinities and has hu-cyno cross-reactivity

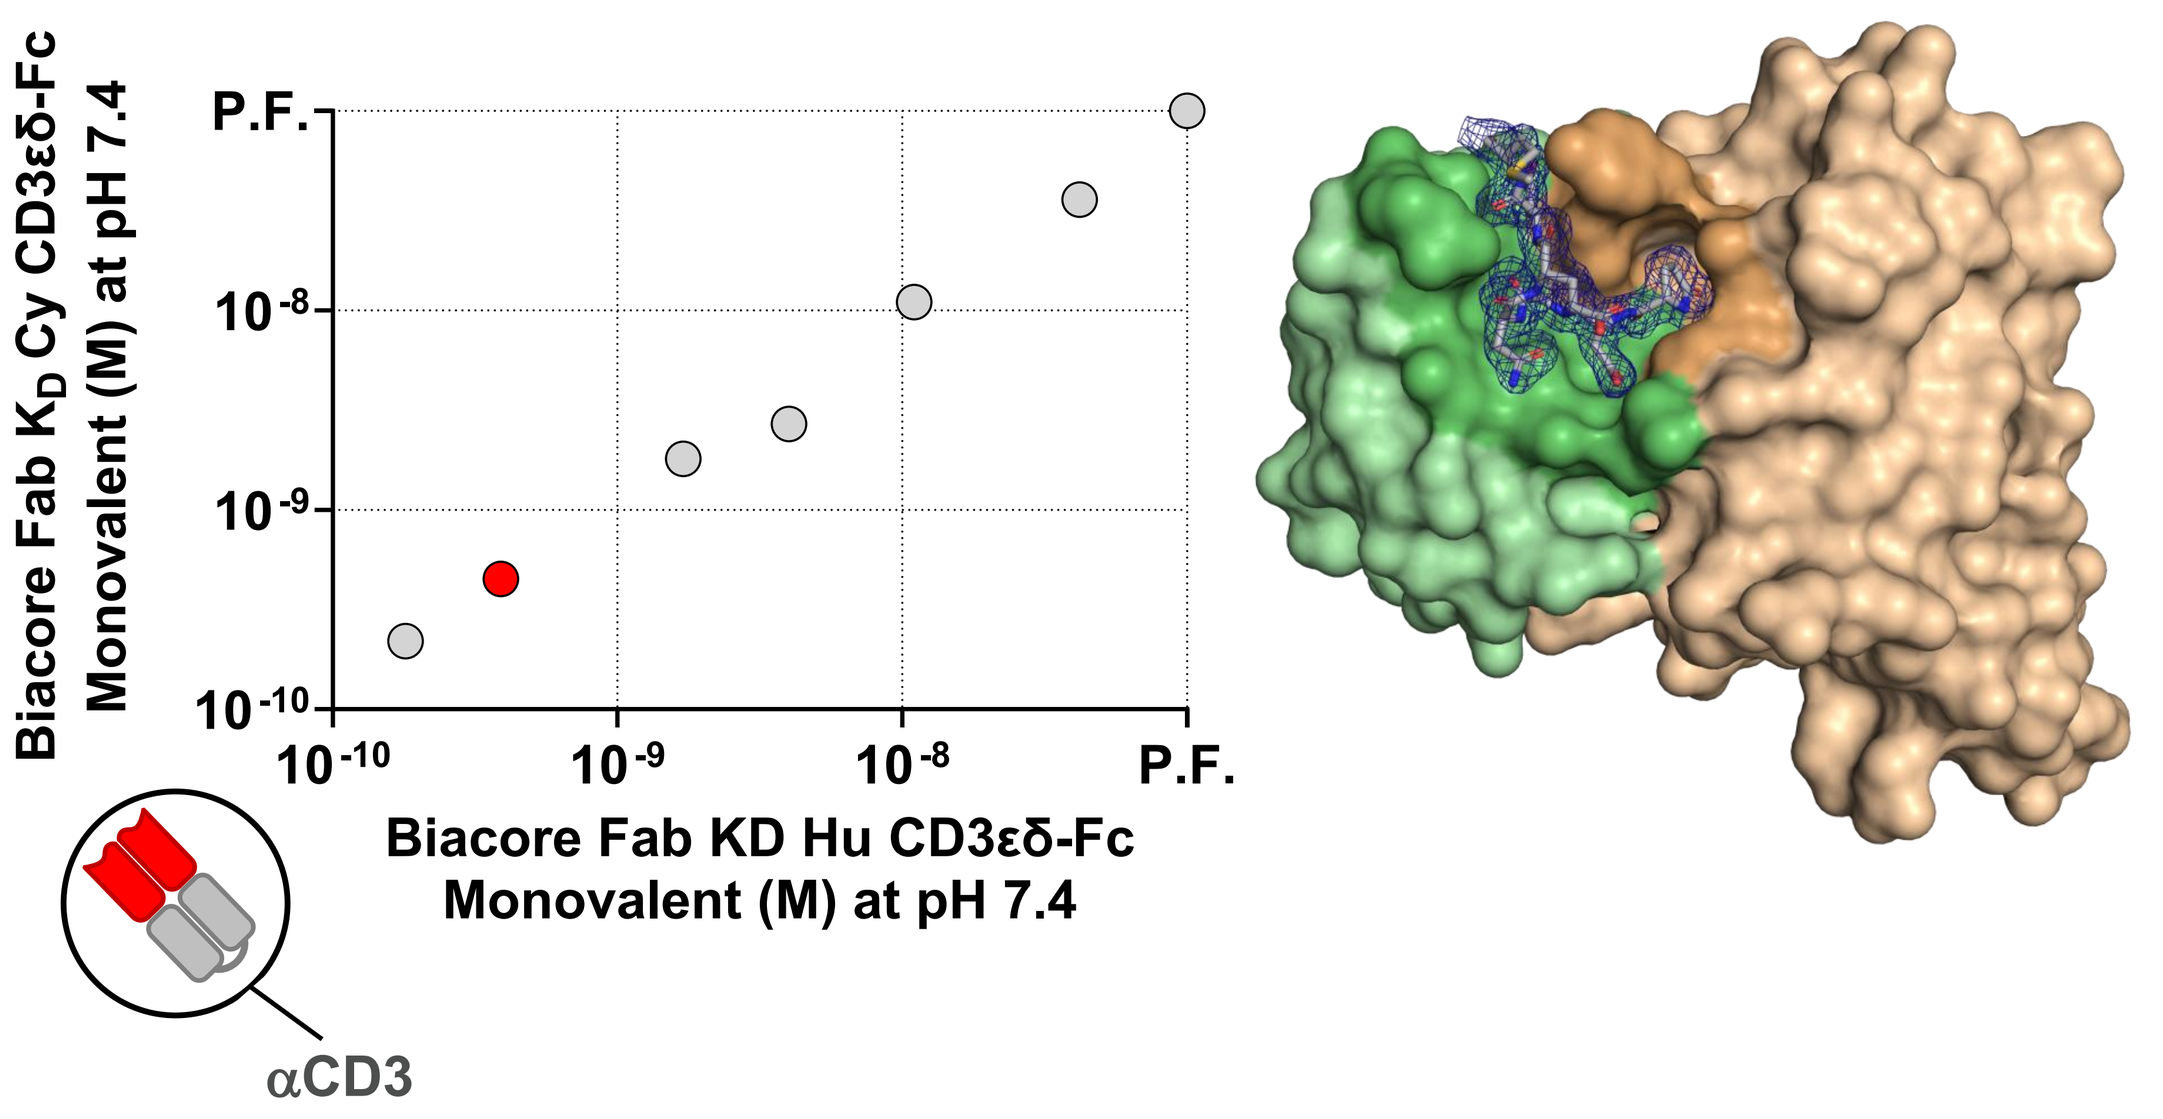
Biacore Fab K_D Cy CD3εδ-Fc Monovalent (M) at pH 7.4
P.F.
10^{-8}
10^{-9}
10^{-10}
10^{-10}
10^{-9}
10^{-8}
P.F.
Biacore Fab KD Hu CD3εδ-Fc Monovalent (M) at pH 7.4
αCD3

Bispecific CD3 x HER2 TCEs generated with Adimab's CD3 panel facilitate cytotoxicity (SKOV3)

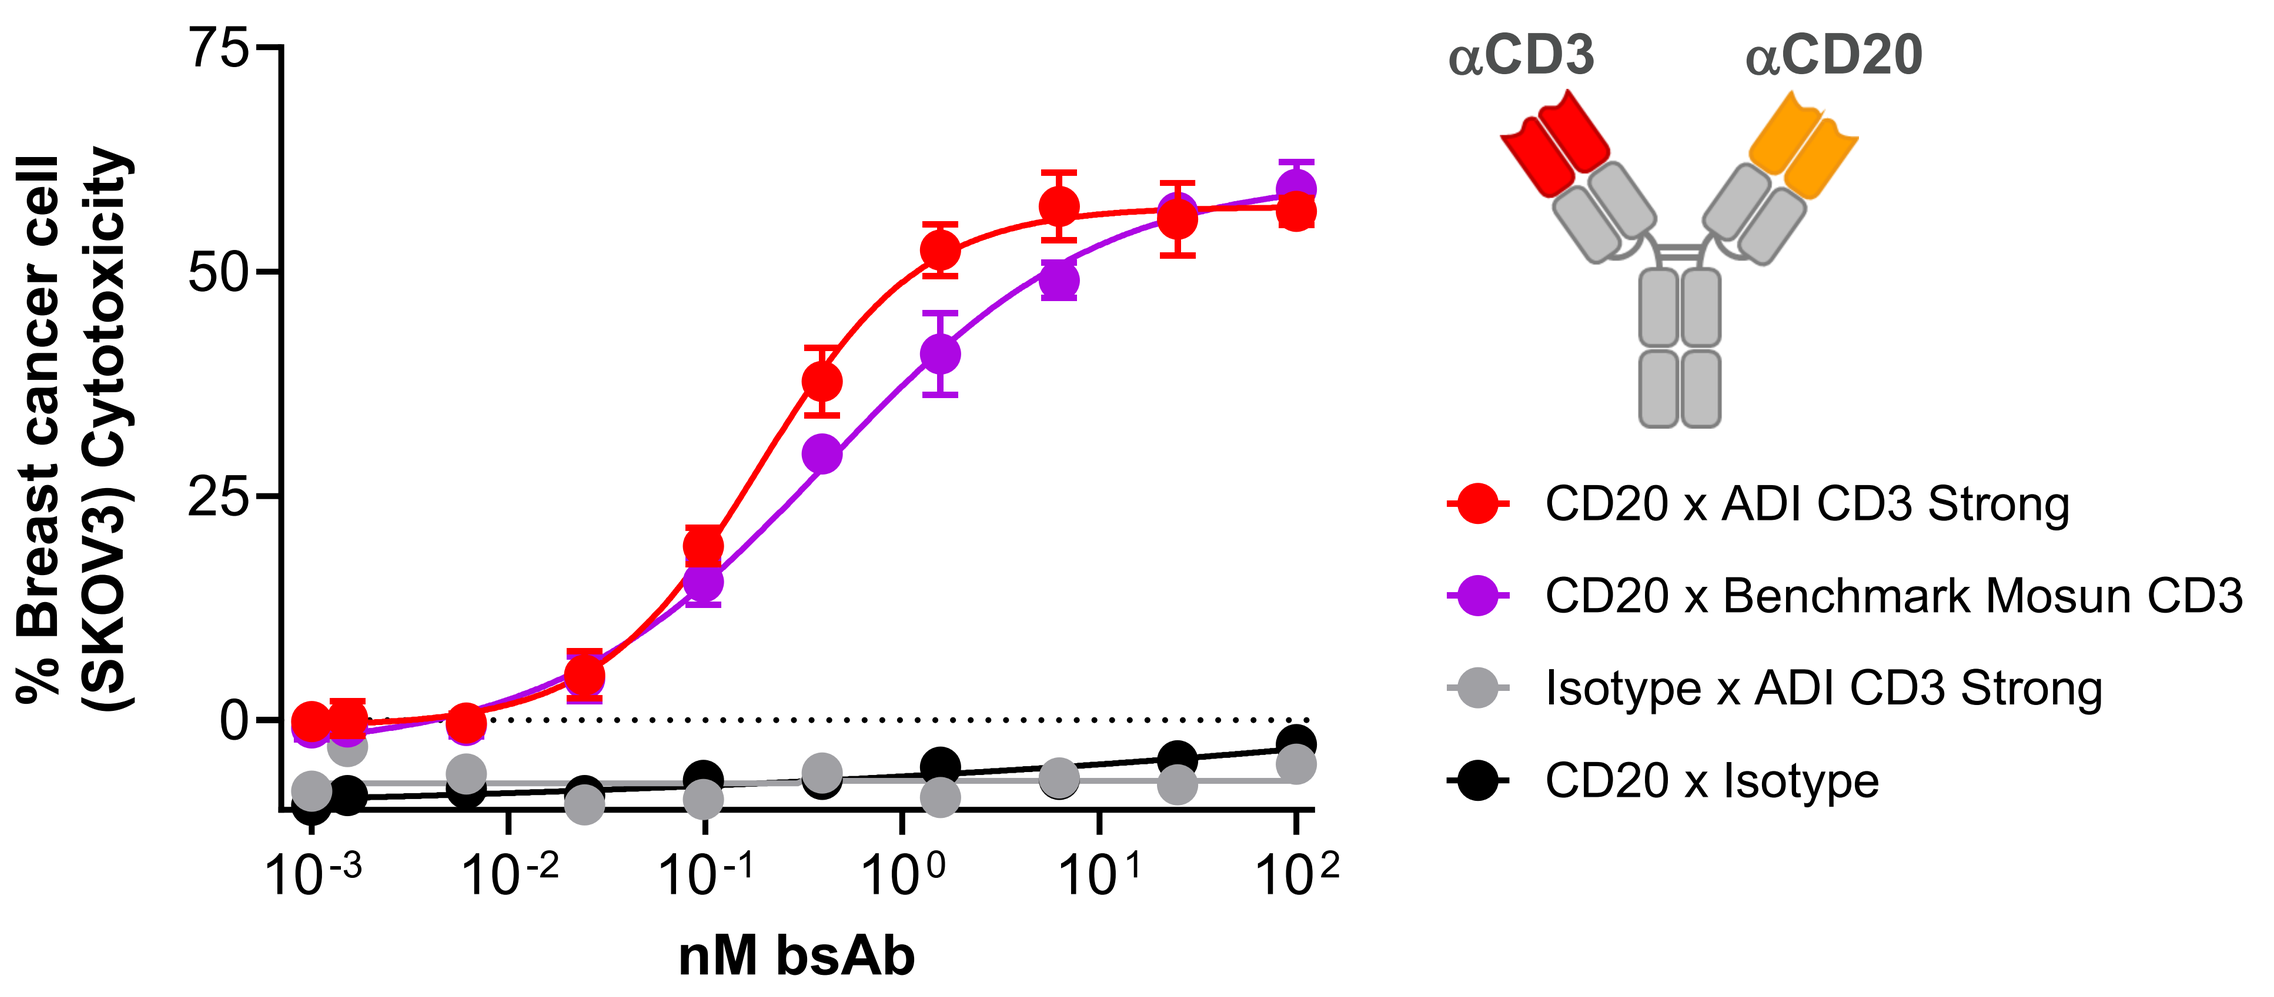
% Breast cancer cell (SKOV3) Cytotoxicity
75
50
25
0
10^{-3} 10^{-2} 10^{-1} 10^{0} 10^{1} 10^{2}
nM bsAb
αCD3
αCD20
CD20 x ADI CD3 Strong
CD20 x Benchmark Mosun CD3
Isotype x ADI CD3 Strong
CD20 x Isotype

EXPANDING ADIMAB'S CD3 BINDERS: HCAB DISCOVERY

Libraries built from llamas immunized against CD3

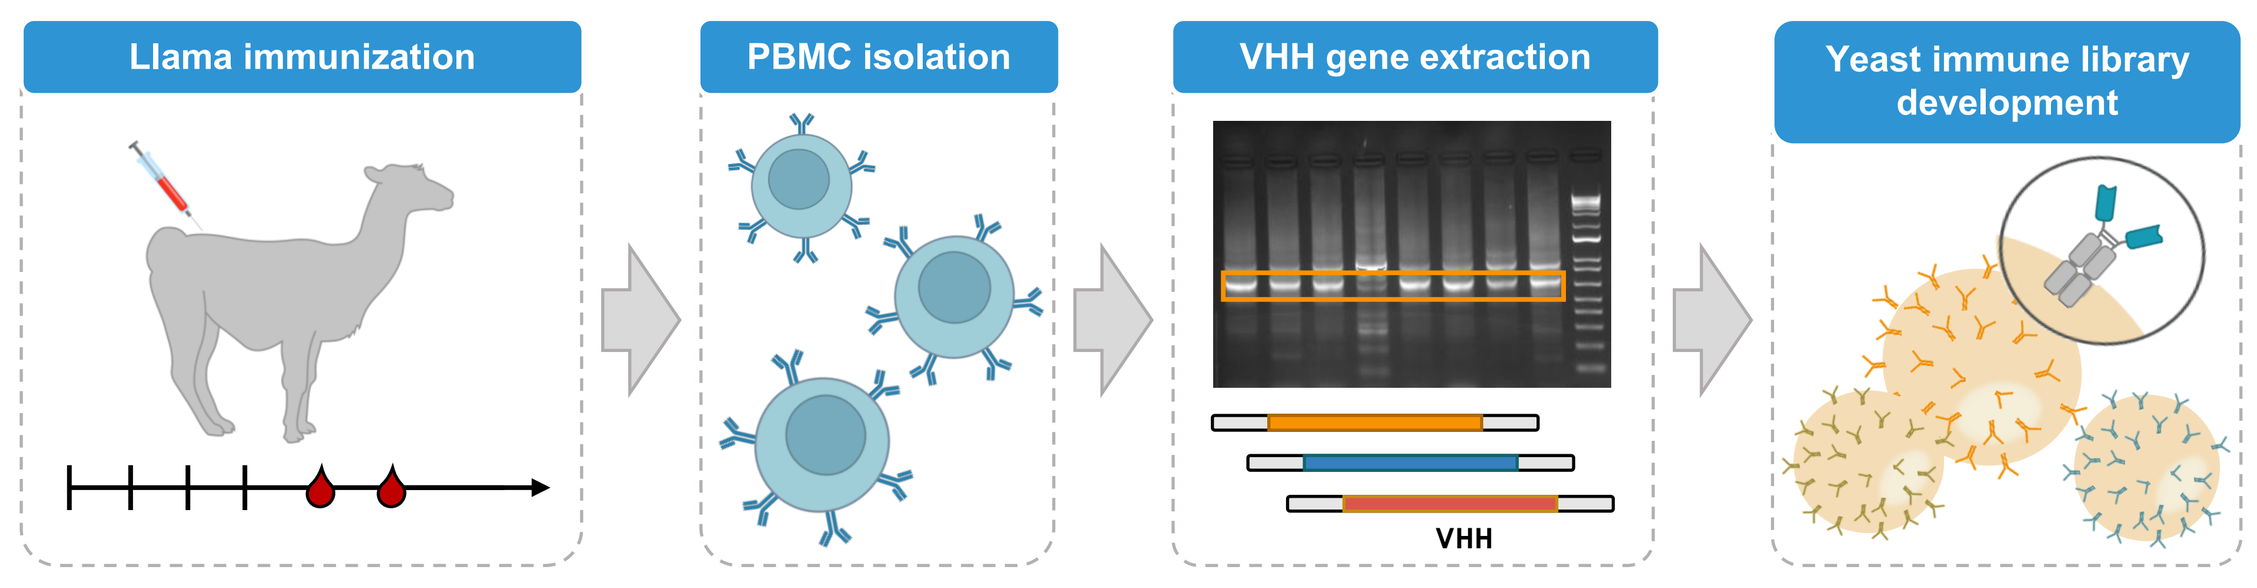
Llama immunization
PBMC isolation
VHH gene extraction
Yeast immune library development
VHH

Rounds of selections performed on the Adimab yeast immune libraries to isolate aCD3 HCAbs with desirable properties

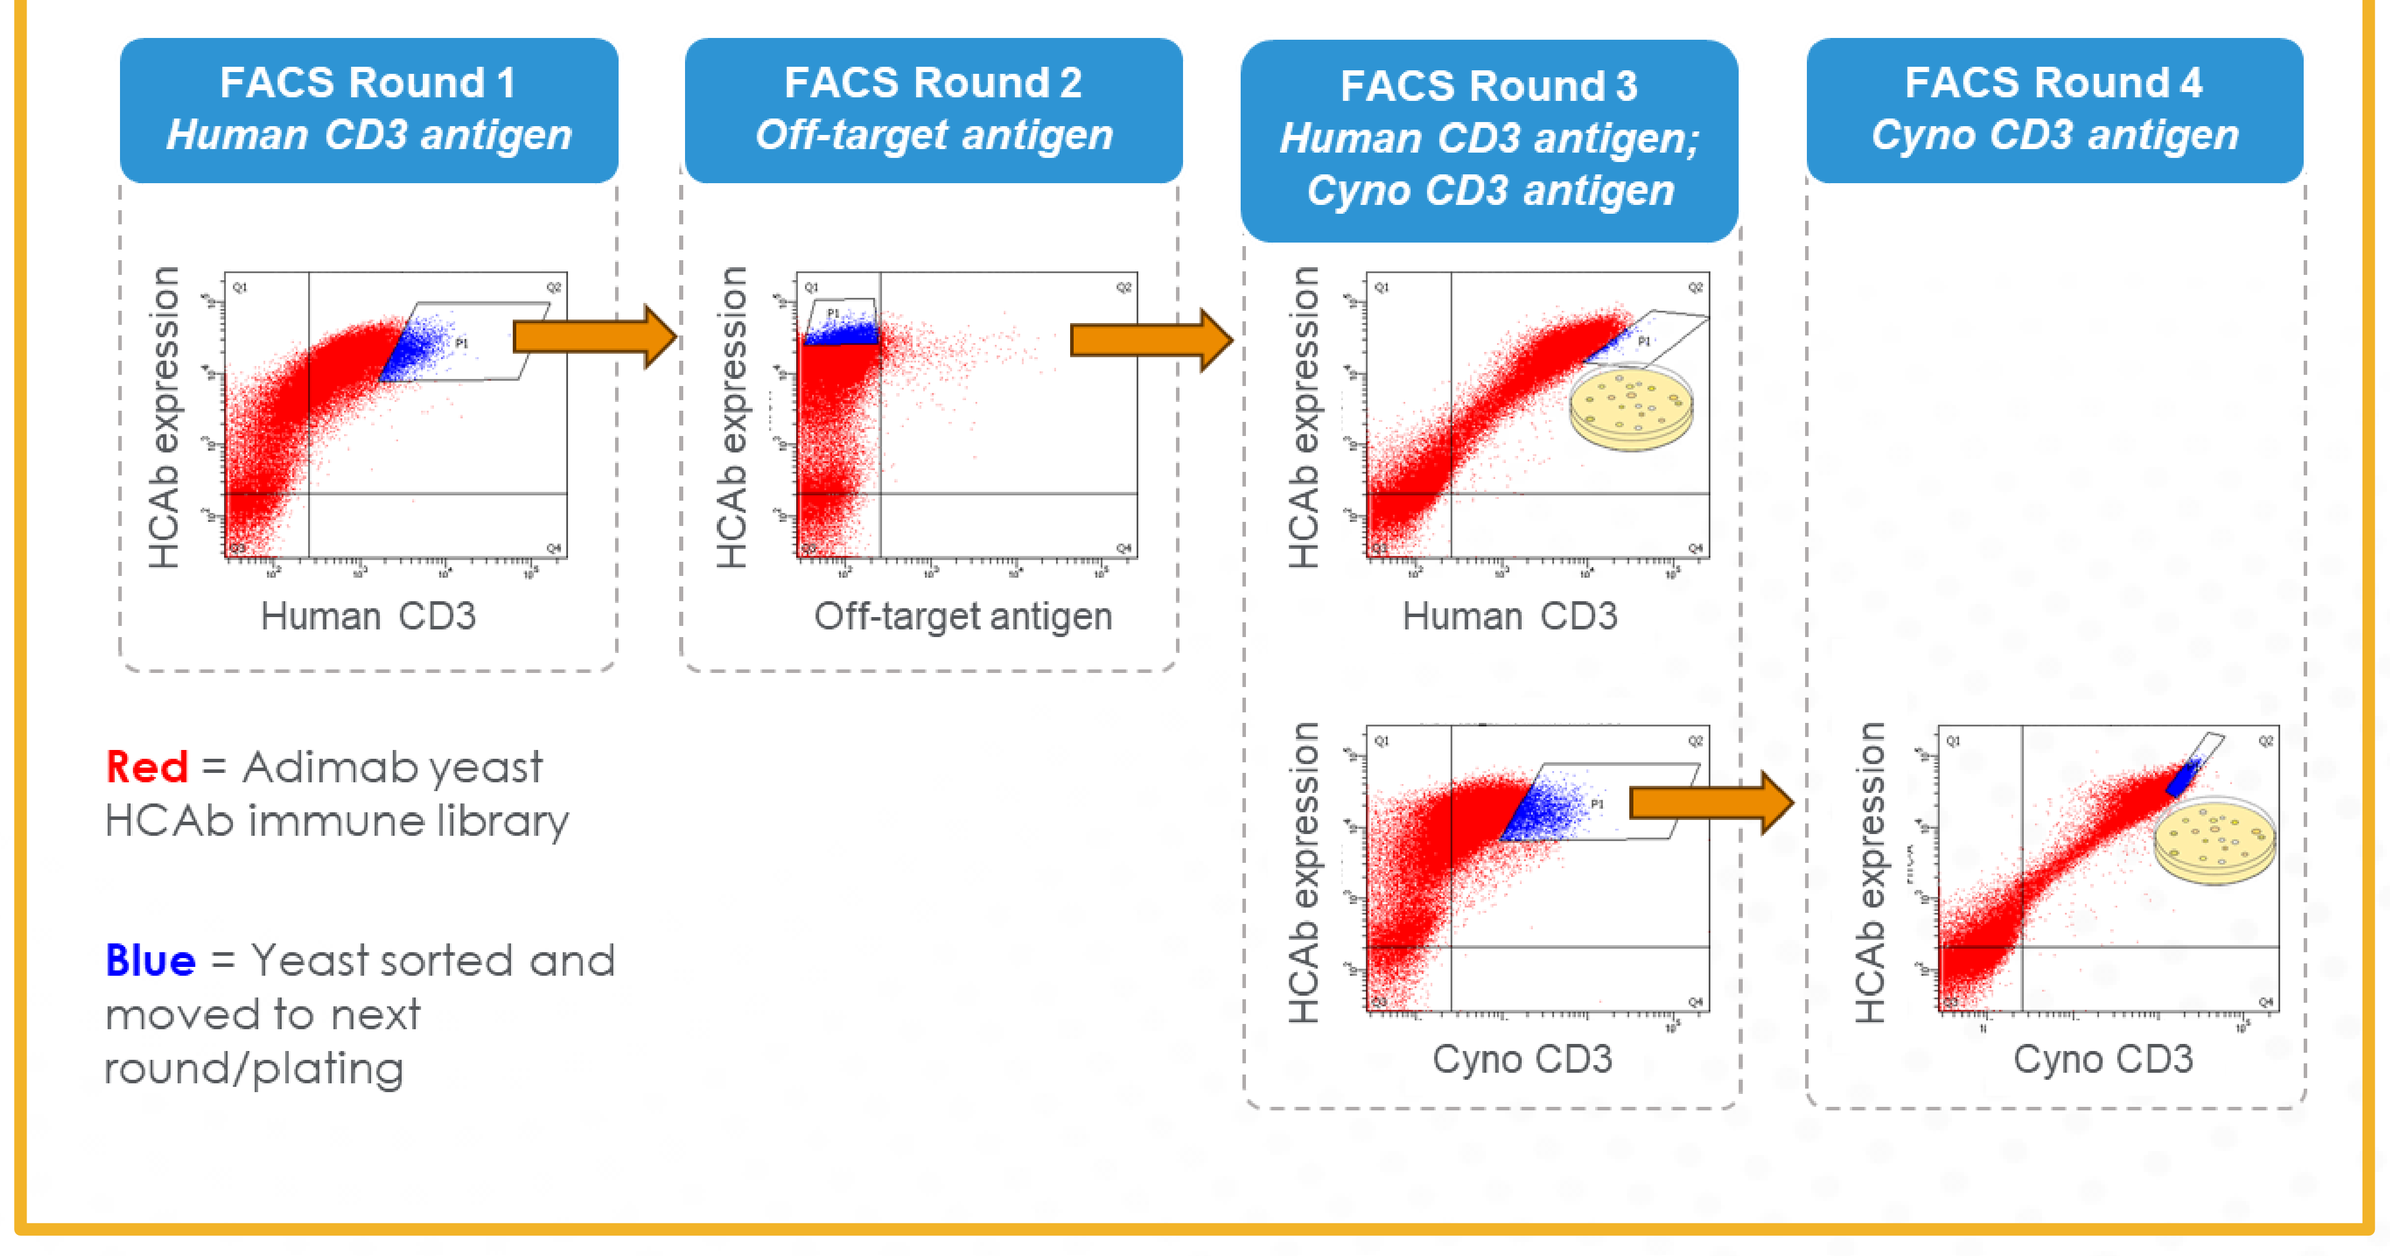
FACS Round 1
Human CD3 antigen
FACS Round 2
Off-target antigen
FACS Round 3
Human CD3 antigen; Cyno CD3 antigen
FACS Round 4
Cyno CD3 antigen
HCAb expression
Human CD3
Off-target antigen
Cyno CD3

Red = Adimab yeast HCAb immune library

Blue = Yeast sorted and moved to next round/plating

STIMULATING aCD3 HCABS DISCOVERED WITH FAVORABLE DEVELOPABILITY

T-cell stimulation assay

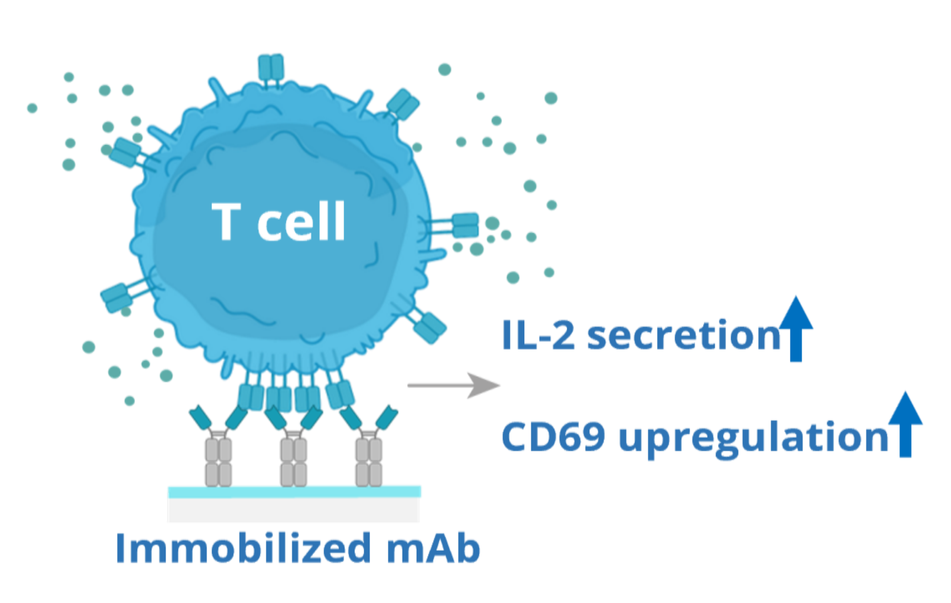
T cell
IL-2 secretion
CD69 upregulation
Immobilized mAb

1. HCAbs plated at 100nM
2. Jurkat T-cells incubated for 24h

MFI = Median Fluorescence Intensity

~10% of discovered HCAbs stimulate T-cells

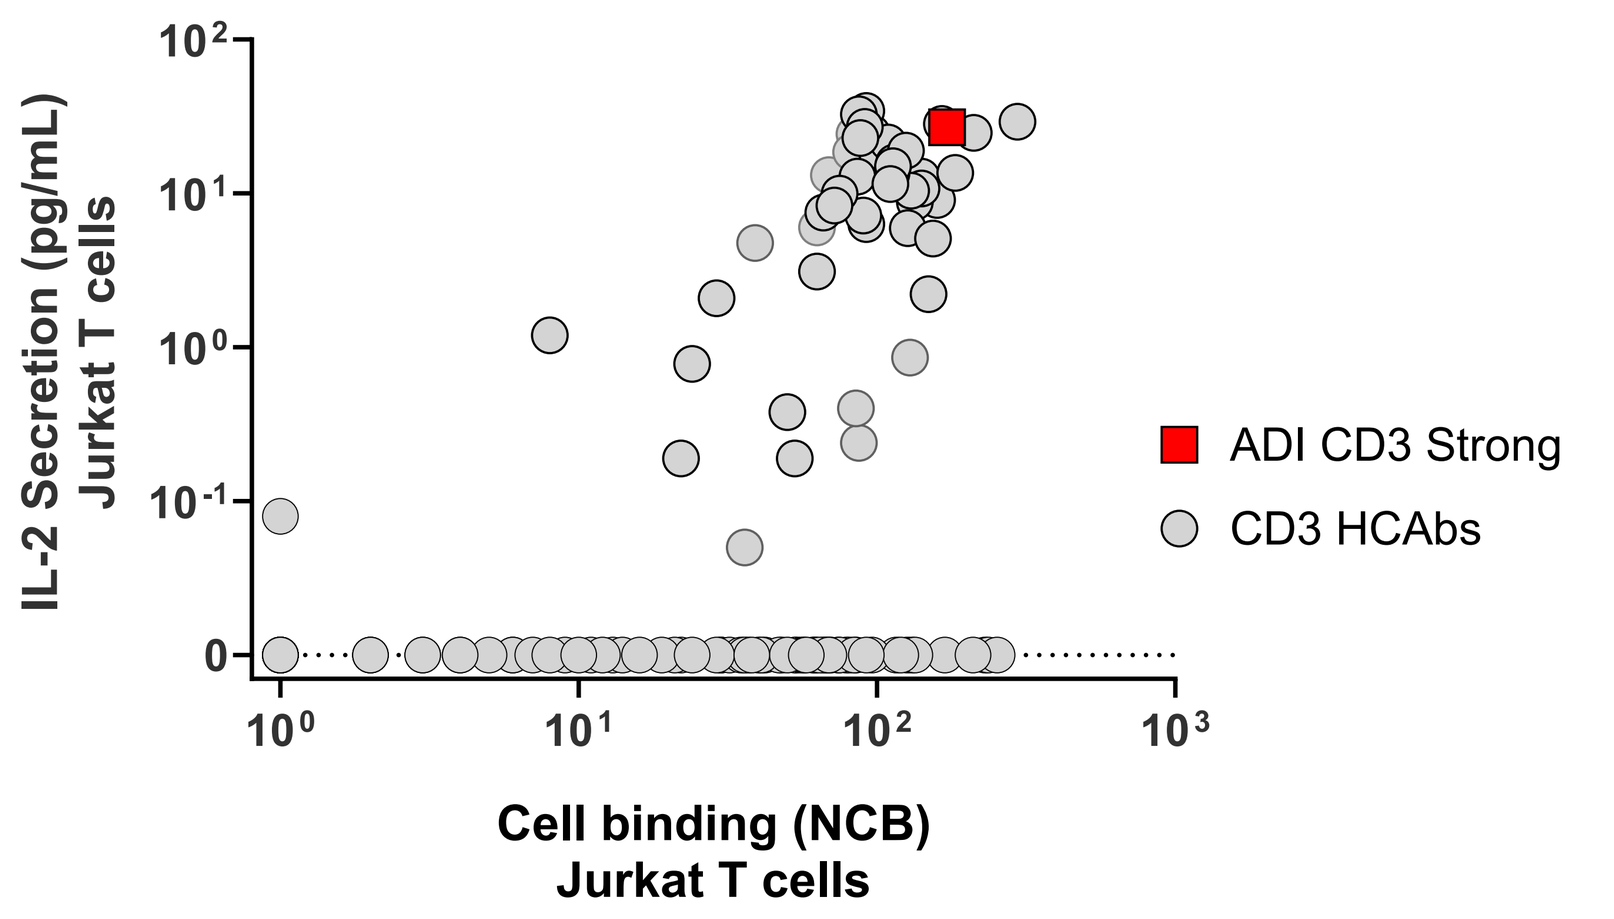
IL-2 Secretion (pg/mL) Jurkat T cells
10^{2}
10^{1}
10^{0}
10^{-1}
0
10^{0} 10^{1} 10^{2} 10^{3}
Cell binding (NCB) Jurkat T cells
ADI CD3 Strong
CD3 HCAbs

Stimulating HCAbs have favorable developability

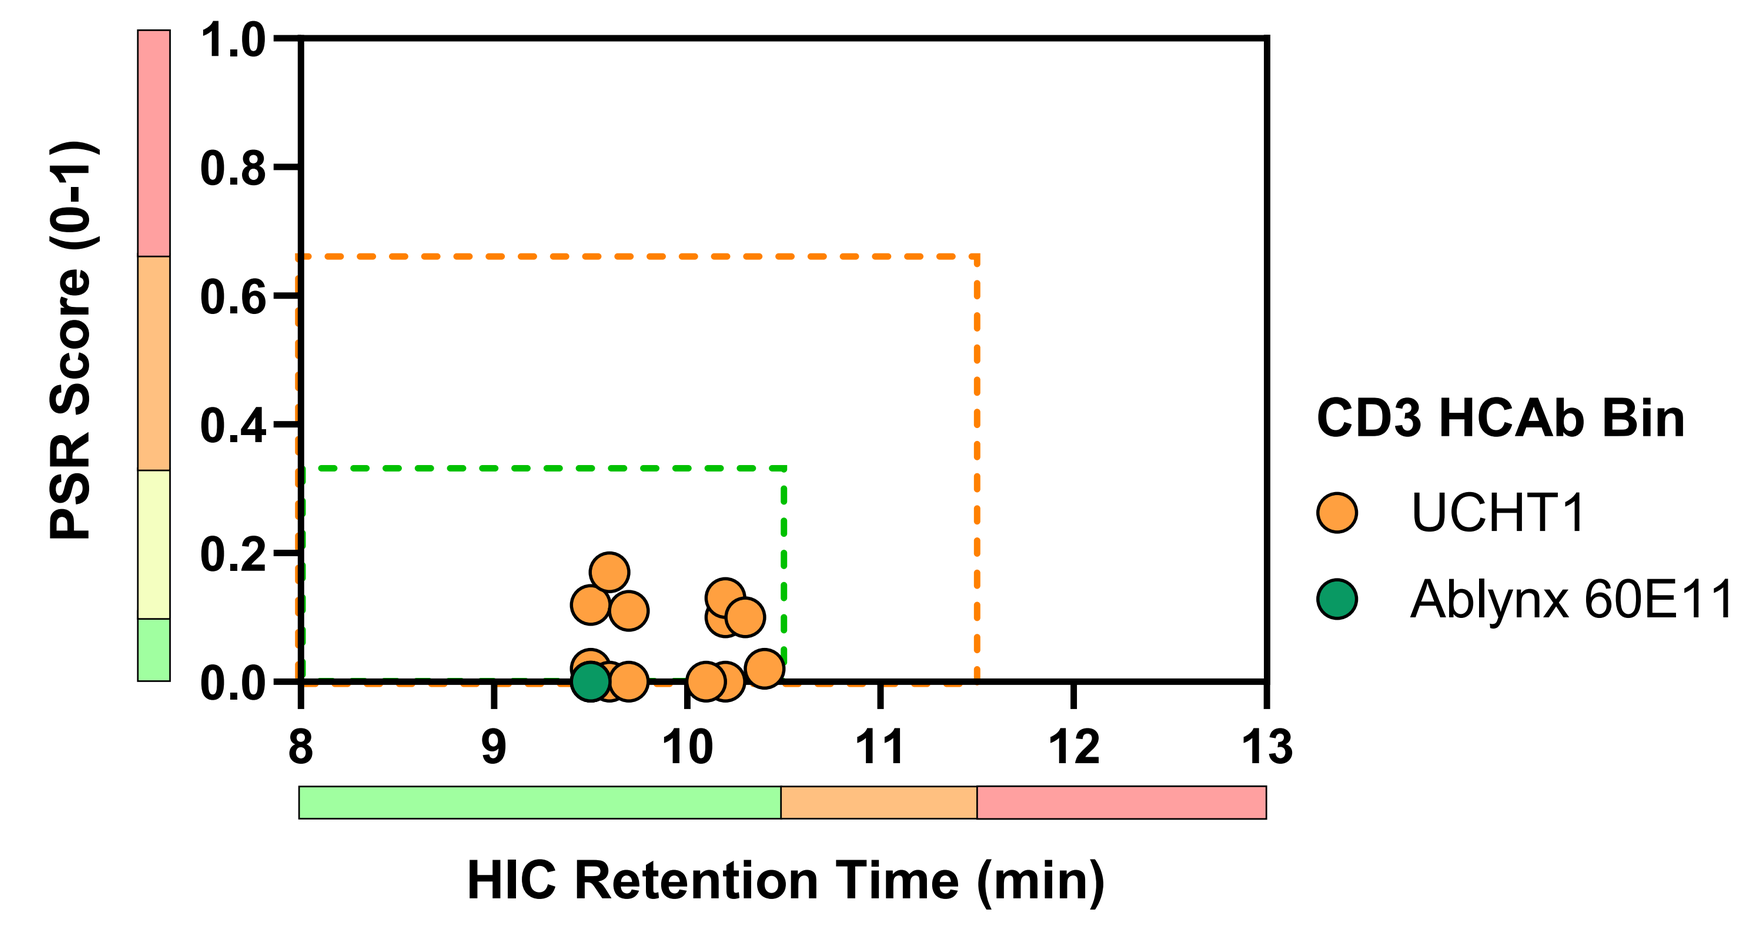
PSR Score (0-1)
1.0
0.8
0.6
0.4
0.2
0.0
8 9 10 11 12 13
HIC Retention Time (min)
CD3 HCAb Bin
UCHT1
Ablynx 60E11

VHH CRYSTAL STRUCTURES DEMONSTRATE BINDING TO CD3δε

Humanized ADI VHH Leads have overlapping binding patch with UCHT1

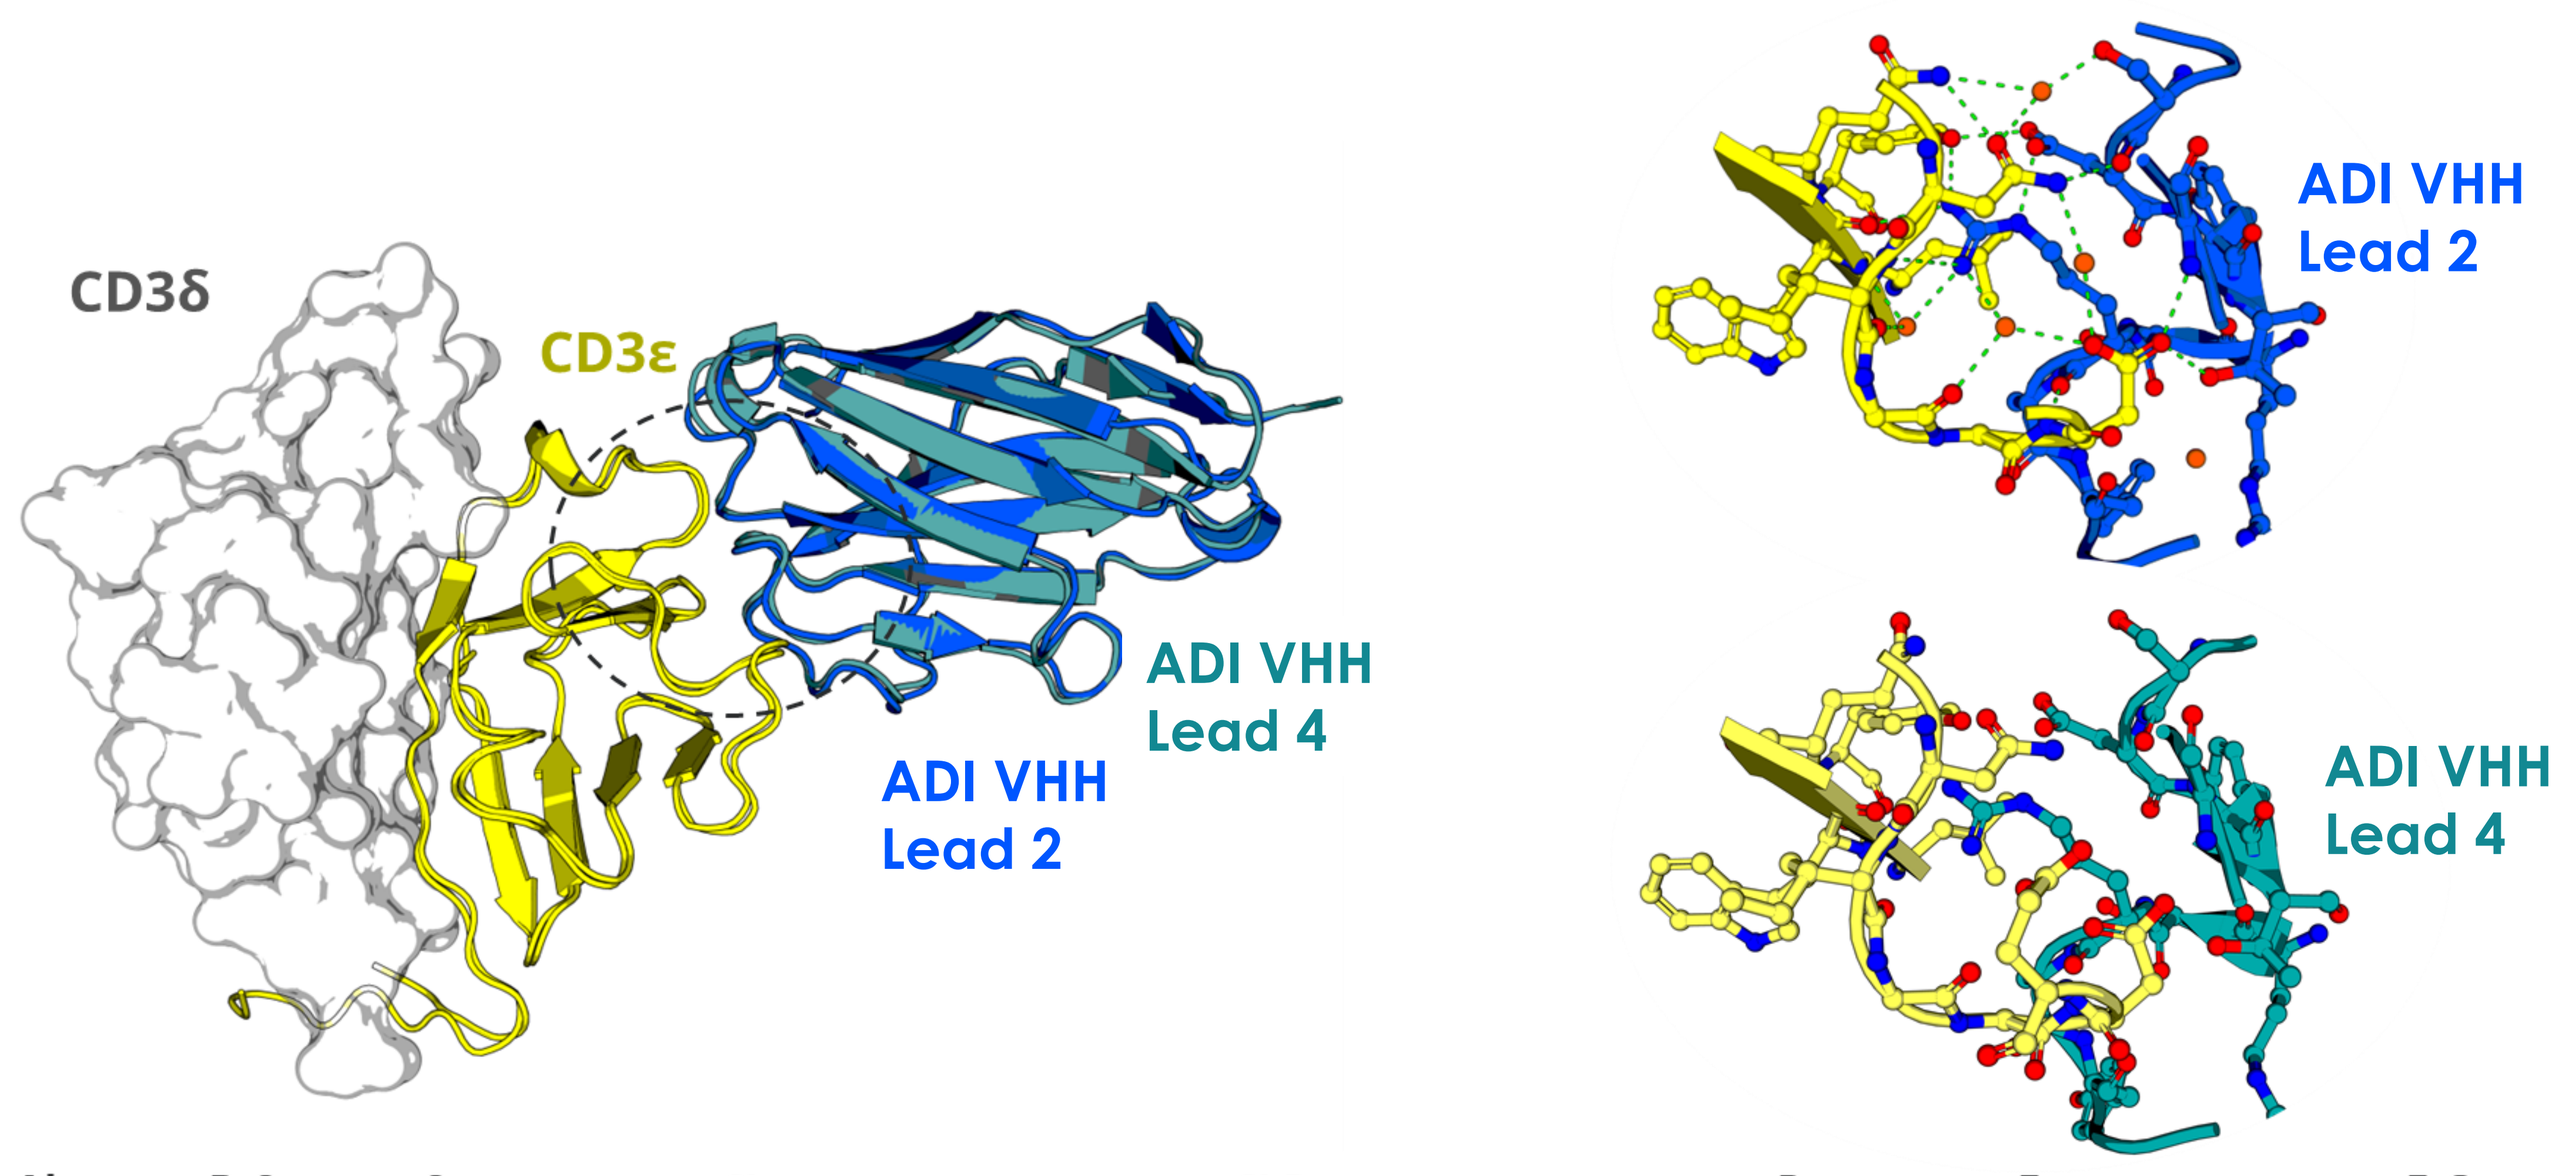
CD3δ
CD3ε
ADI VHH Lead 4
ADI VHH Lead 2
ADI VHH Lead 2
ADI VHH Lead 4

mAbs	B-C	C		C'			C'-D						D		F	F-G					
UCHT1	E35	L37			N44	I45	G46	G47	D48	E49		K52	S56	D57	Y78	R80	G81	S82	K83	P84	E85
ADI Lead 4	E35	L37		K43	N44			G47	D48	E49	D50		S56		Y78	R80	G81	S82	K83	P84	
ADI Lead 2	E35	L37	Q39		N44			G47	D48	E49	D50		S56		Y78		G81	S82	K83	P84	

contact residues

ADI VHH Leads are specific to Hu CD3ε

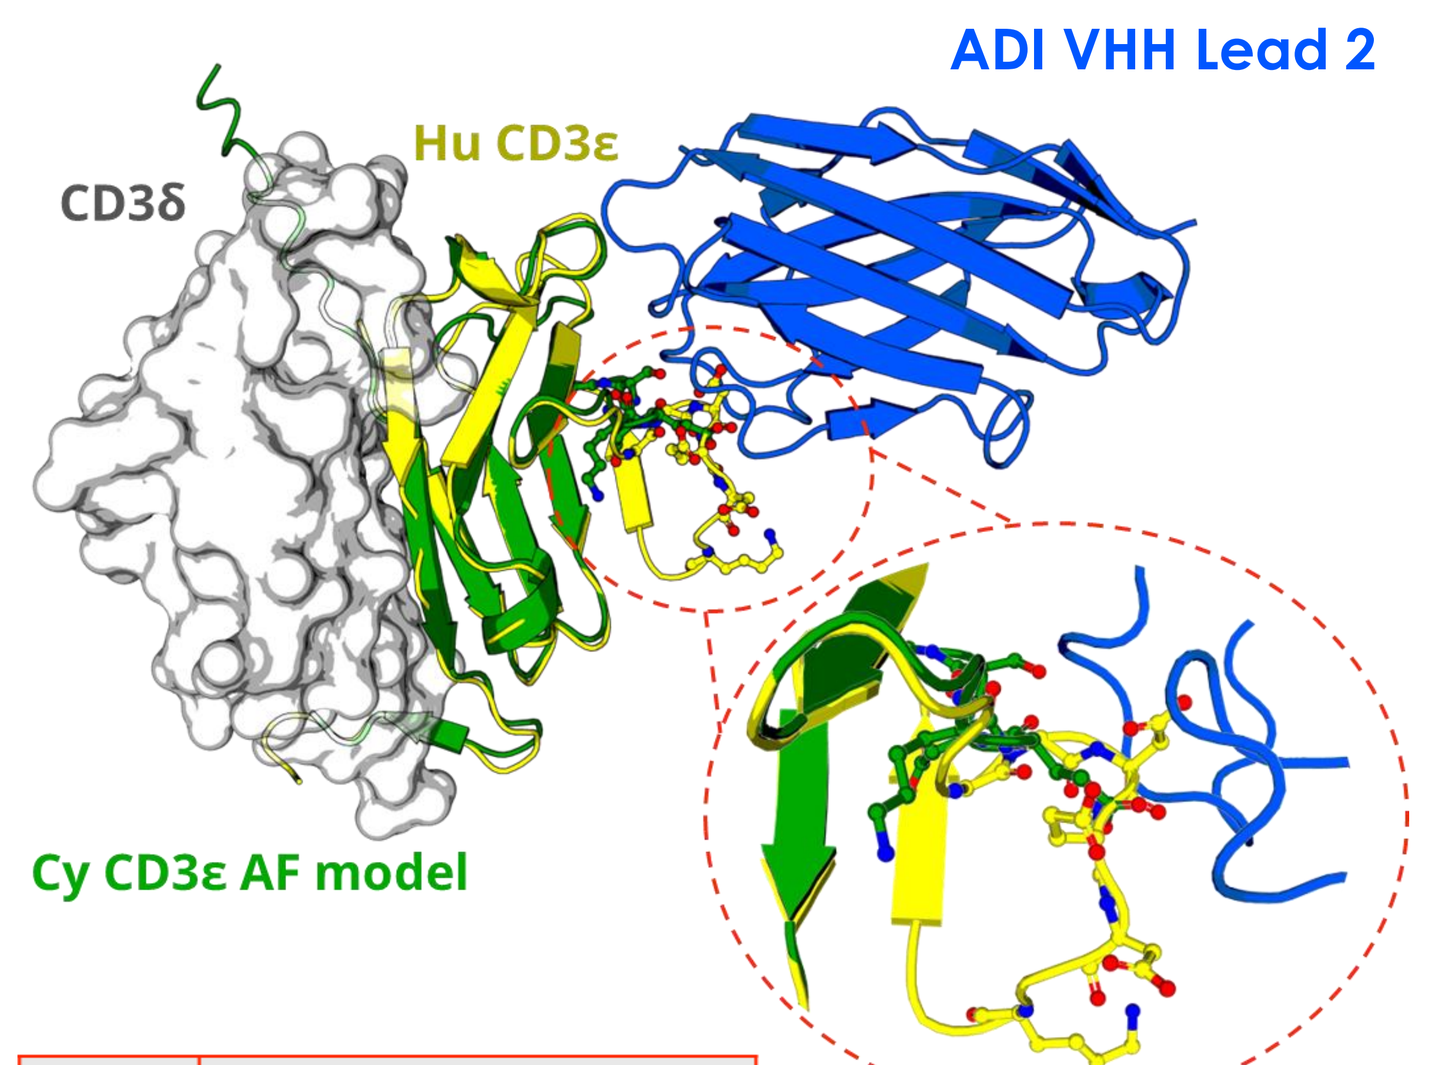
ADI VHH Lead 2
Hu CD3ε
CD3δ
Cy CD3ε AF model

Species	CD3ε C'-D loop
Hu	KNIGGDEDDKNIGSDEDH
Cy	KN---KEDSG------DR
Rh	KN---KEDSG------DR

aCD3 T-CELL ENGAGERS SHOW FUNCTIONAL ACTIVITY IN MULTIPLE FORMATS

CD3 VHH x CD20 bispecific TCEs elicit killing

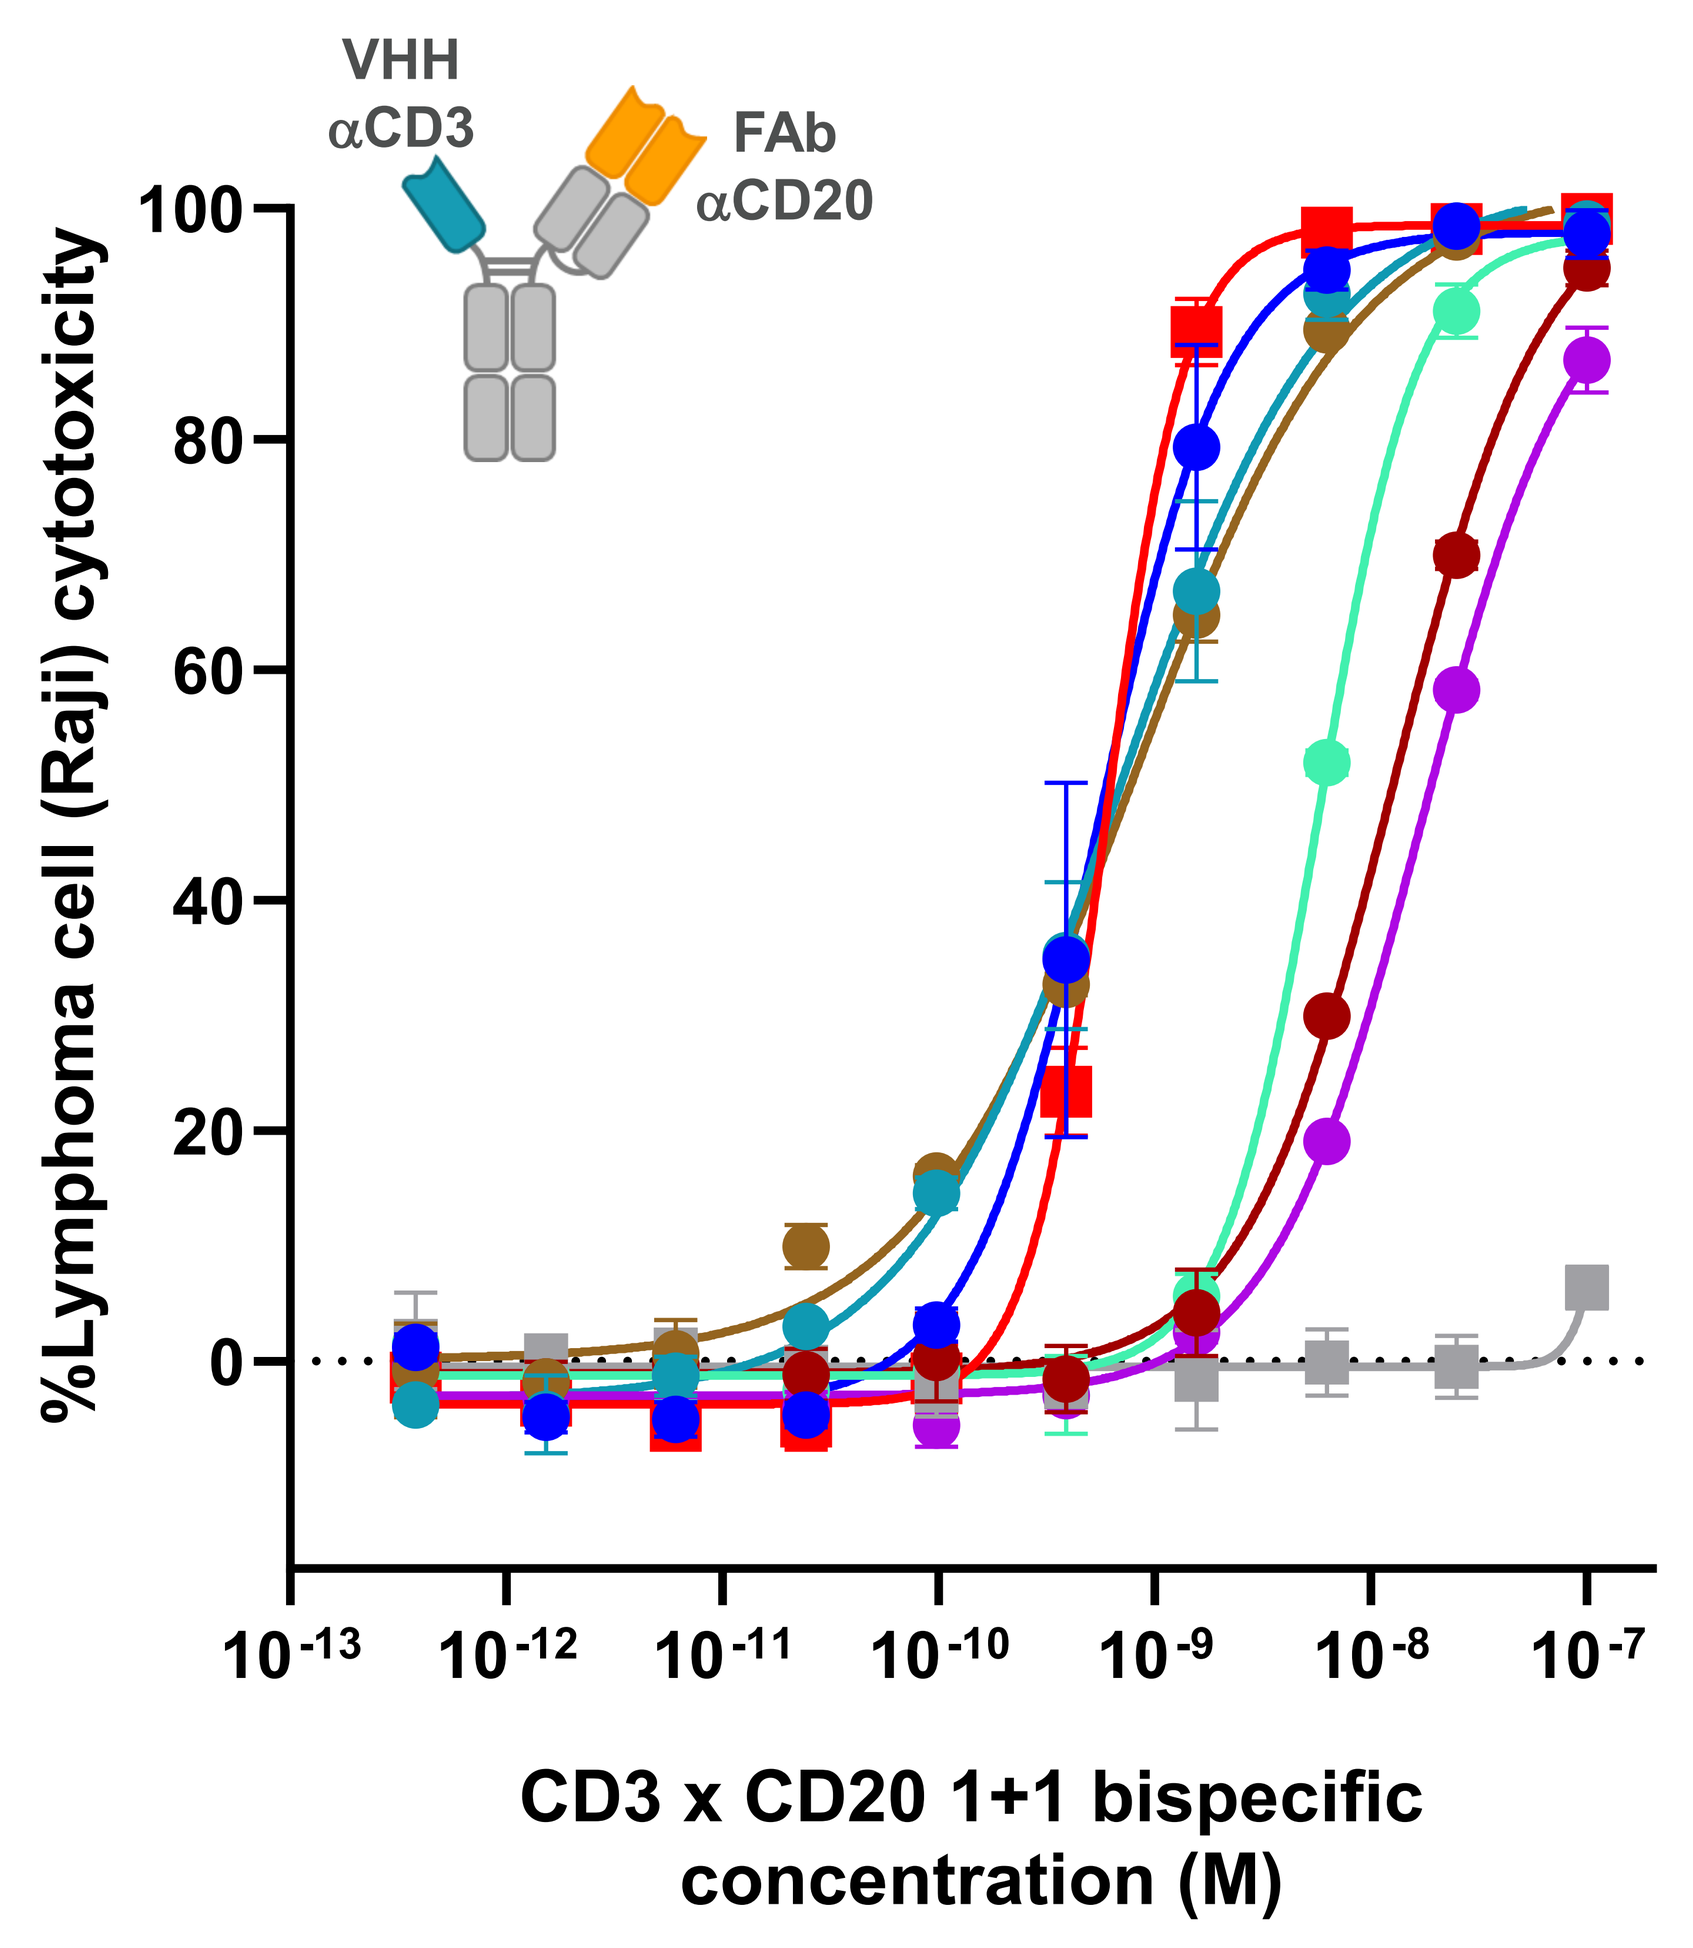
VHH αCD3
FAb αCD20
%Lymphoma cell (Raji) cytotoxicity
100
80
60
40
20
0
10^{-13} 10^{-12} 10^{-11} 10^{-10} 10^{-9} 10^{-8} 10^{-7}
CD3 x CD20 1+1 bispecific concentration (M)

CD3 VHH – TCR fusion TCEs elicit killing

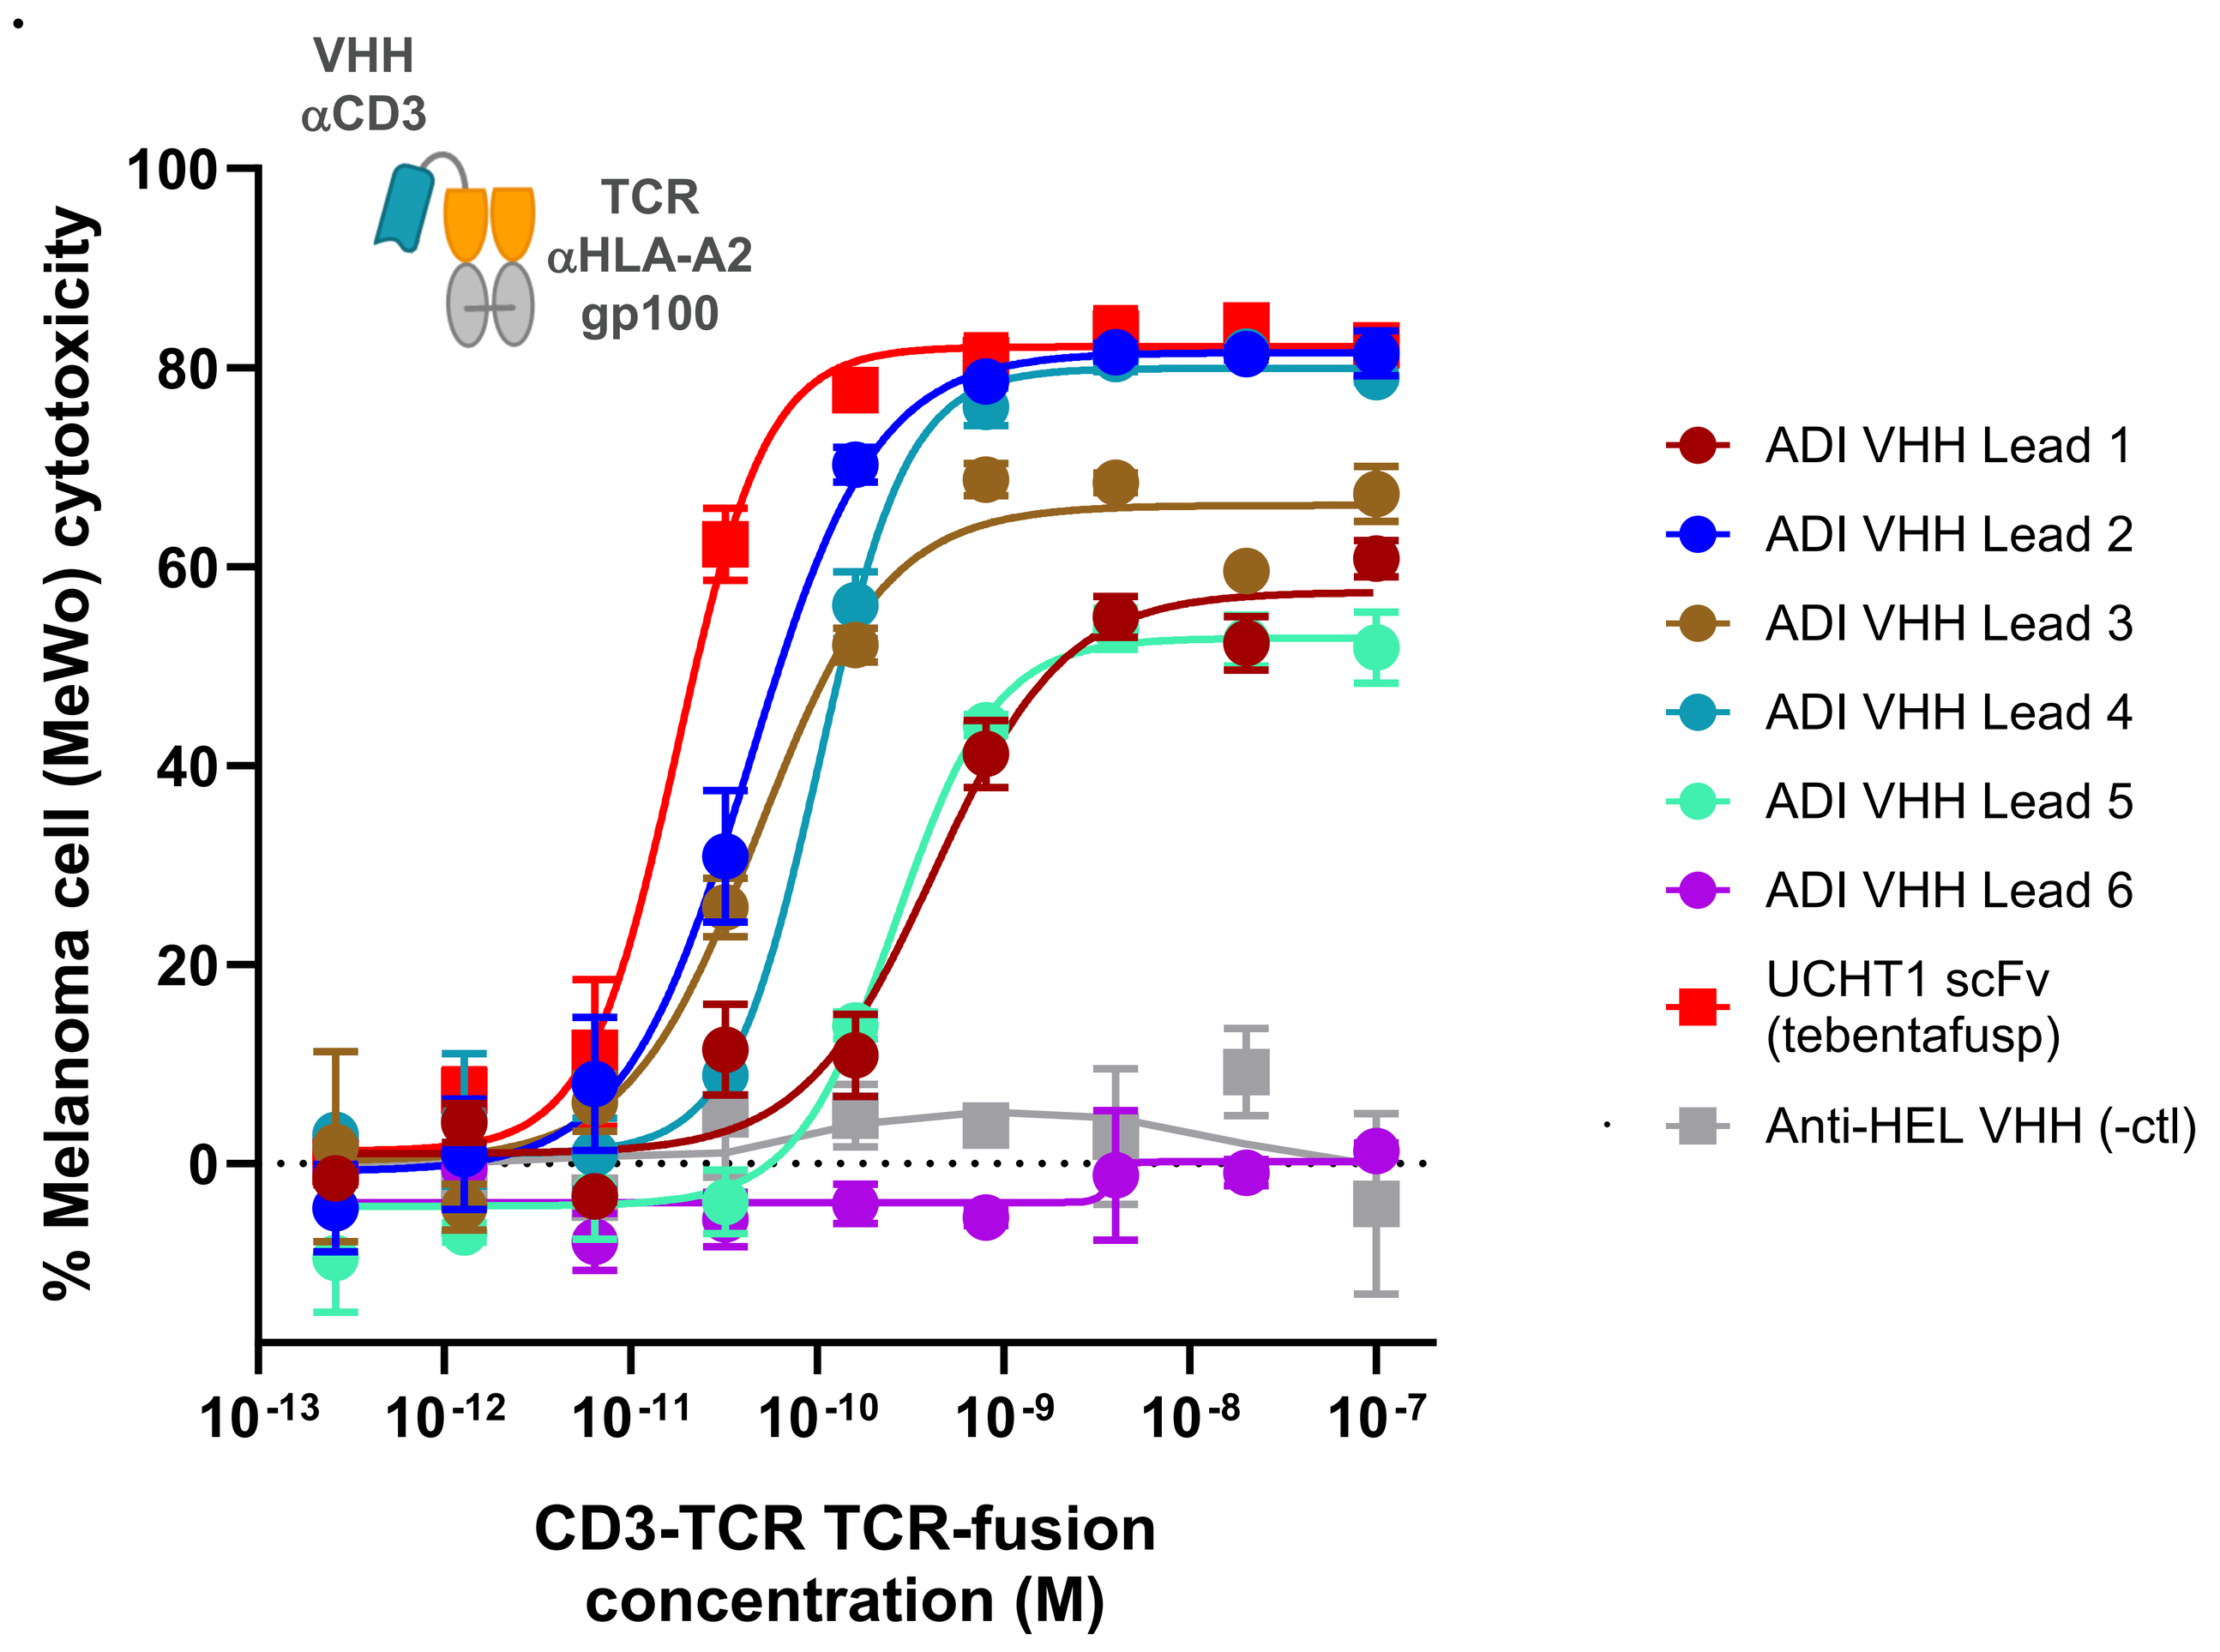
VHH αCD3
TCR αHLA-A2 gp100
% Melanoma cell (MeWo) cytotoxicity
100
80
60
40
20
0
10^{-13} 10^{-12} 10^{-11} 10^{-10} 10^{-9} 10^{-8} 10^{-7}
CD3-TCR TCR-fusion concentration (M)
ADI VHH Lead 1
ADI VHH Lead 2
ADI VHH Lead 3
ADI VHH Lead 4
ADI VHH Lead 5
ADI VHH Lead 6
UCHT1 scFv (tebentafusp)
Anti-HEL VHH (-ctl)

ADI VHH panel exhibits range in T-cell binding

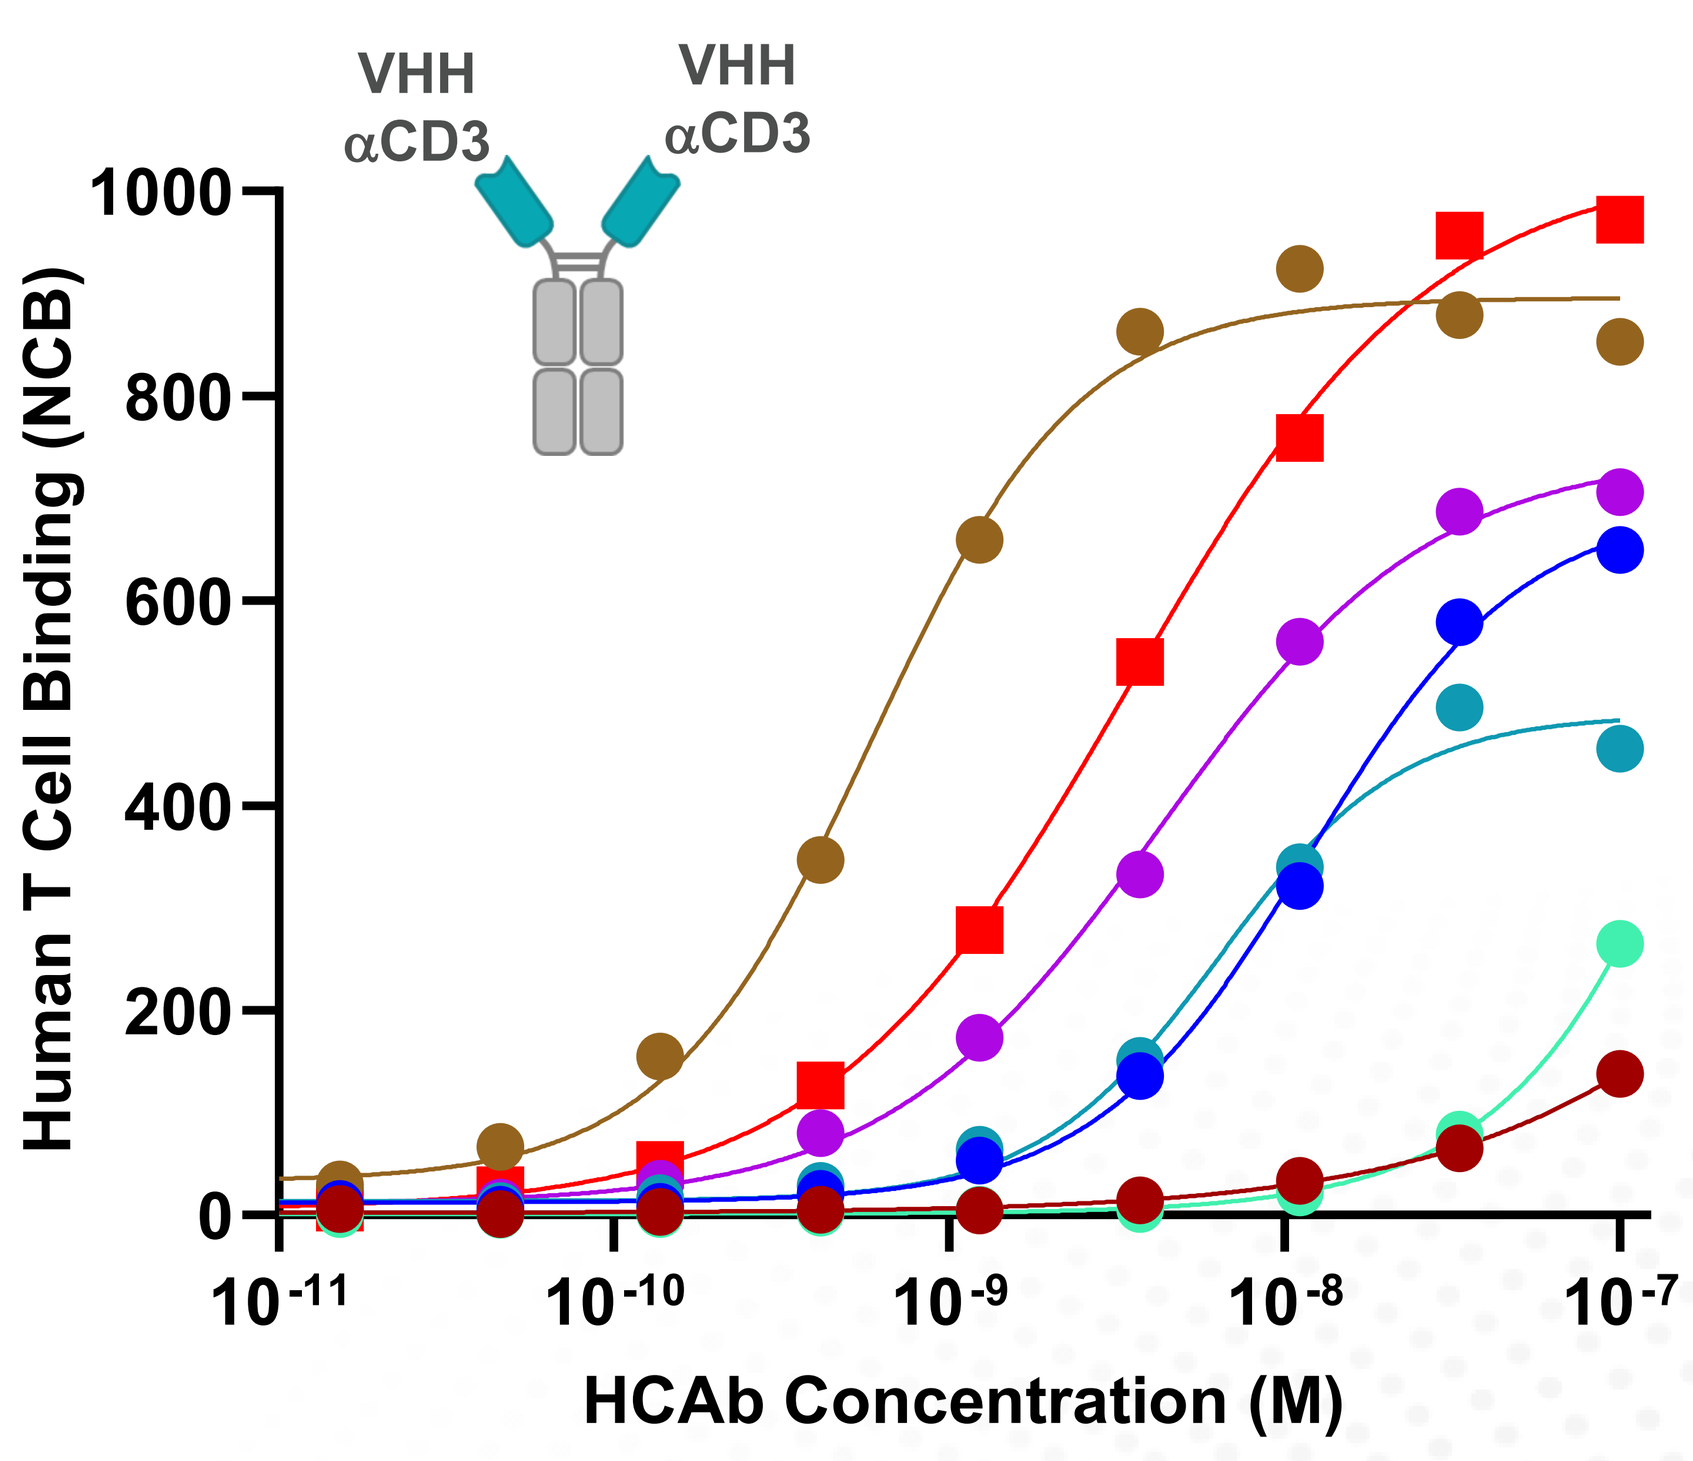
VHH αCD3
VHH αCD3
Human T Cell Binding (NCB)
1000
800
600
400
200
0
10^{-11} 10^{-10} 10^{-9} 10^{-8} 10^{-7}
HCAb Concentration (M)

ADI VHH panel spans wide affinity range

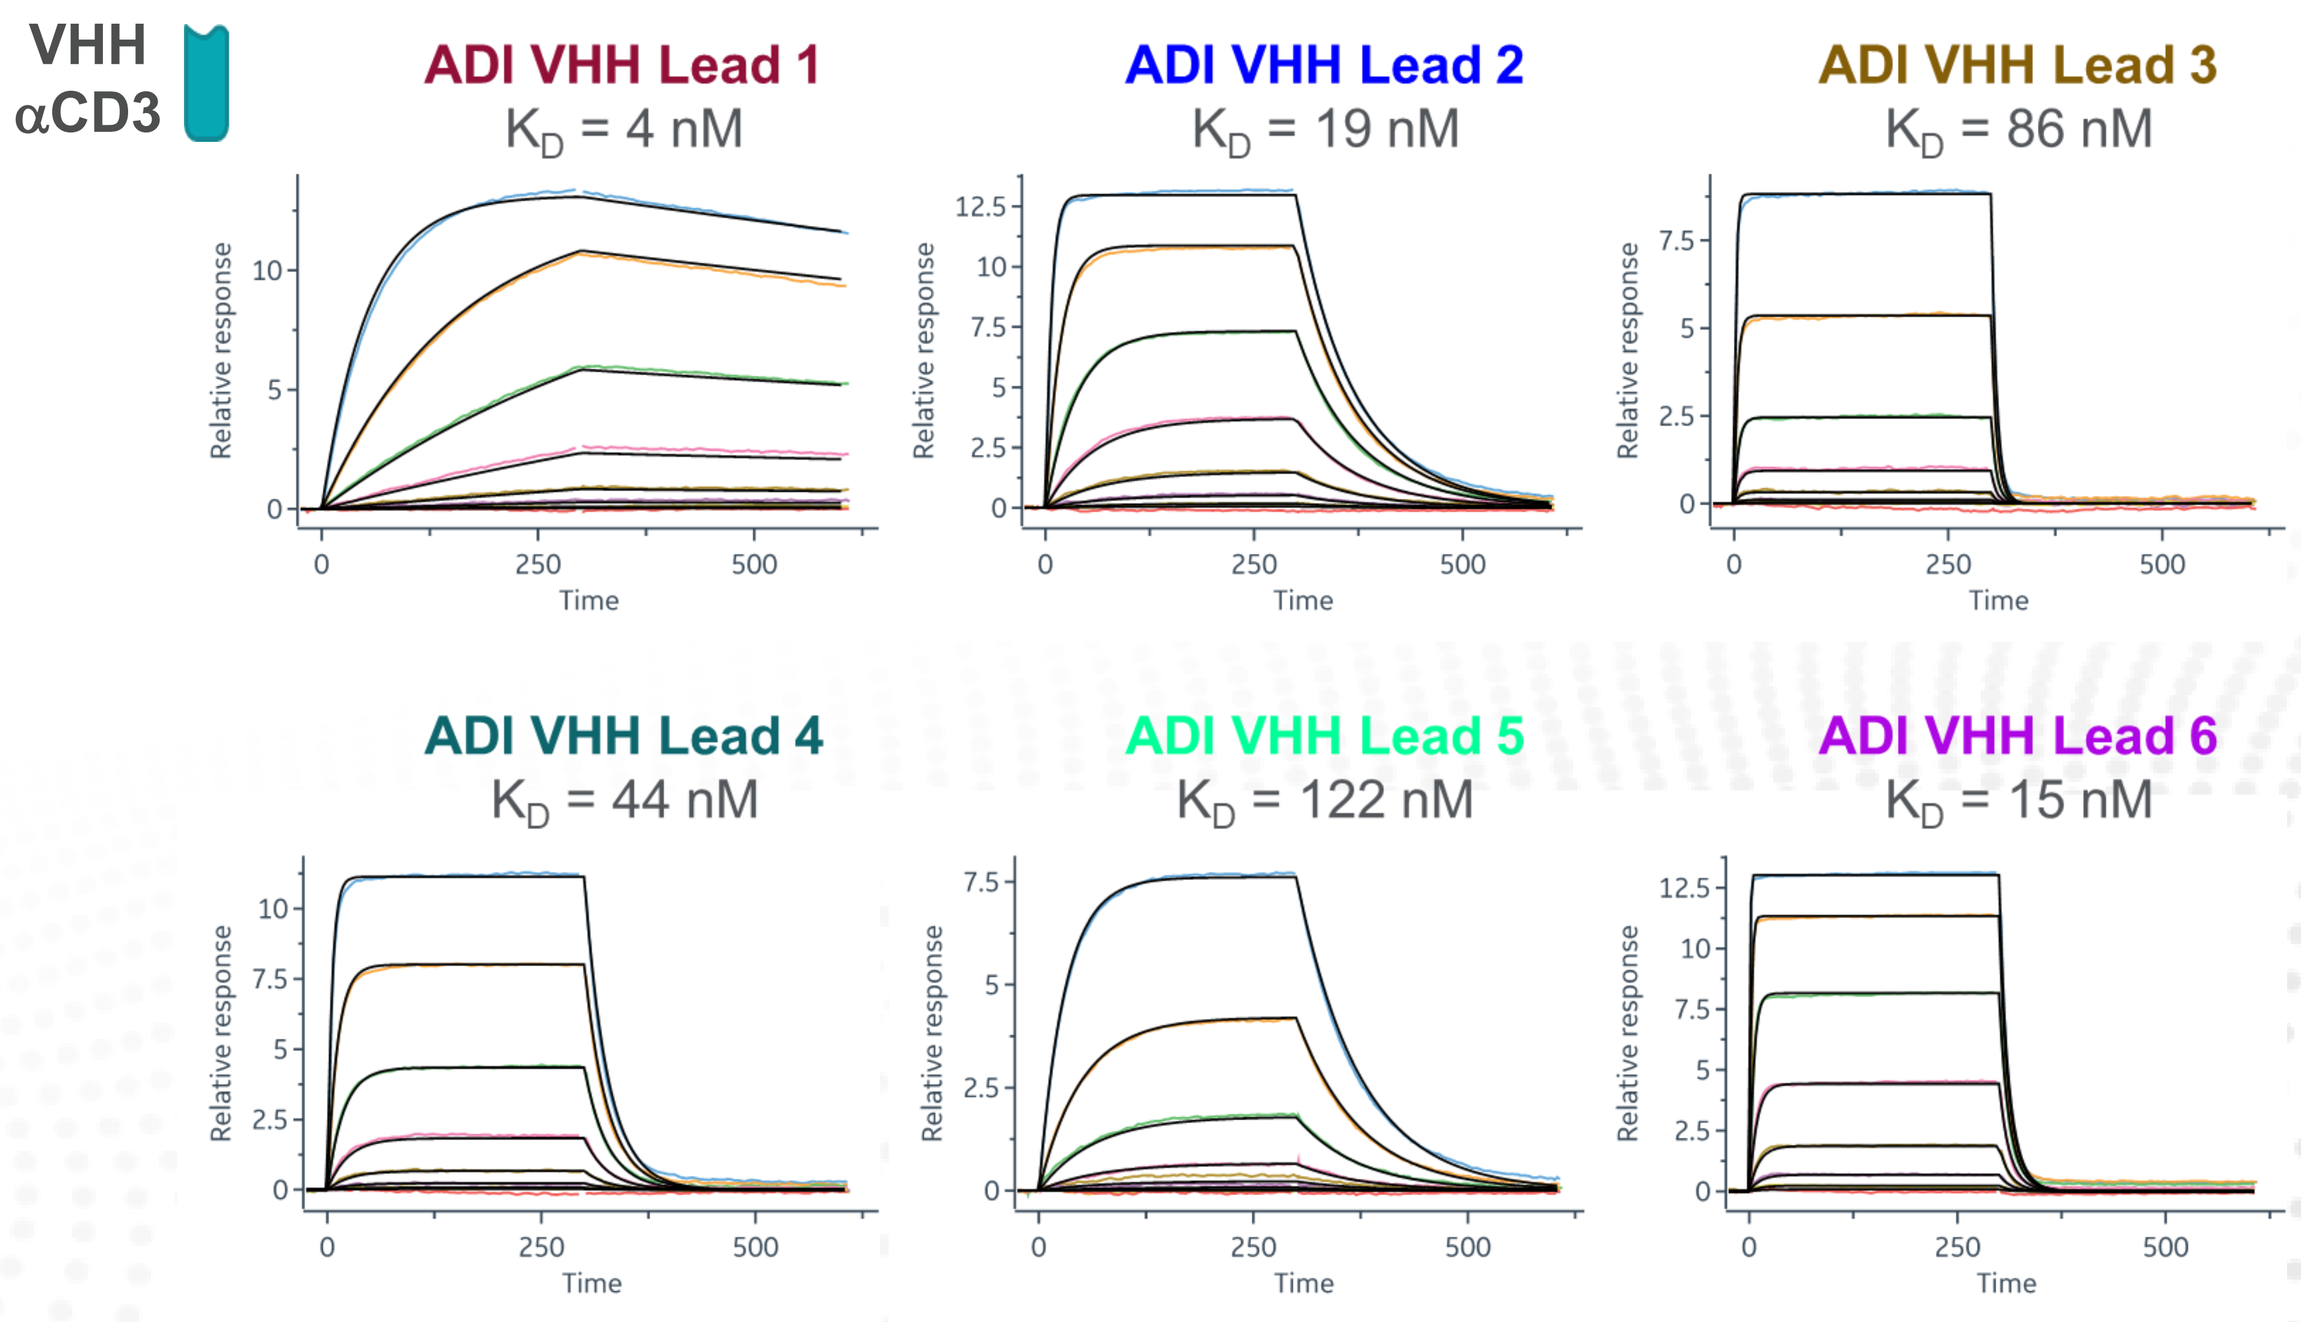
VHH αCD3
ADI VHH Lead 1
K_D = 4 nM
ADI VHH Lead 2
K_D = 19 nM
ADI VHH Lead 3
K_D = 86 nM
ADI VHH Lead 4
K_D = 44 nM
ADI VHH Lead 5
K_D = 122 nM
ADI VHH Lead 6
K_D = 15 nM
Relative response
Time

SUMMARY

- CD3 VHHs reduce the manufacturing complexity of TCEs
- Adimab's VHHs have functional activity comparable to clinically validated TCEs in both IgG bispecific and TCR fusion formats
- Crystal structures obtained for two of the leads show over an overlapping binding patch with UCHT1
- CD3 VHHs are accessible as part of Adimab's non-exclusive TCE offering

Partnering with Adimab

Please reach out to our Business Development department at bd@adimab.com.

The QR code on the right links to Adimab posters and other resources.